Imprime PGG, a soluble yeast β-glucan PAMP, enhancement of anti-tumor responses in combination with tumor targeting antibody is highly dependent on NK cell killing

AACR 2018
Poster # 3767

Kathryn A. Fraser, Takashi Kangas, Ross B. Fulton, Steven M. Leonardo, Ben Harrison, Yumi Yokoyama, Nandita Bose, Jeremy R. Graff, Mark Uhlik, and Keith B. Gorden.
Biothera Pharmaceuticals, Inc., Eagan MN, 55121

Abstract

Cancer therapy has been reshaped by checkpoint inhibitors (CPIs), making it possible for durable responses against cancers with traditionally low cure rates. Current efforts are focused on combination therapies in the hopes of evading resistance to CPIs and improving overall response. One escape mechanism attributed to acquired resistance to CPIs includes defective antigen presentation, namely a loss in MHC class I expression. This leads to loss of CD8 T cell-mediated tumor killing and disease relapse. This recent revelation has stimulated a need for therapies that activate other cytotoxic effector cells such as NK cells to kill tumors. Imprime PGG (Imprime) is a soluble, systemically delivered yeast 1,3/1,6 β-glucan PAMP (pathogen-associated molecular pattern) capable of triggering innate immune cell function leading to a cascade of immune activation and enhanced anti-tumor killing. Imprime activates the innate immune system via dectin-1, eliciting production of a variety of chemokines and cytokines, including type I IFN, leading to the mobilization and stimulation of innate cell types including dendritic cells and monocytes. Unlike other PAMPs for which systemic administration often leads to toxic side effects, Imprime has been administered safely by intravenous infusion to >400 human subjects. Currently, Imprime PGG is being evaluated in combination with αPD1 therapy in multiple clinical trials.

Previously we have shown that combination therapy of anti-Trp1 antibody and Imprime leads to a significant reduction in both number and size of lung metastases in the B16F10 metastatic melanoma tumor model over αTrp1 antibody alone. This reduction of metastases is highly dependent on NK cells but not CD8 T cells. To explore the impact of Imprime on NK-mediated cytotoxicity, we further evaluated in vivo killing of MHC class I deficient TapKO cells after intravenous administration of Imprime. In these experiments Imprime was able to enhance the NK cytotoxic killing of the targets. All NK cell killing observed was dependent on type I IFN, phagocytic cells and dectin 1. Imprime treatment increased cytokines that drive enhanced NK activation and effector phenotype. Significant increases were observed in the cytokines IL15/IL15rα, IL18, IL12p70 in lymph node lysates as well as increases in the effector molecules CD69, GrB, and CD107a on splenic NK cells. IL15/IL15rα production was dependent on type I IFN, Dectin 1, and phagocytic cells. Interestingly, Ly6c hi monocytes, which are increased after Imprime treatment, also show enhancement of IL15rα expression.

Collectively, these data demonstrate that Imprime contributes to enhanced NK functionality and killing which may provide a unique immunotherapeutic approach to complement existing therapies.

Structure of Imprime PGG

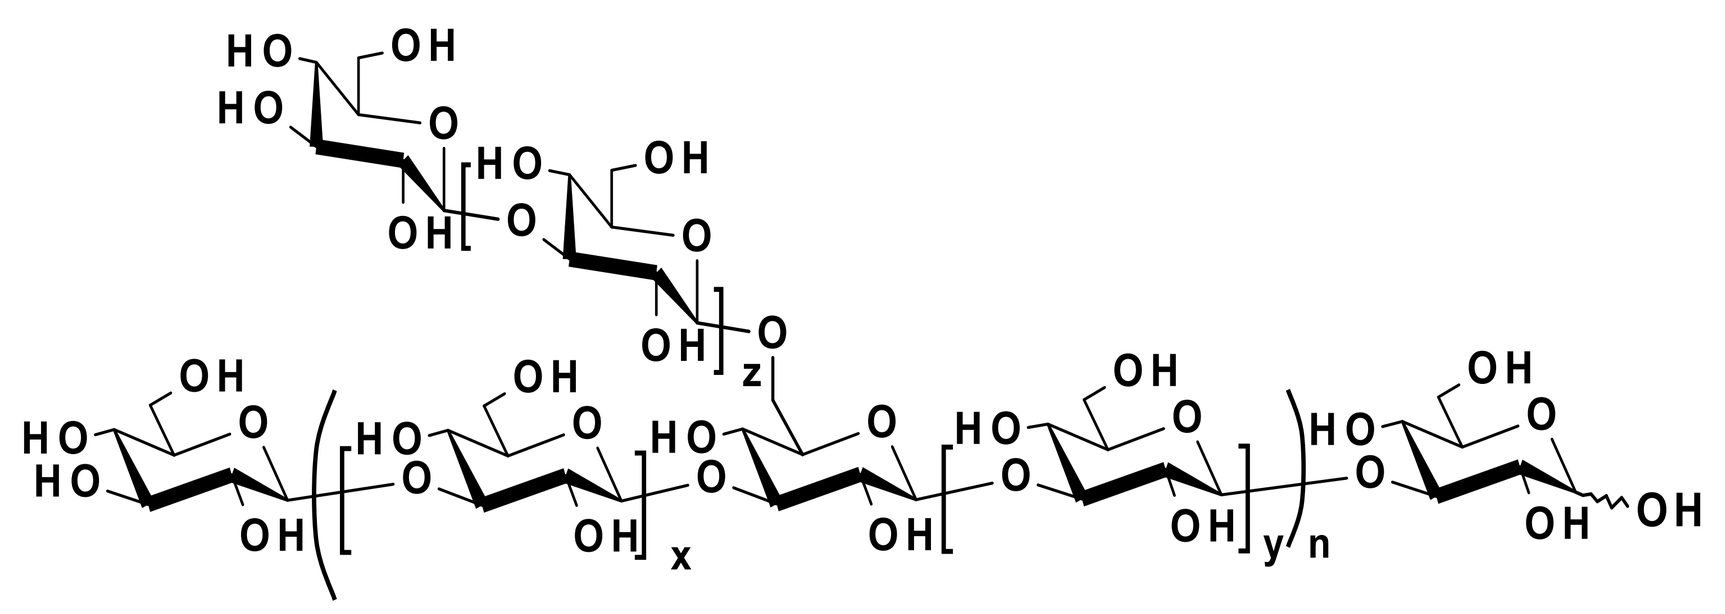

Figure 1. Combination of Imprime PGG with a tumor targeting antibody enhances NK mediated killing/ADCC of B16 metastatic melanoma

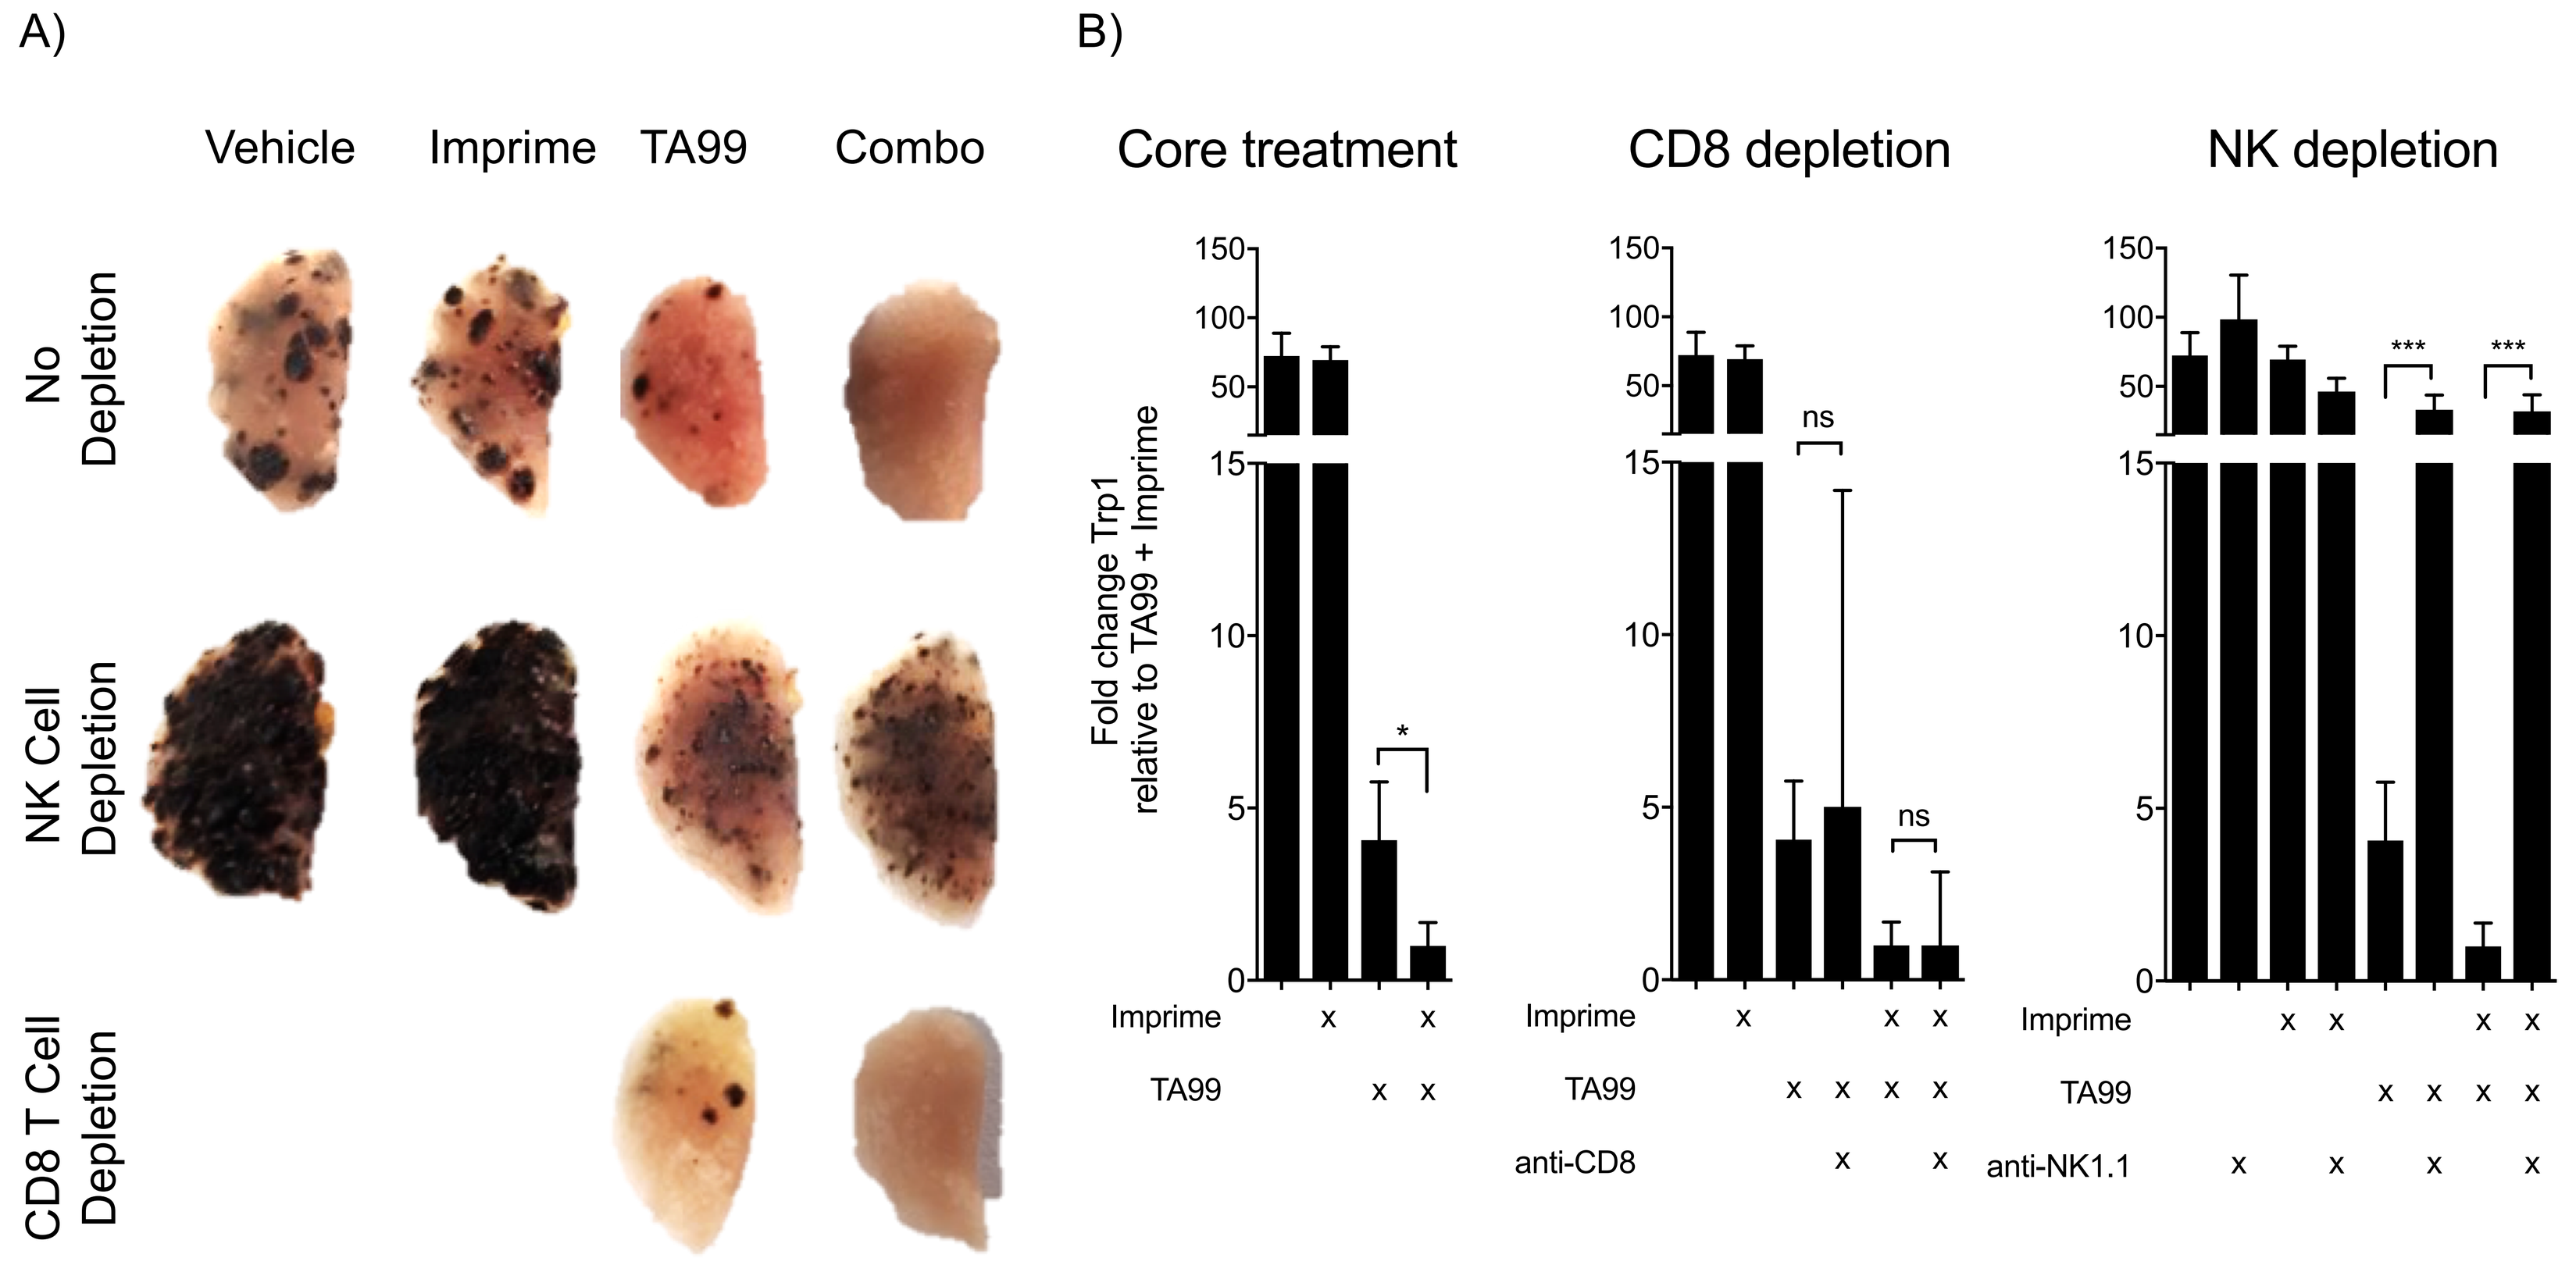
A)
Vehicle
Imprime
TA99
Combo
No Depletion
NK Cell Depletion
CD8 T Cell Depletion
B)
Core treatment
CD8 depletion
NK depletion
Fold change Trp1 relative to TA99 + Imprime
Imprime
TA99
anti-CD8
anti-NK1.1

B16 lung melanoma model. 1e5 B16F10 cells were administered i.v. and treatments were administered as follows. Imprime (1.2mg) D1, 3, 7, 10, 14; TA99 (50ug) D1,D3,D5,D7,D10;anti-NK1.1 and anti-CD8 200ug D1, 100ugx2/wk. Mice were euthanized on D21 and lungs removed. (A) Picture of mouse lungs day 21 post tumor inoculation. (B) Relative expression of Trp1 message in the lungs on day 21 post tumor inoculation. Combination treatment showed enhance tumor efficacy vs TA99 alone. Upon NK depletion (right), but not CD8 T cell depletion (center), the enhancement was lost indicating that the tumor efficacy mediated via combination treatment was NK cell dependent.

Figure 2. Imprime PGG treatment results in enhanced killing of NK cell targets

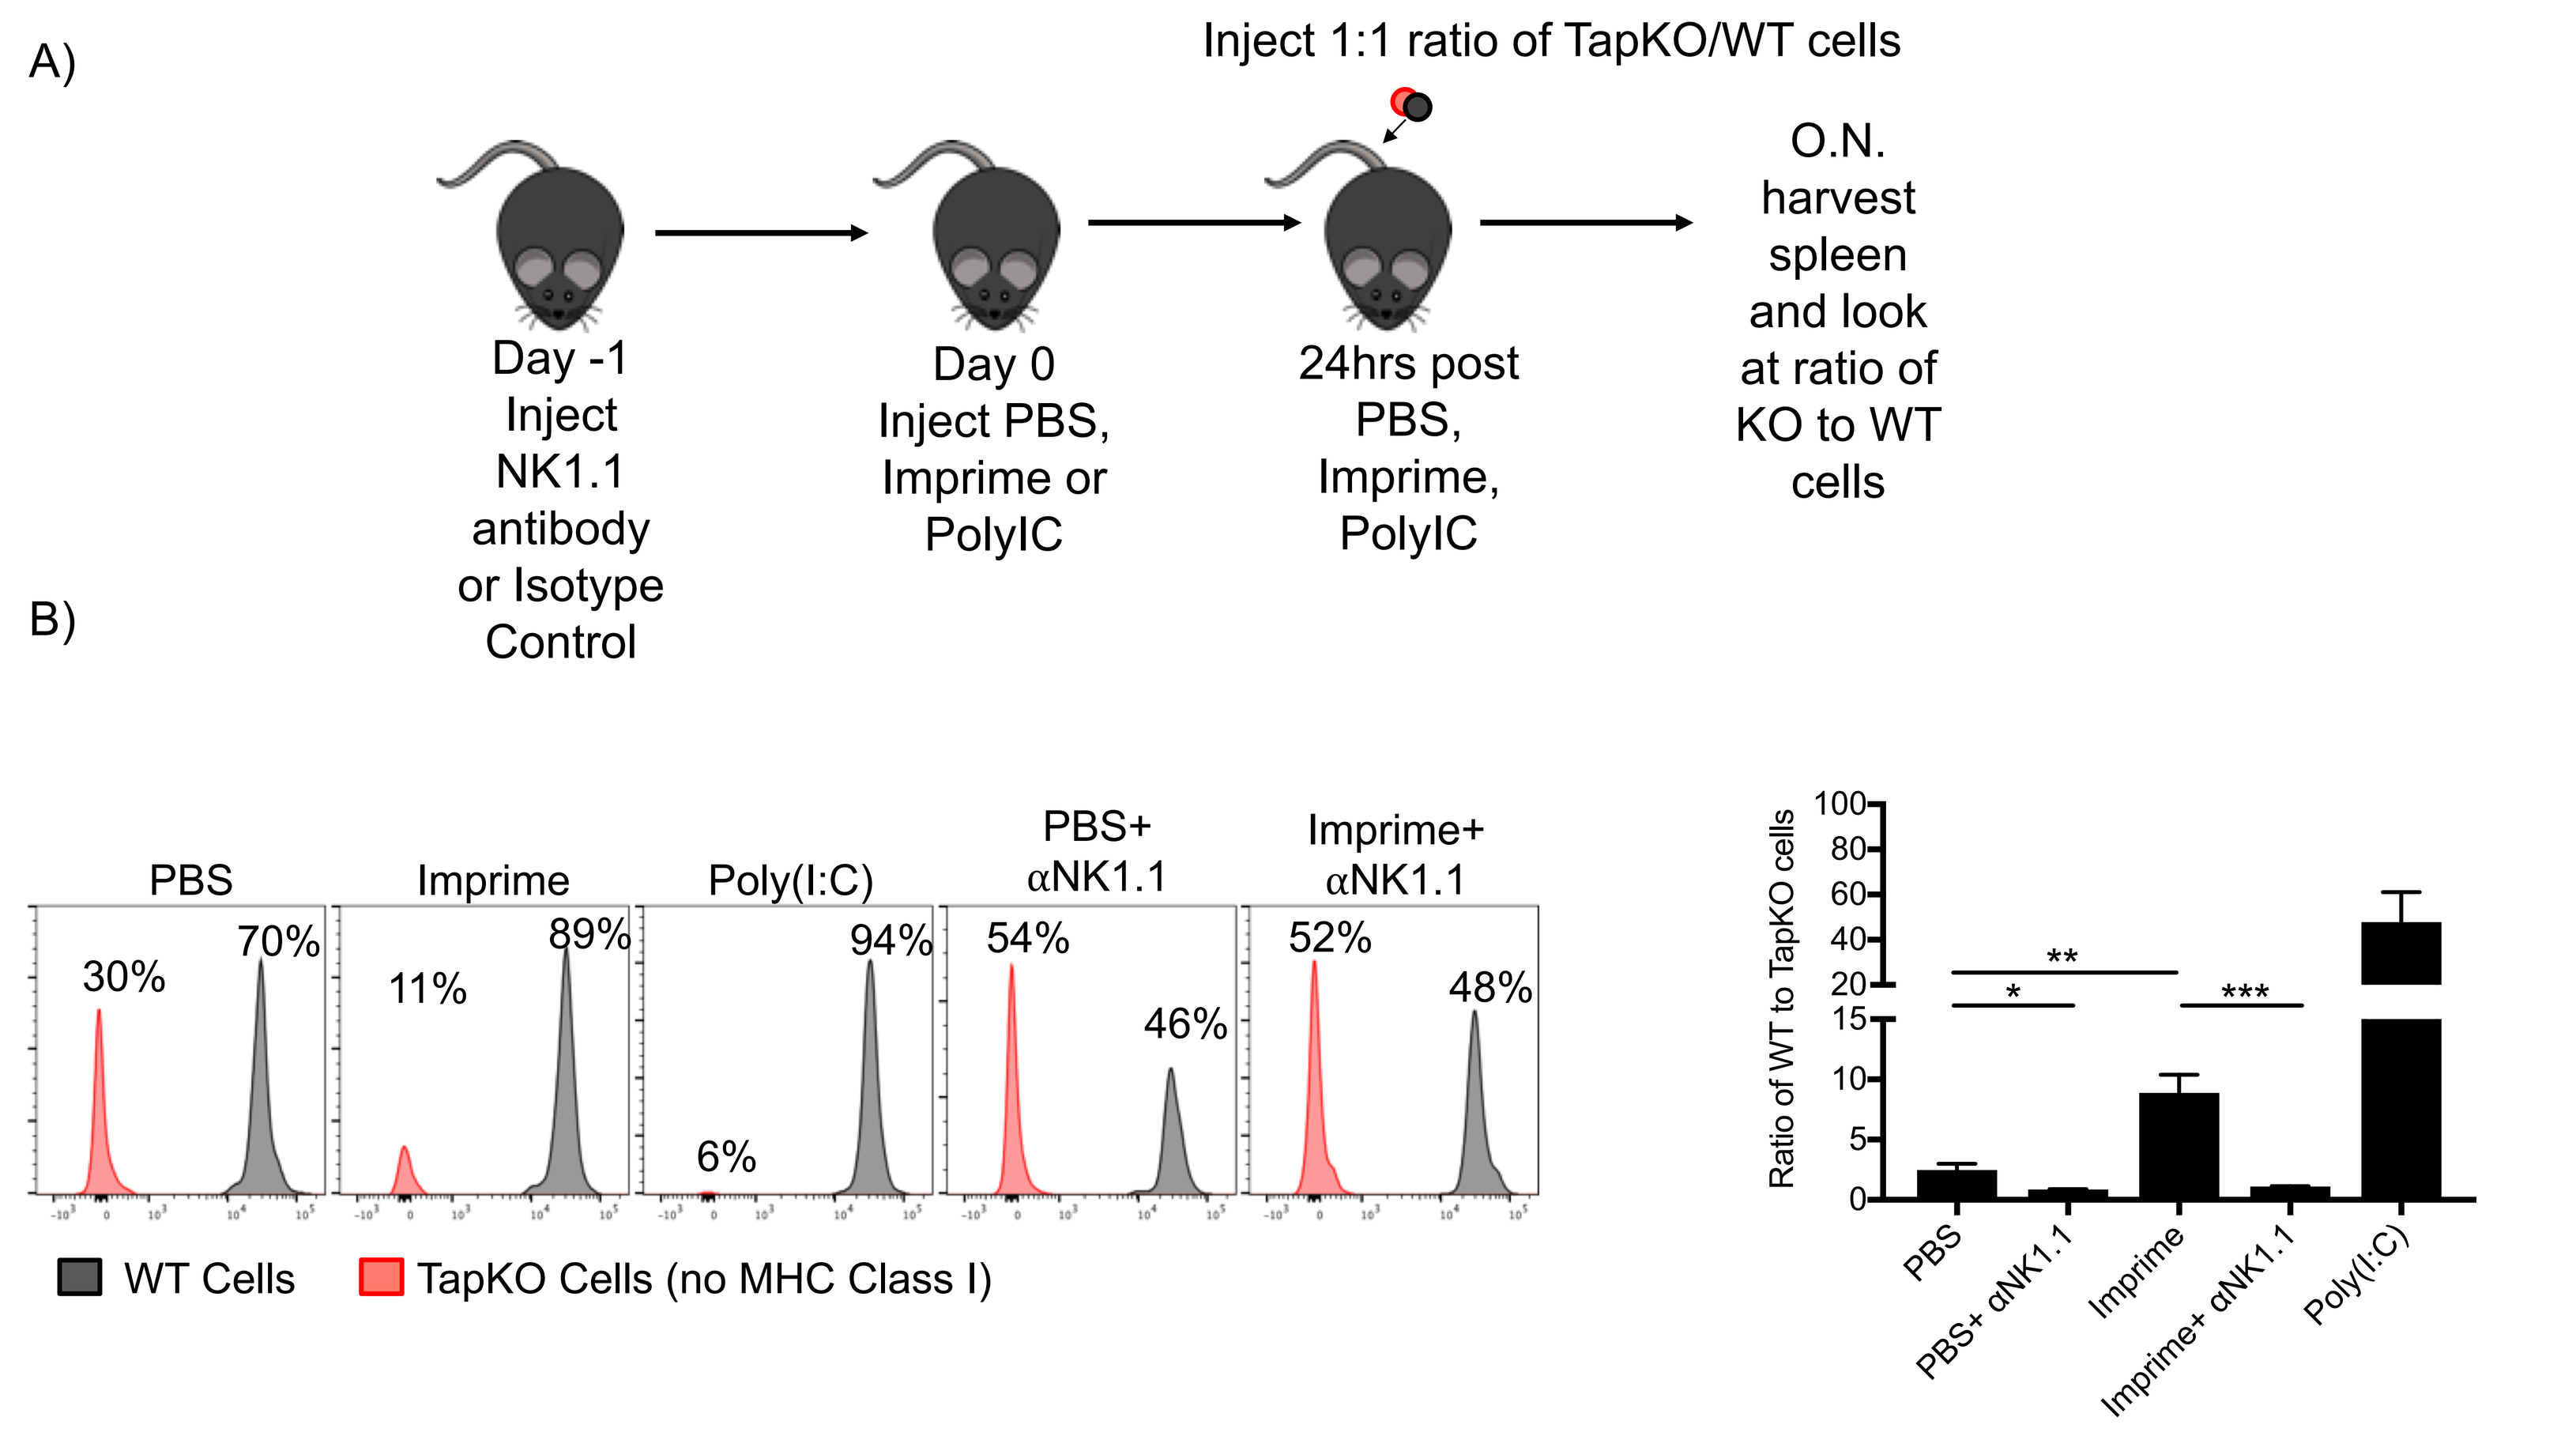
A)
Inject 1:1 ratio of TapKO/WT cells
Day -1 Inject NK1.1 antibody or Isotype Control
Day 0 Inject PBS, Imprime or PolyIC
24hrs post PBS, Imprime, PolyIC
O.N. harvest spleen and look at ratio of KO to WT cells
B)
PBS
Imprime
Poly(I:C)
PBS+ αNK1.1
Imprime+ αNK1.1
30%
70%
11%
89%
6%
94%
54%
46%
52%
48%
WT Cells
TapKO Cells (no MHC Class I)
Ratio of WT to TapKO cells
PBS
PBS+ αNK1.1
Imprime
Imprime+ αNK1.1
Poly(I:C)

(A) C57BL/6 mice were injected i.p. with NK1.1 antibody (PK136) or isotype control. The next day, mice were treated with PBS, Imprime (1.2 mg) or PolyIC (20ug). 24hrs post treatment, mice were injected with a 1:1 ratio of labelled WT (CFSE) to TapKO (CTV) splenic cells. Recipient mice splenic cells were harvested after O.N. incubation. (B) Representative experiment depicting the significant decrease in frequency of TapKO cells in comparison to WT cells in Imprime treated mice. All decreases in TAPKO frequency were dependent on NK1.1 expressing cells.

Figure 3. Enhanced killing of NK cell targets is dependent on phagocytic cells, type I IFN, and Dectin-1

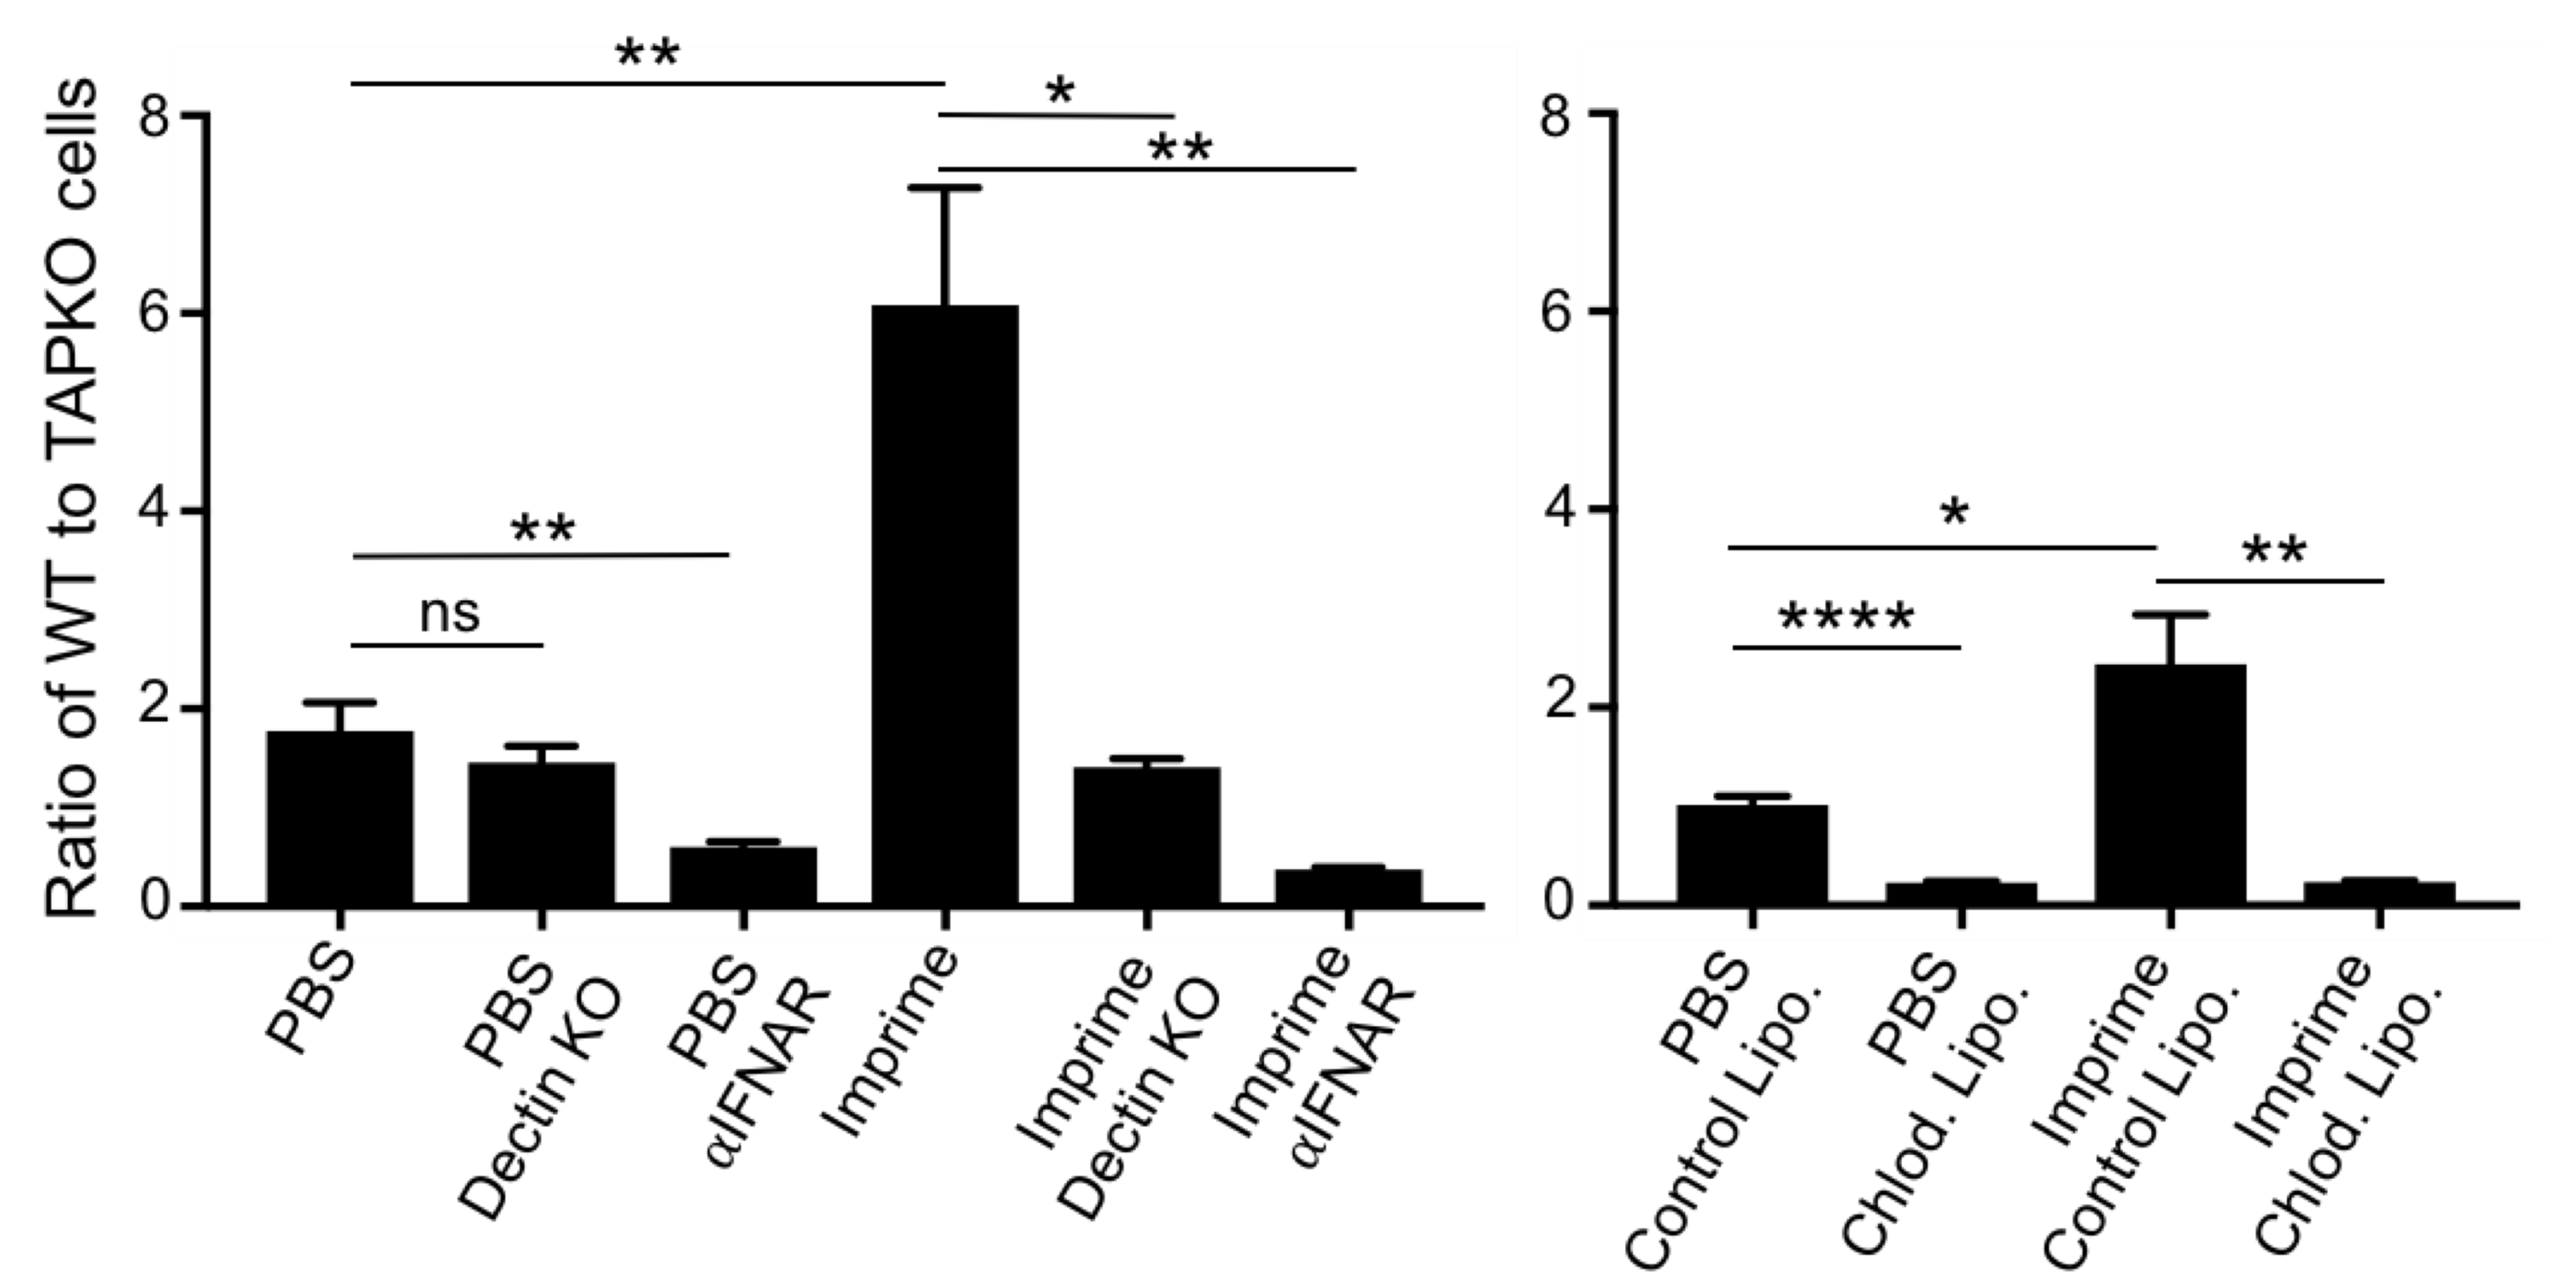
Ratio of WT to TAPKO cells
PBS
PBS Dectin KO
PBS αIFNAR
Imprime
Imprime Dectin KO
Imprime αIFNAR
PBS Control Lipo.
PBS Chlod. Lipo.
Imprime Control Lipo.
Imprime Chlod. Lipo.

(A) C57BL/6 mice were injected i.p. or i.v. with IFNAR antibody, clodronate liposomes or appropriate controls. Dectin-1 KO mice were used as recipients where indicated. As in Fig 2, the next day, mice were treated with PBS or Imprime (1.2 mg). 24hrs post treatment, mice were injected with a 1:1 ratio of labelled WT (CFSE) to TapKO (CTV) splenic cells. Recipient mice splenic cells were harvested after O.N. incubation. Graph shows N of 3-5 mice per group.

Figure 4. Imprime PGG treatment results in increased cytokines that drive enhanced NK activation and functional phenotype

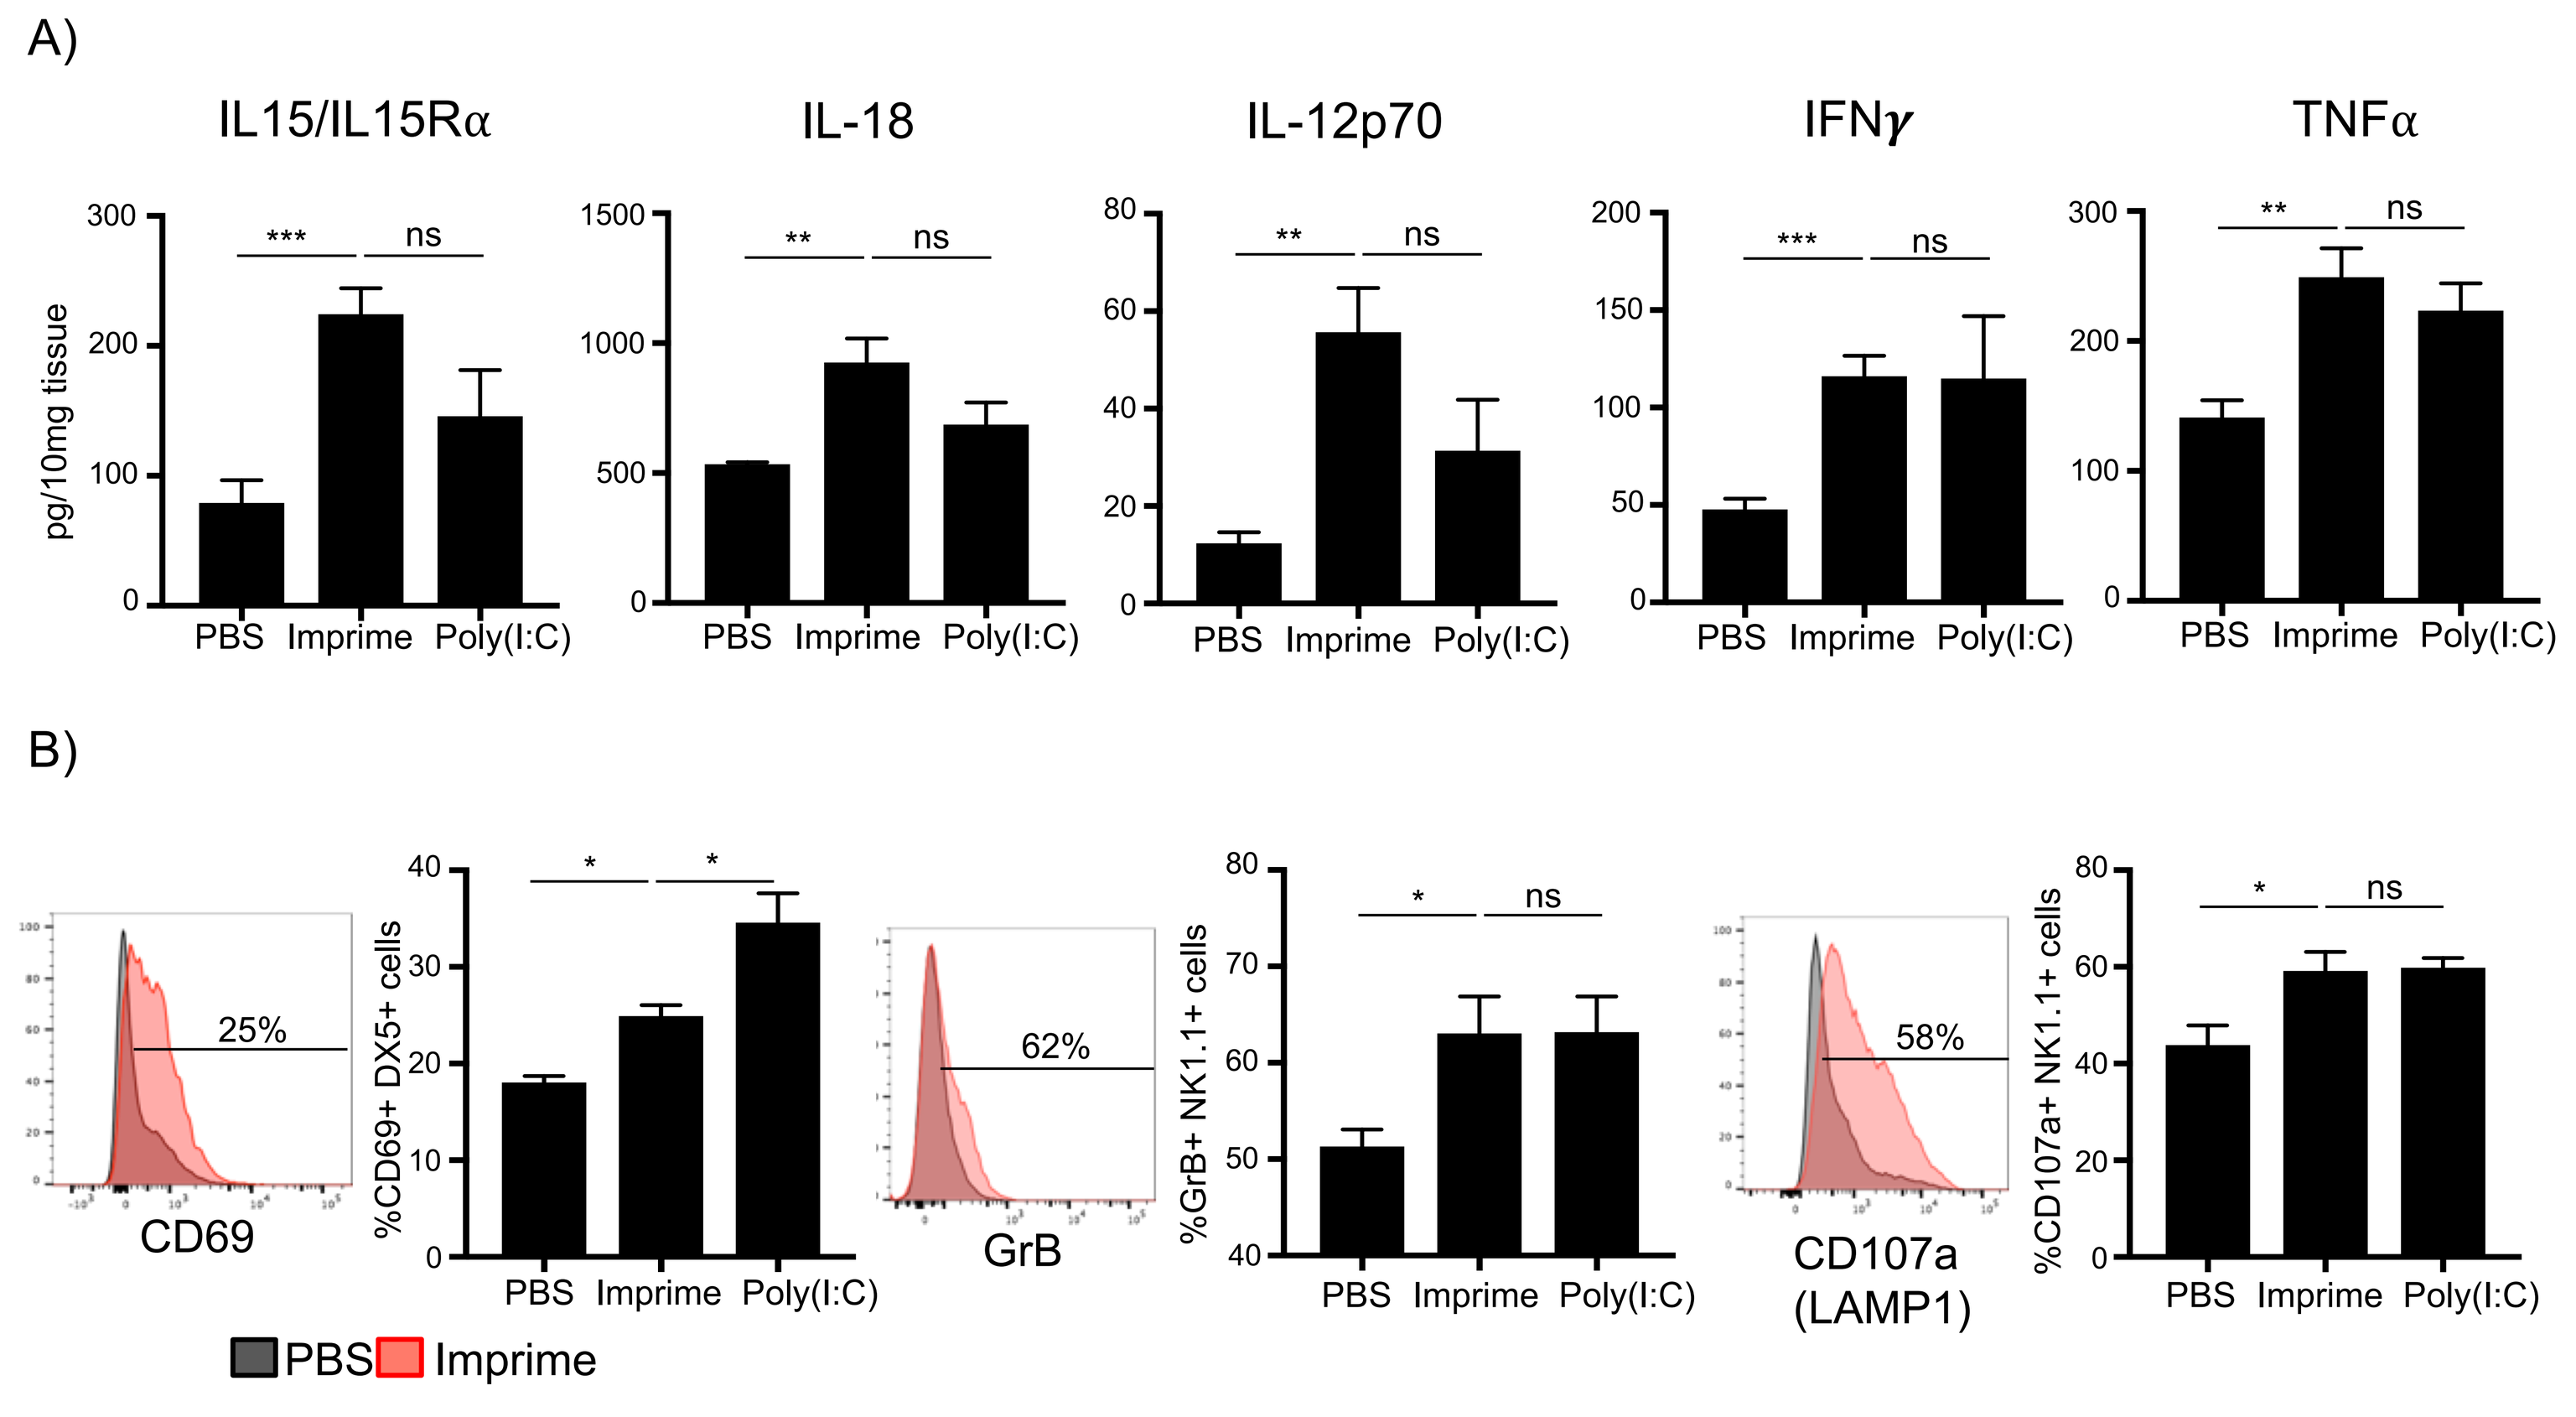
A)
IL15/IL15Rα
IL-18
IL-12p70
IFNγ
TNFα
pg/10mg tissue
PBS
Imprime
Poly(I:C)
B)
CD69
25%
%CD69+ DX5+ cells
GrB
62%
%GrB+ NK1.1+ cells
CD107a (LAMP1)
58%
%CD107a+ NK1.1+ cells
PBS
Imprime

C57BL/6 mice were treated as in Fig 2 and 3. Recipient mice lymph node and splenic cells were harvested after O.N. incubation. (A) Lysates were generated from LNs, and samples were analyzed on a Luminex cytokine 36-plex. *n* = 5 mice and data are from a single experiment. (B) Splenic cell lysates were stained with indicated antibodies and analyzed via Flow Cytometry. Percent expression of activation markers on NK1.1+, CD3-, CD45+ live cells.

Figure 5. Enhancement of IL15 occurs on Ly6c hi monocytes after Imprime PGG treatment

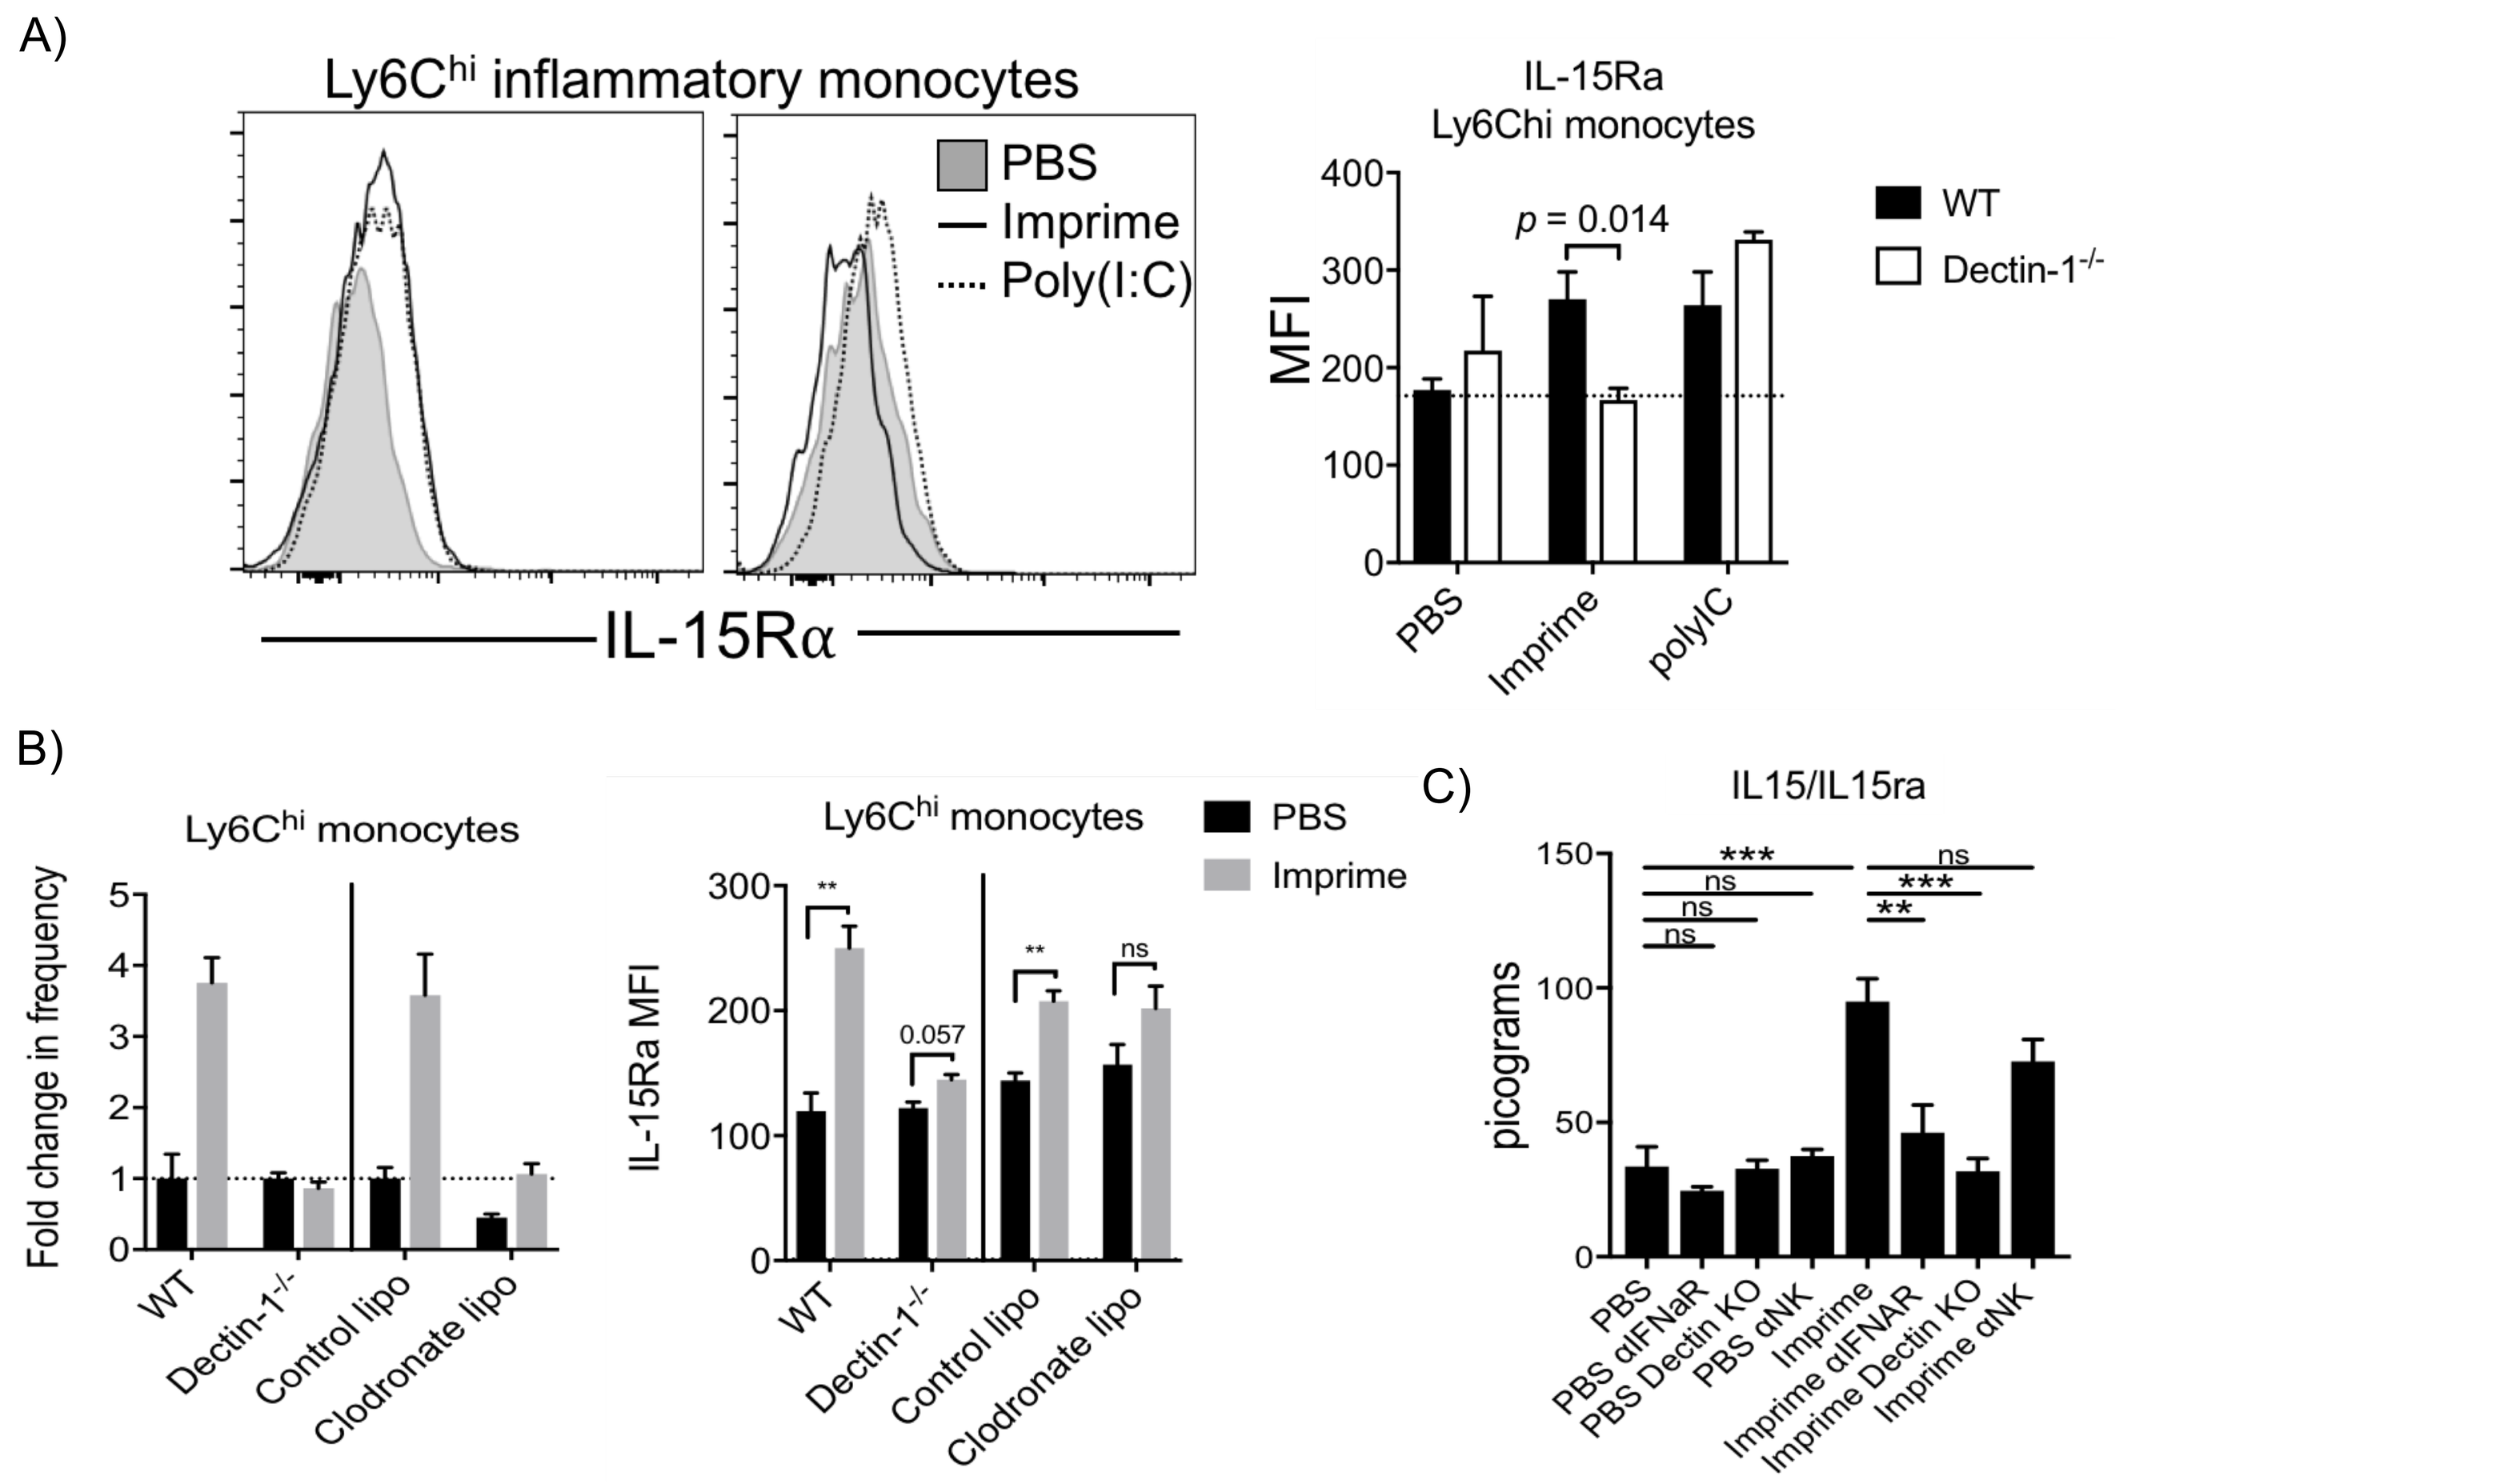
A)
Ly6Chi inflammatory monocytes
PBS
Imprime
Poly(I:C)
IL-15Rα
IL-15Ra Ly6Chi monocytes
MFI
p = 0.014
WT
Dectin-1-/-
PBS
Imprime
poly/C
B)
Ly6Chi monocytes
Fold change in frequency
IL-15Ra MFI
PBS
Imprime
WT
Dectin-1-/-
Control lipo
Clodronate lipo
0.057
C)
IL15/IL15ra
picograms
PBS
PBS αIFNaR
PBS Dectin KO
PBS αNK
Imprime
Imprime αIFNAR
Imprime Dectin KO
Imprime αNK

Imprime-treated animals express higher levels of Dectin 1 dependent IL-15Rα on Ly6Chi monocytes. Lymph nodes were harvested from C57BL/6 or Dectin KO mice after undergoing the *in-vivo* NK killing assay. Lysates were stained with IL15Rα and markers for various myeloid subsets. (A and B) Graphs depicts median fluorescent intensity of IL-15Rα on- and frequency of- Ly6Chi monocytes.(C) Luminex cytokine 36-plex on LN lysates.

Conclusions

Proposed model of Imprime PGG mediated enhancement of NK killing

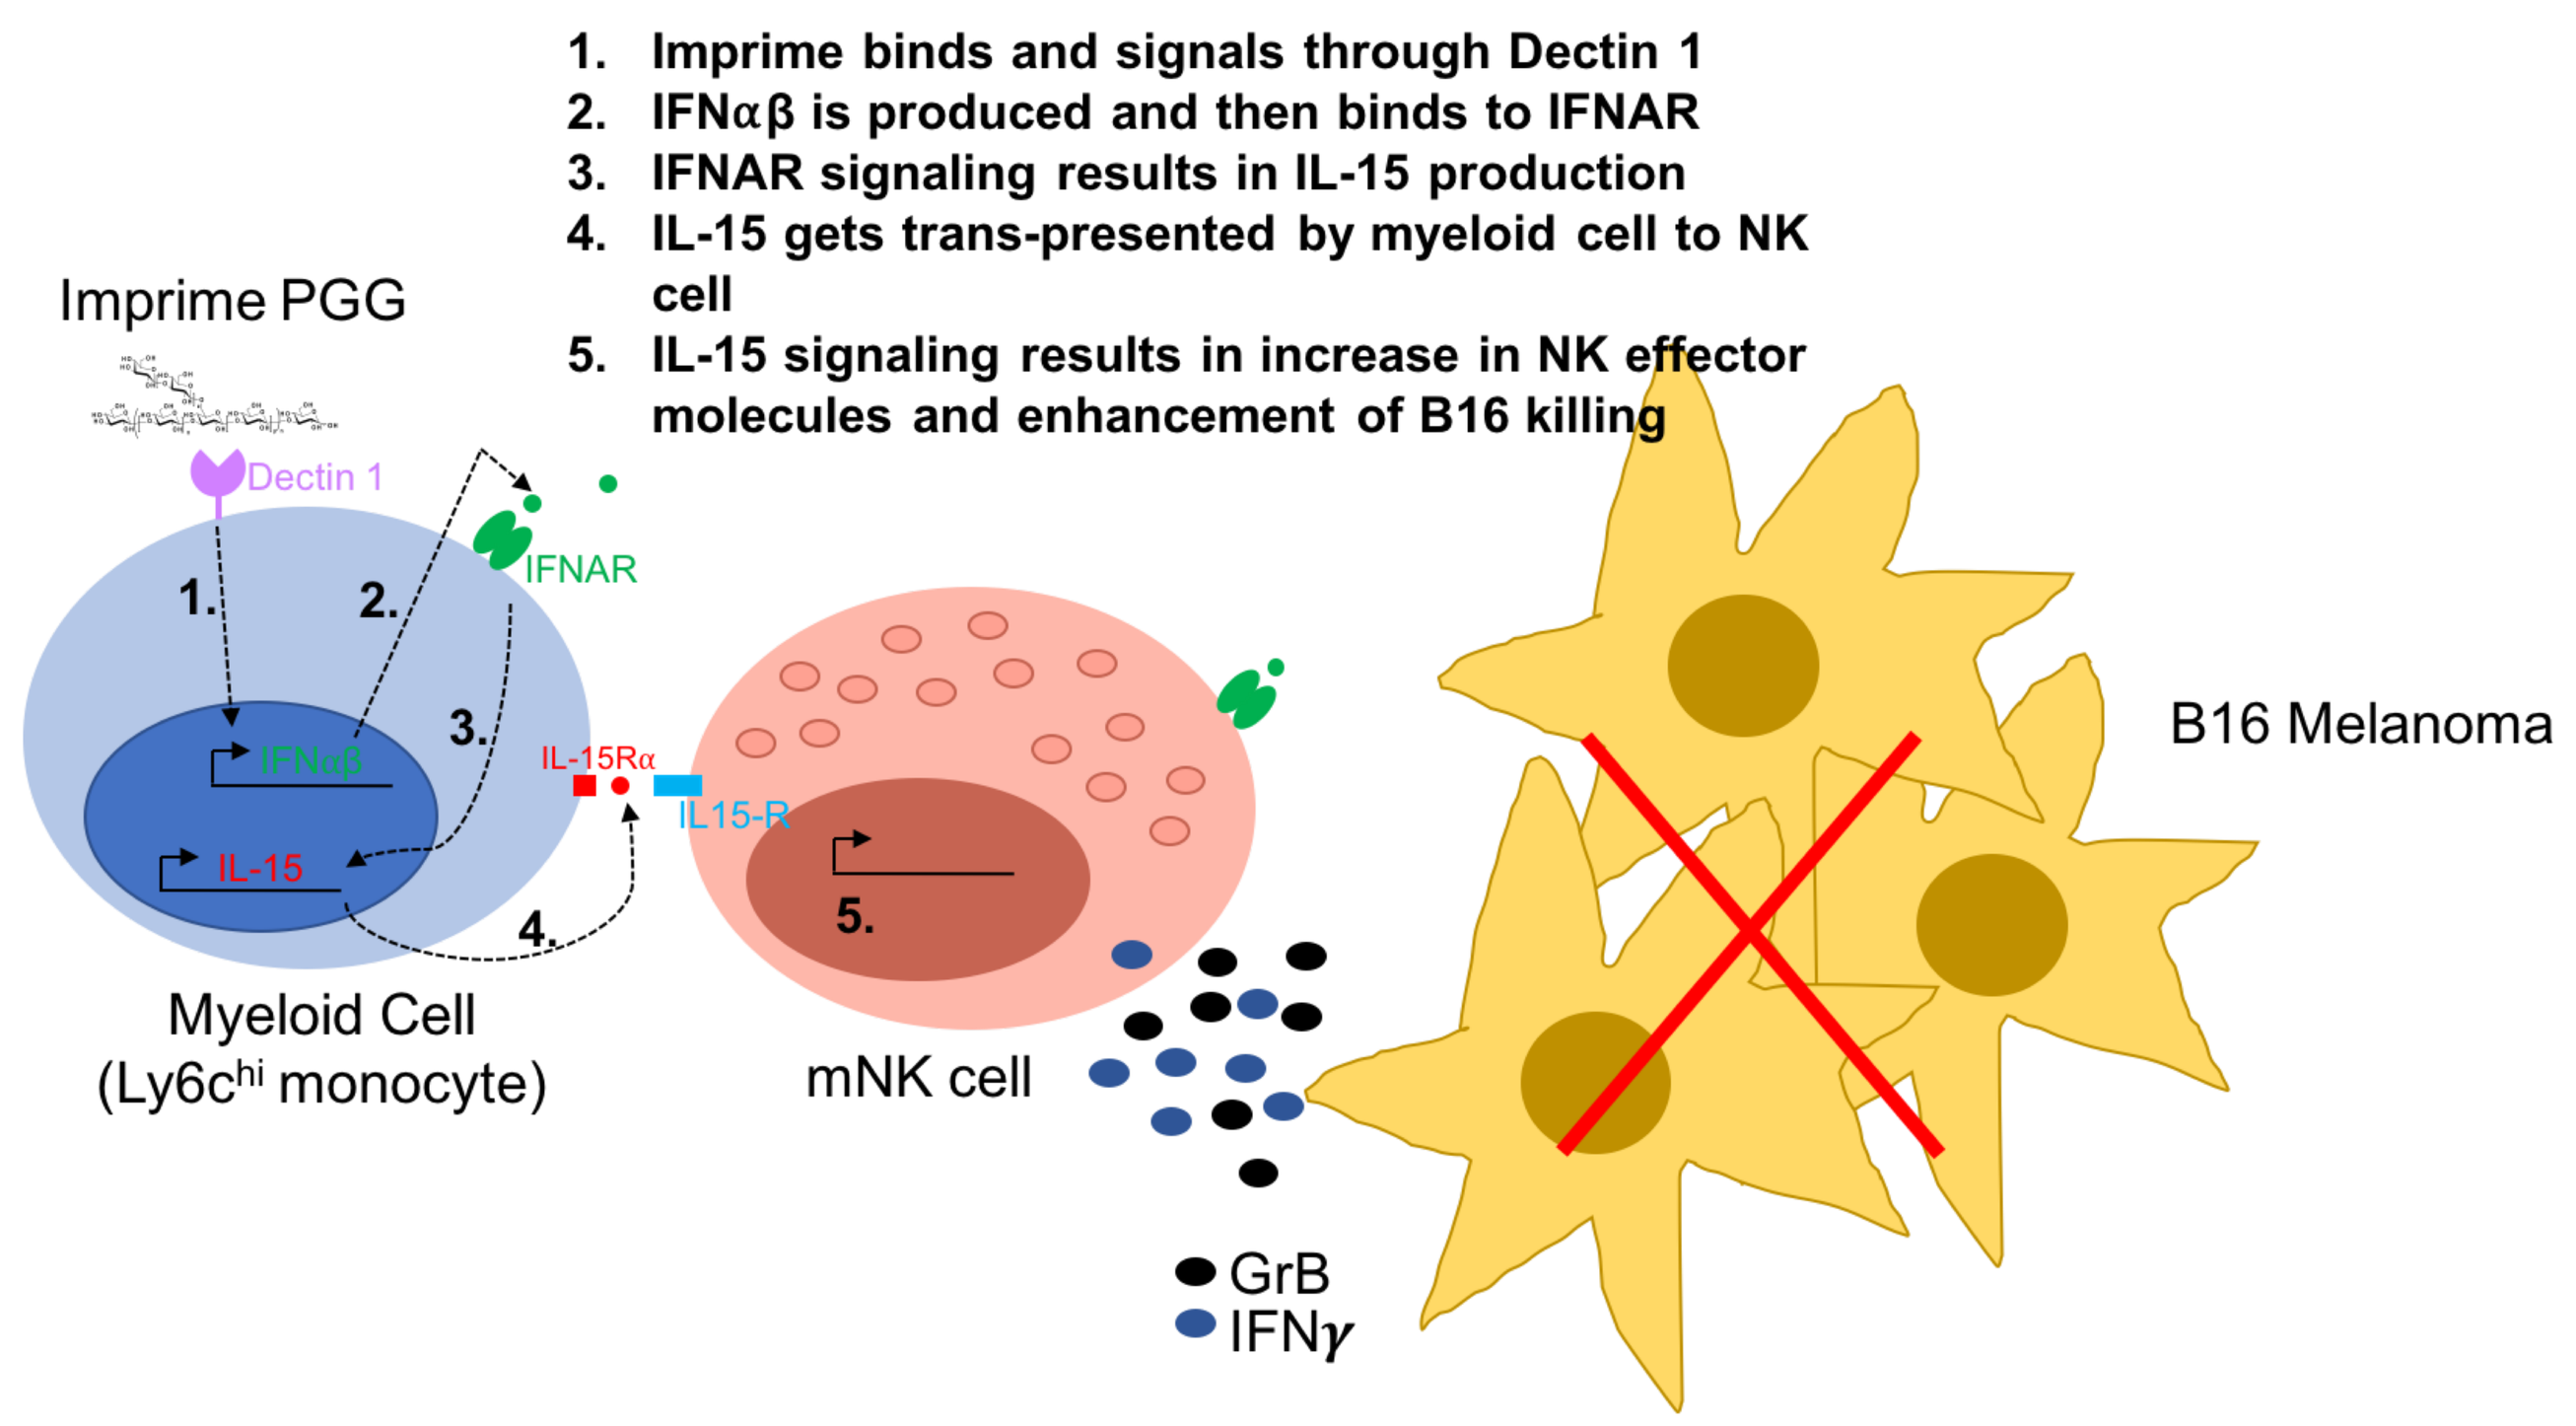
1. Imprime binds and signals through Dectin 1
2. IFNαβ is produced and then binds to IFNAR
3. IFNAR signaling results in IL-15 production
4. IL-15 gets trans-presented by myeloid cell to NK cell
5. IL-15 signaling results in increase in NK effector molecules and enhancement of B16 killing
Imprime PGG
Dectin 1
IFNAR
IL-15Rα
IL-15-R
IL-15
Myeloid Cell (Ly6chi monocyte)
mNK cell
B16 Melanoma
GrB
IFNγ

- The combination of Imprime and TA99 enhances NK mediated tumor killing/ADCC in metastatic melanoma.
- Imprime treatment results in a significant increase in NK activation and functional markers as well as increased killing of MHC Class I negative cells.

BIOTHERA
pharmaceuticals