SITC 2019
P859

Association of Immunopharmacodynamic Responses of Imprime PGG Plus Pembrolizumab with Clinical Benefit in Metastatic Triple Negative Breast Cancer (TNBC) Subjects Failing Front-line Chemotherapy

Nadine R. Ottoson[1], Anissa SH Chan[1], Xiaohong Qiu[1], Ben Harrison[1], Richard Walsh, Paulette Mattson[1], Michele Gargano[1], Joanna Cox[1], Michael Chisamore[2], Mark Uhlik[1], Jeremy R. Graff[1], and Nandita Bose[1].
[1]Biothera Pharmaceuticals, Inc., Eagan MN, 55121
[2]Merck & Co., Inc., Kenilworth, NJ, USA

Abstract

Background: Checkpoint inhibitor (CPI) monotherapies, including pembrolizumab (KEYTRUDA®, pembro), avelumab and atezolizumab have demonstrated modest clinical benefit in chemotherapy-relapsed/refractory TNBC patients (pts) with ~5-10% response rate, median overall survival (mOS) of 7-9 months, and 1 year OS ~37-40%. TNBC, although more immunogenic relative to the other breast cancer subtypes, is also the most heterogenous, resulting in substantial variability in immune responses. There is a dire need for immunotherapeutic agents that could consistently induce anti-cancer immune responses.

Methods: The primary analyses of our Phase 2 study (NCT02981303; collaboration with Merck & Co., Inc.) in 44 (intent-to-treat) chemotherapy-refractory/relapsed TNBC pts treated with Imprime PGG (Imprime), a novel yeast derived, Dectin-1 agonist β-glucan PAMP in combination with pembro has shown enhanced disease control rate (25%, N= 11;1 CR, 6 PR and 4 SD>24 weeks), 12-month OS rate (57.3%) and increased mOS (16.6 months) vs the respective endpoints in Keynote086 pts treated with pembro alone. As part of exploratory translational objectives, peripheral blood from pts receiving the combination in 3-week cycles were collected at various time points. Results from serum and cellular immunopharacodynamic (IPD) evaluations from 41 pts are presented.

Results: Peak levels of serum circulating immune complexes (~3 to 22-fold) and complement protein SC5b-9 (~1.4 to 41-fold) in stage 1 pts provided evidence for Imprime-anti beta glucan antibody immune complex formation. A significant increase in the frequency of HLA-DR+ myeloid cells was observed in the overall population (up to 7.4-fold). In pts showing disease control (N=11), a significant increase in complement function (CH50, ~0.8-4 fold range), select chemokines such as MCP-1 production (up to 1000-fold), CD86 expression on monocyte (~0.5-6-fold) and DC subsets (~0.8-11-fold), and increased frequency of Ki-67+, HLA-DR/PD-1+ CD8 T cells (~0.4-14-fold) was observed. Some IPD responses were associated with the 12-month landmark OS analyses. Additionally, enhanced mPFS (HR 0.51; p=0.03) and mOS (HR 0.13; p=0.0013) was observed in 18 pts with >1.25-fold increase in CD86 expression on classical monocytes. Greater than 2-fold increase in the frequency of Ki-67+, HLA-DR/PD-1+ CD8 T cells in 16 pts was also associated with enhanced mPFS (HR 0.395; p=0.01) and mOS (HR 0.183; p=0.008). Additionally, the gene expression profile of these IPD-responders were similar to the RECIST responders with >2-fold upregulation of several genes including IFNg, CD83, GZMA, GZMK, and CD3.

Conclusions: Overall, the strong association of the innate/adaptive IPD responses to the clinical responses are suggestive of interplay between the therapeutic mechanisms of Imprime and pembro combination.

Background

Imprime PGG: A Novel Dectin Receptor Agonist (a PAMP) that Activates the Innate Immune System

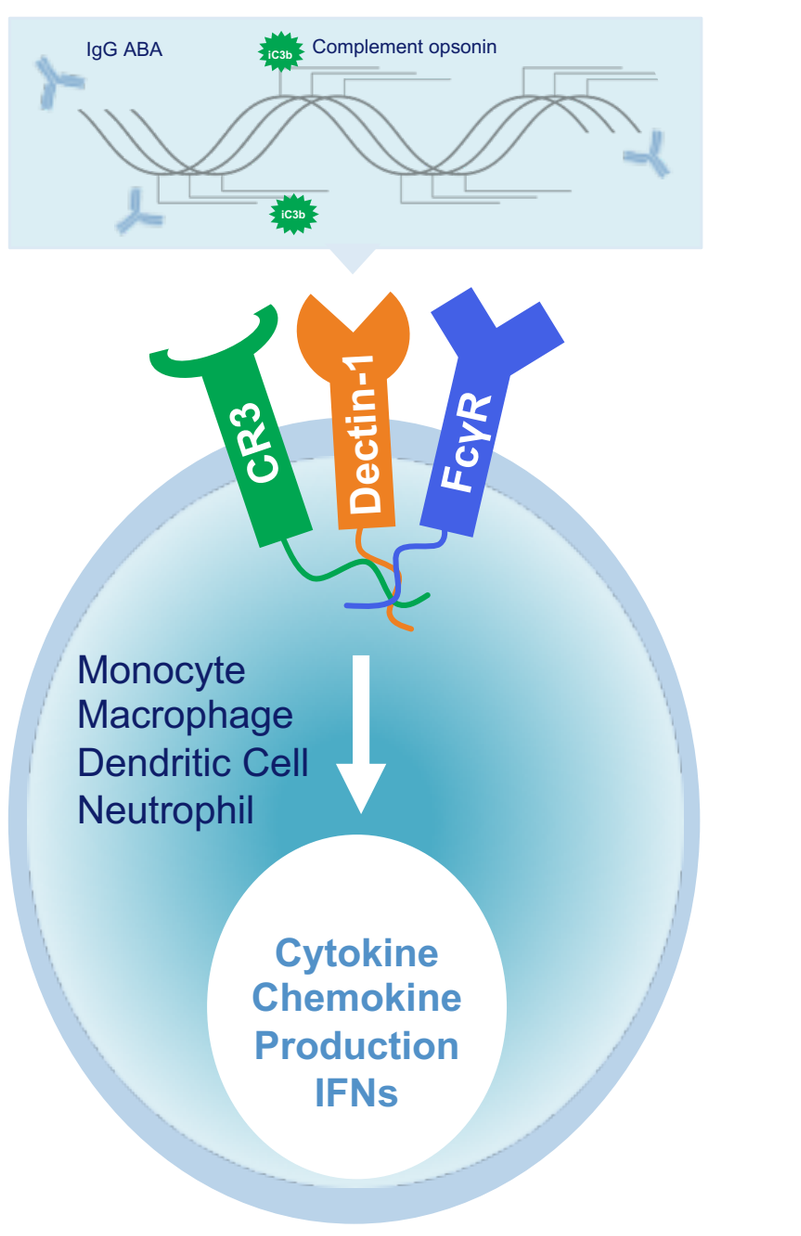
IgG ABA
Complement opsonin
CR3
Dectin-1
FcγR
Monocyte
Macrophage
Dendritic Cell
Neutrophil
Cytokine
Chemokine
Production
IFNs

Imprime PGG binds the Dectin Receptor instigating an integrated anti-cancer immune response involving both innate and adaptive immunity

1. **Imprime PGG: 1,3-1,6 β-glucan isolated from *Saccharomyces***
 - A "non-self danger" signal
 - A Pathogen-Associated Molecular Pattern (PAMP)
2. **Imprime PGG forms an immune complex**
 - Anti-β glucan antibodies (ABA) (IgG2a)
 - Patient selection biomarker
 - Complement fragment (iC3b)
3. **Immune Complex binds Dectin and Co-Receptors**
 - **Immune complex is "active" drug**
1. **Innate Immune Cell Activation**
 - Promotes tumor killing by macrophages, neutrophils, and NK
 - Alleviates immunosuppression
 - Promotes shift from immunosuppressive M2 state
 - Drives infiltration of PD-L1+, CD80+ immune cells (M1)
 - Activates antigen presenting cells
 - Dendritic Cells, M1 APCs

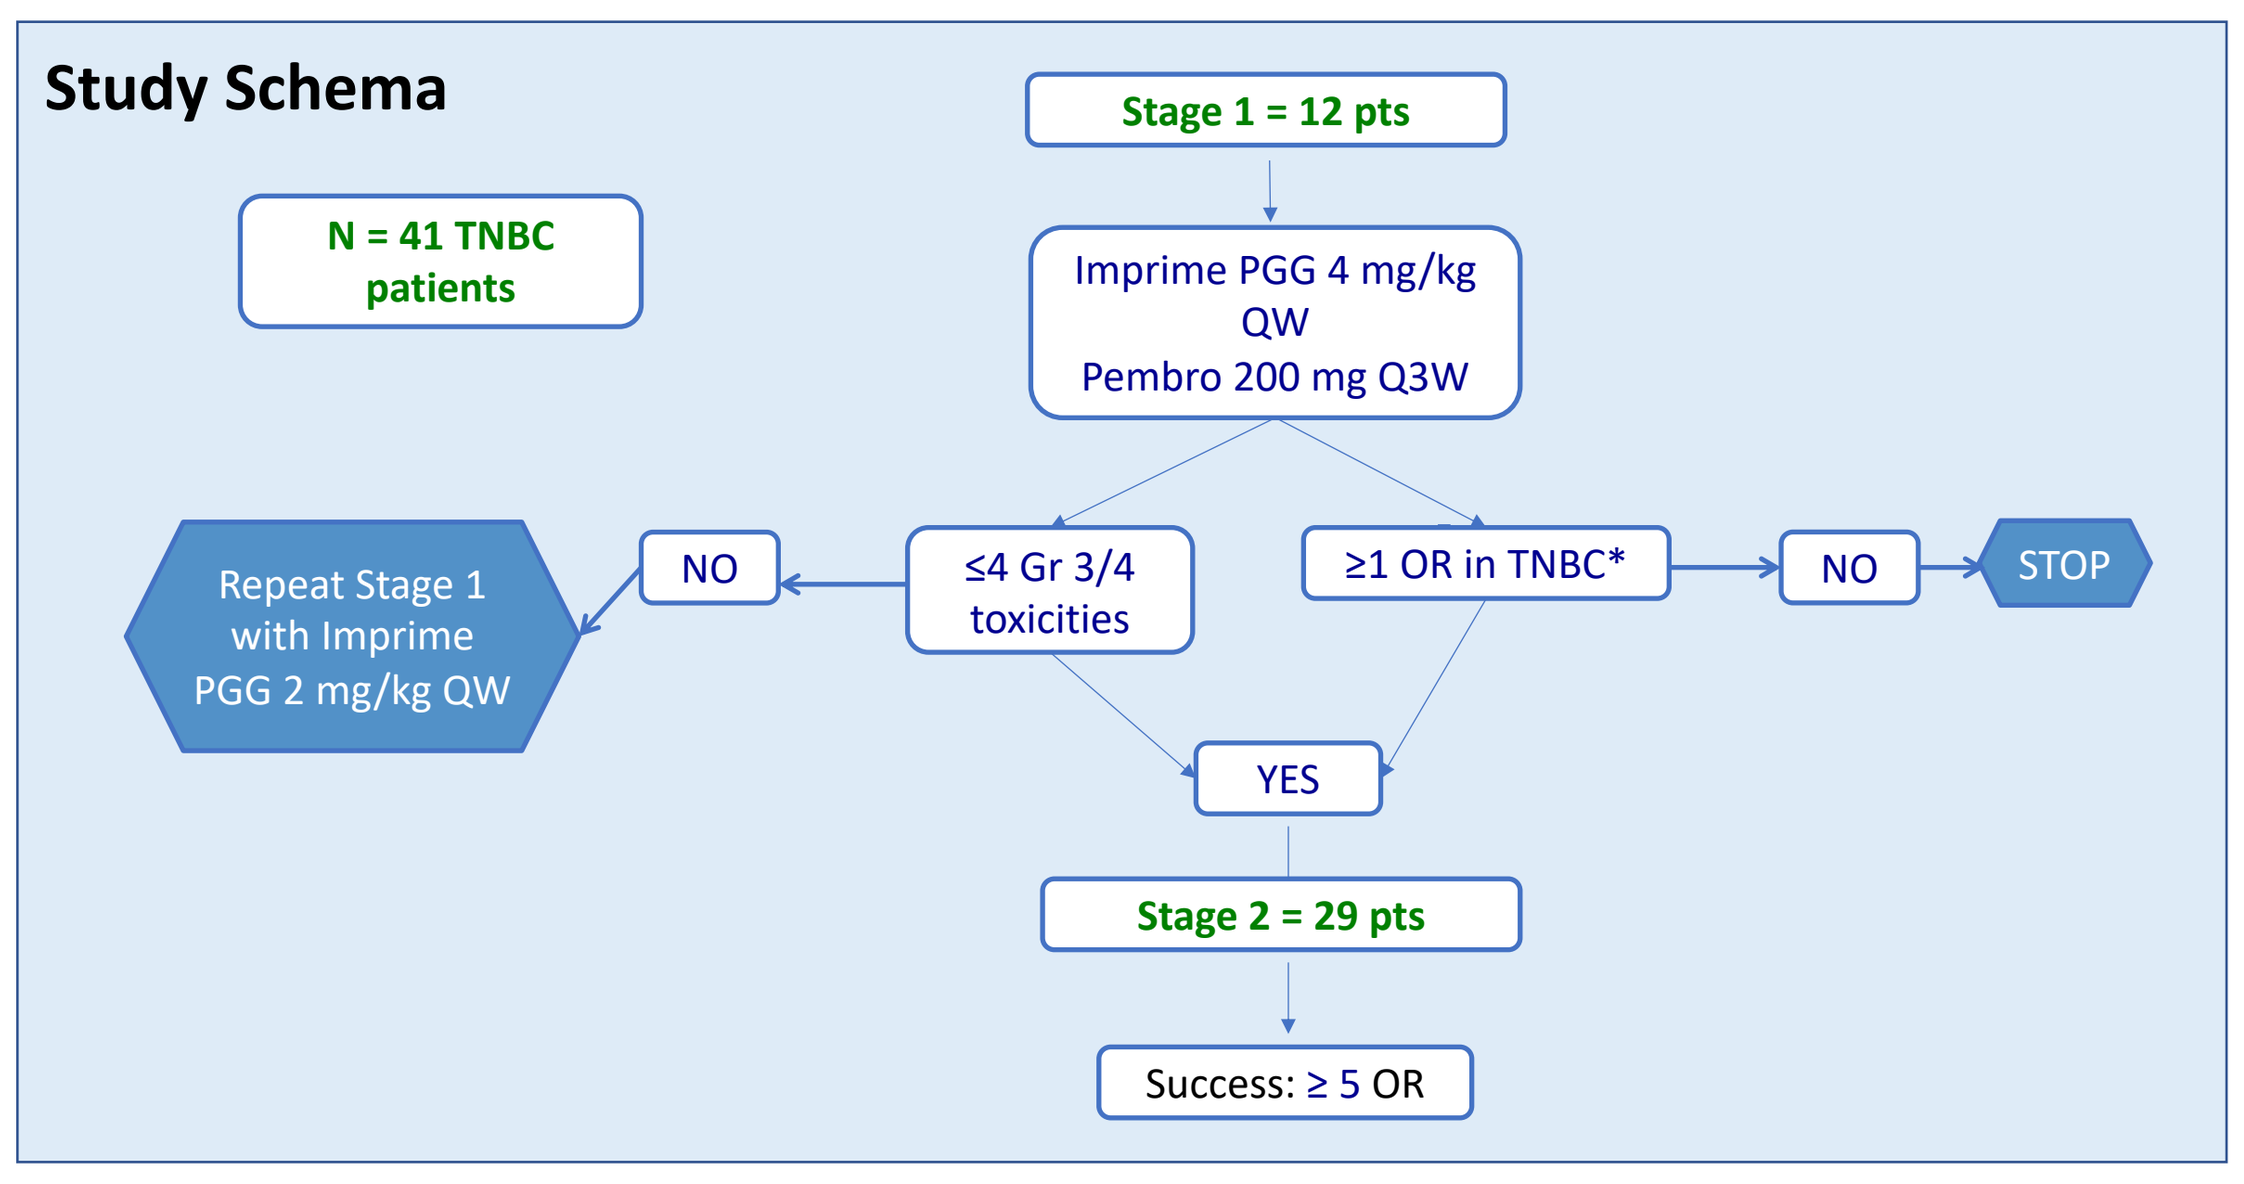
Study Schema
N = 41 TNBC patients
Stage 1 = 12 pts
Imprime PGG 4 mg/kg QW
Pembro 200 mg Q3W
≤4 Gr 3/4 toxicities
≥1 OR in TNBC*
NO
STOP
NO
Repeat Stage 1 with Imprime PGG 2 mg/kg QW
YES
Stage 2 = 29 pts
Success: ≥ 5 OR

Clinical Benefit in mTNBC: Previous CPI Monotherapy Studies and IMPRIME 1

	Bavencio[c] % (N=58)	Tecentriq[b] % (N=94)	Keytruda[a] % (N=170)	Keytruda[d] % (N=312)	IMPRIME 1 % (N=44[#])
Overall Response Rate (ORR)	5.2	6.4	5.3	9.6	**15.9**
Stable Disease (SD)	26.0	13.0	18.0	NR	**38.6**
Progressive Disease (PD)	65.0	64.0	60.6	NR	**40.9**
Disease Control Rate (DCR)					
- CR+PR+SD any time	31.2	19.4	23.3		**54.5**
- CR+PR+SD ≥ 24 weeks	NR	10.0	7.6	12.2	**25.0**
Median Overall Survival (mos)	9.2	7.3	9.0	9.9	**16.6***
Overall Survival Rate (%)					
- 6 month	NR	60.0	69.7	NR	**76.8**
- 9 month	~50.0**	44.0	50.0	NR	**69.6**
- 12 month	37.1	37.0	39.8	NR	**57.3**

CR = Confirmed Complete Responder
PR = Confirmed Partial Responder
NR = Not Reported
#- ITT population, n = 44 patients, 2 not evaluable for response

Latest IMPRIME 1 data from September 4, 2019.

* Median follow up time 16.5 months
**Estimated from reported median OS
[a] Keynote-086 Adams et al., 2018- Merck
[b] PCD4989g Emens et al., 2019- Genentech
[c] Javelin Dirix et al., 2018- Pfizer
[d] Keynote-119 Cortes et al, ESMO 2019- Merck

Treatment Promotes a Shift from the Immunosuppressive M2 State (Pink) toward the M1 State (Green)

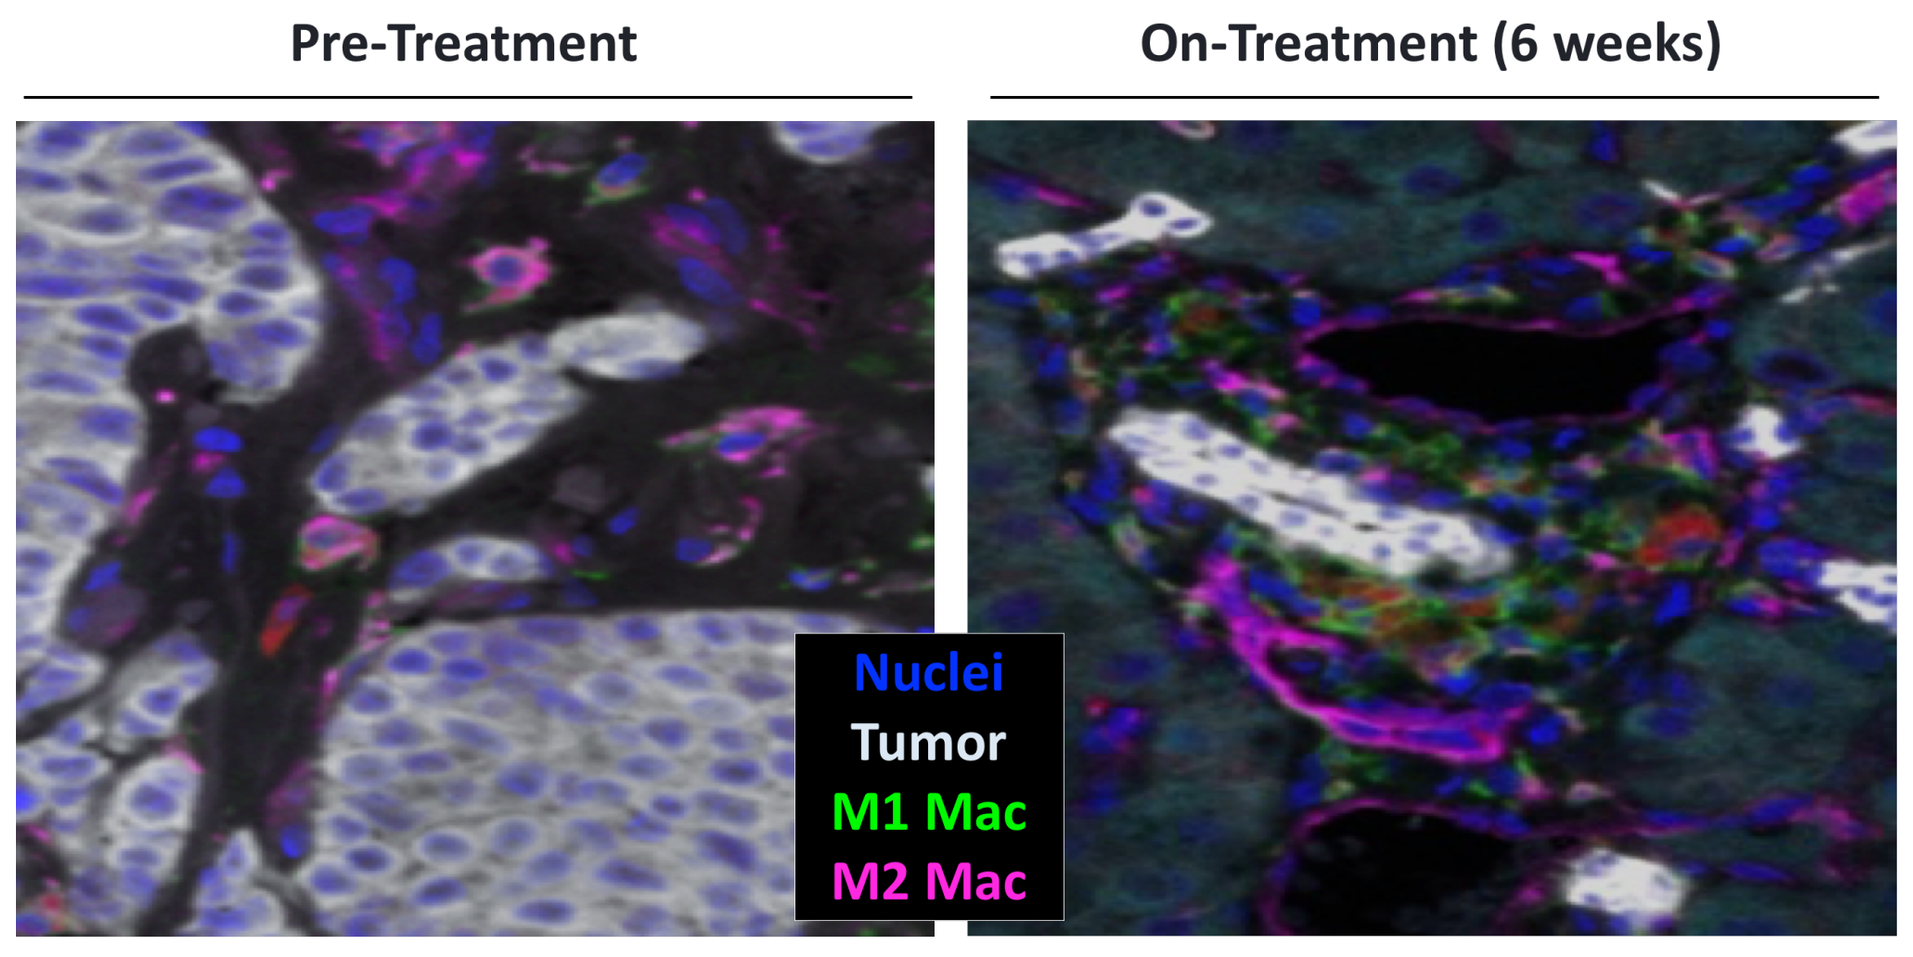
Pre-Treatment
On-Treatment (6 weeks)
Nuclei
Tumor
M1 Mac
M2 Mac

Figure 1. mTNBC patient 109128. The pre-treatment tumor sample shows continuous sheets of tumor cells with predominant M2 (CD206, pink) myeloid character. On-treatment, the tumor is evident as smaller nests of white cells and necrotic tumor cells. Especially evident surrounding the central lesion are infiltrating myeloid cells that are in the desired M1 state (CD80+). The ratio of M1:M2 cells on treatment increased ~40 fold vs pre-treatment as quantitated using the Perkin Elmer Vectra 3.0 system.

Acknowledgements

All experiments funded by Biothera Pharmaceuticals Inc. No external funding was received to support the work.

Results

Rationale for IPD Evaluations in Imprime 1: TNBC

- Pembrolizumab has been shown to increase the activation of **pre-existing exhausted T cells in highly immunogenic cancers like melanoma** (PD1+/HLA-DR+ Ki67+ CD8). TNBC is not as immunogenic as melanoma and there is less pre-existing T Cell immunity
- In TNBC, the challenge is to inspire T cell immunity. Robust innate immune activation is key to inducing anti-tumor T cell responses

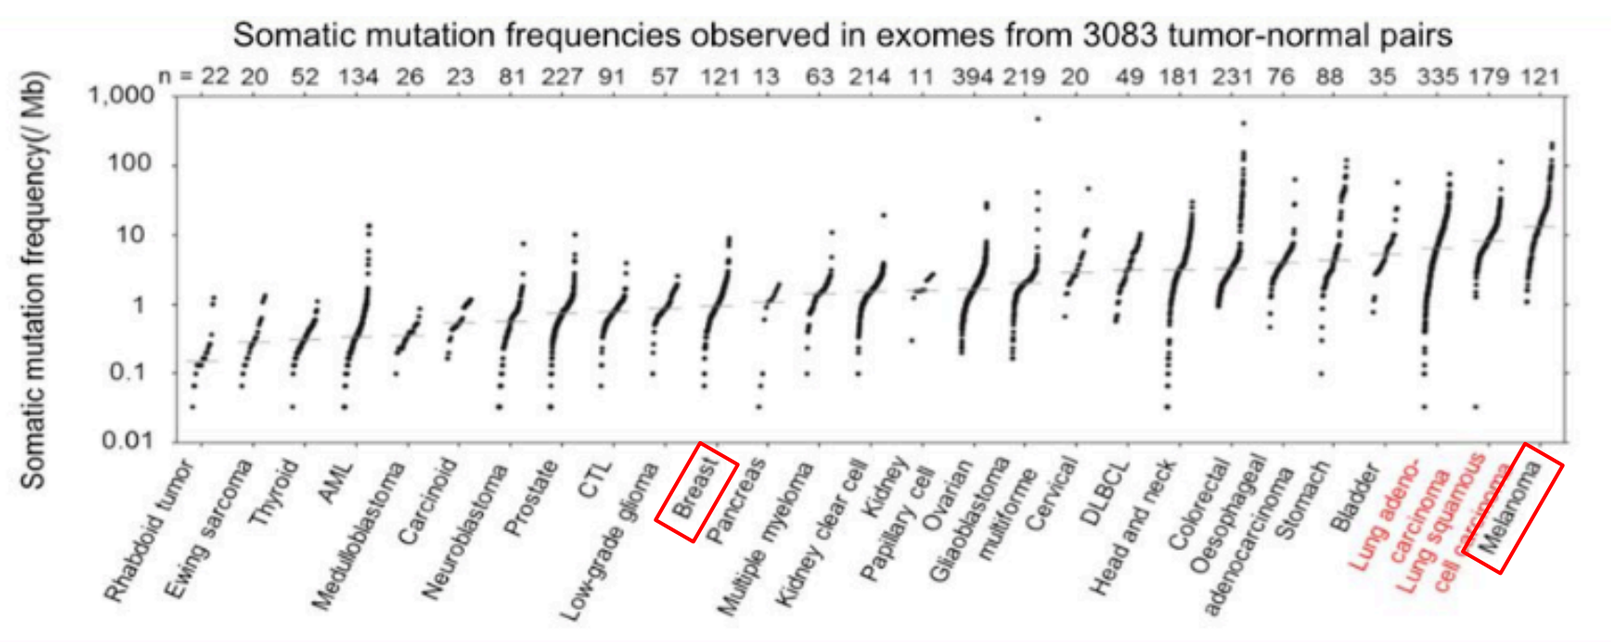
Somatic mutation frequencies observed in exomes from 3083 tumor-normal pairs
Somatic mutation frequency(/ Mb)
Breast
Melanoma

Lawrence et al., Nature 2013

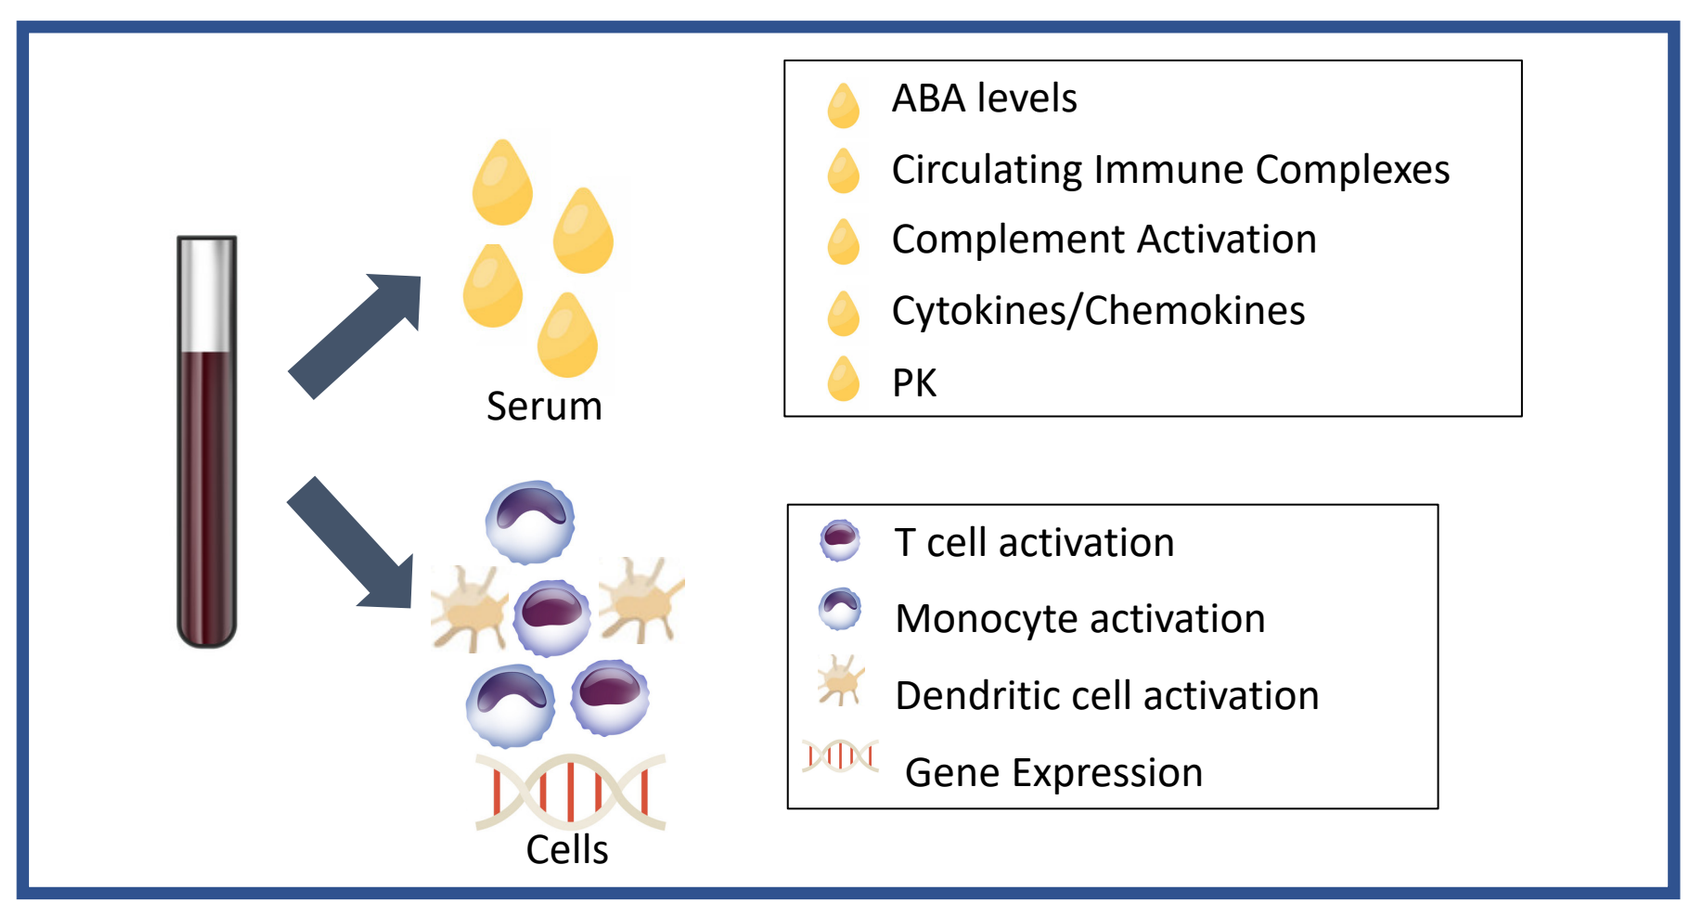
Serum
Cells
ABA levels
Circulating Immune Complexes
Complement Activation
Cytokines/Chemokines
PK
T cell activation
Monocyte activation
Dendritic cell activation
Gene Expression

Serum and Cellular IPD Evaluations in Imprime 1: TNBC

In vivo Evidence of Immune Complex Formation, Complement Activation and Cytokine Production

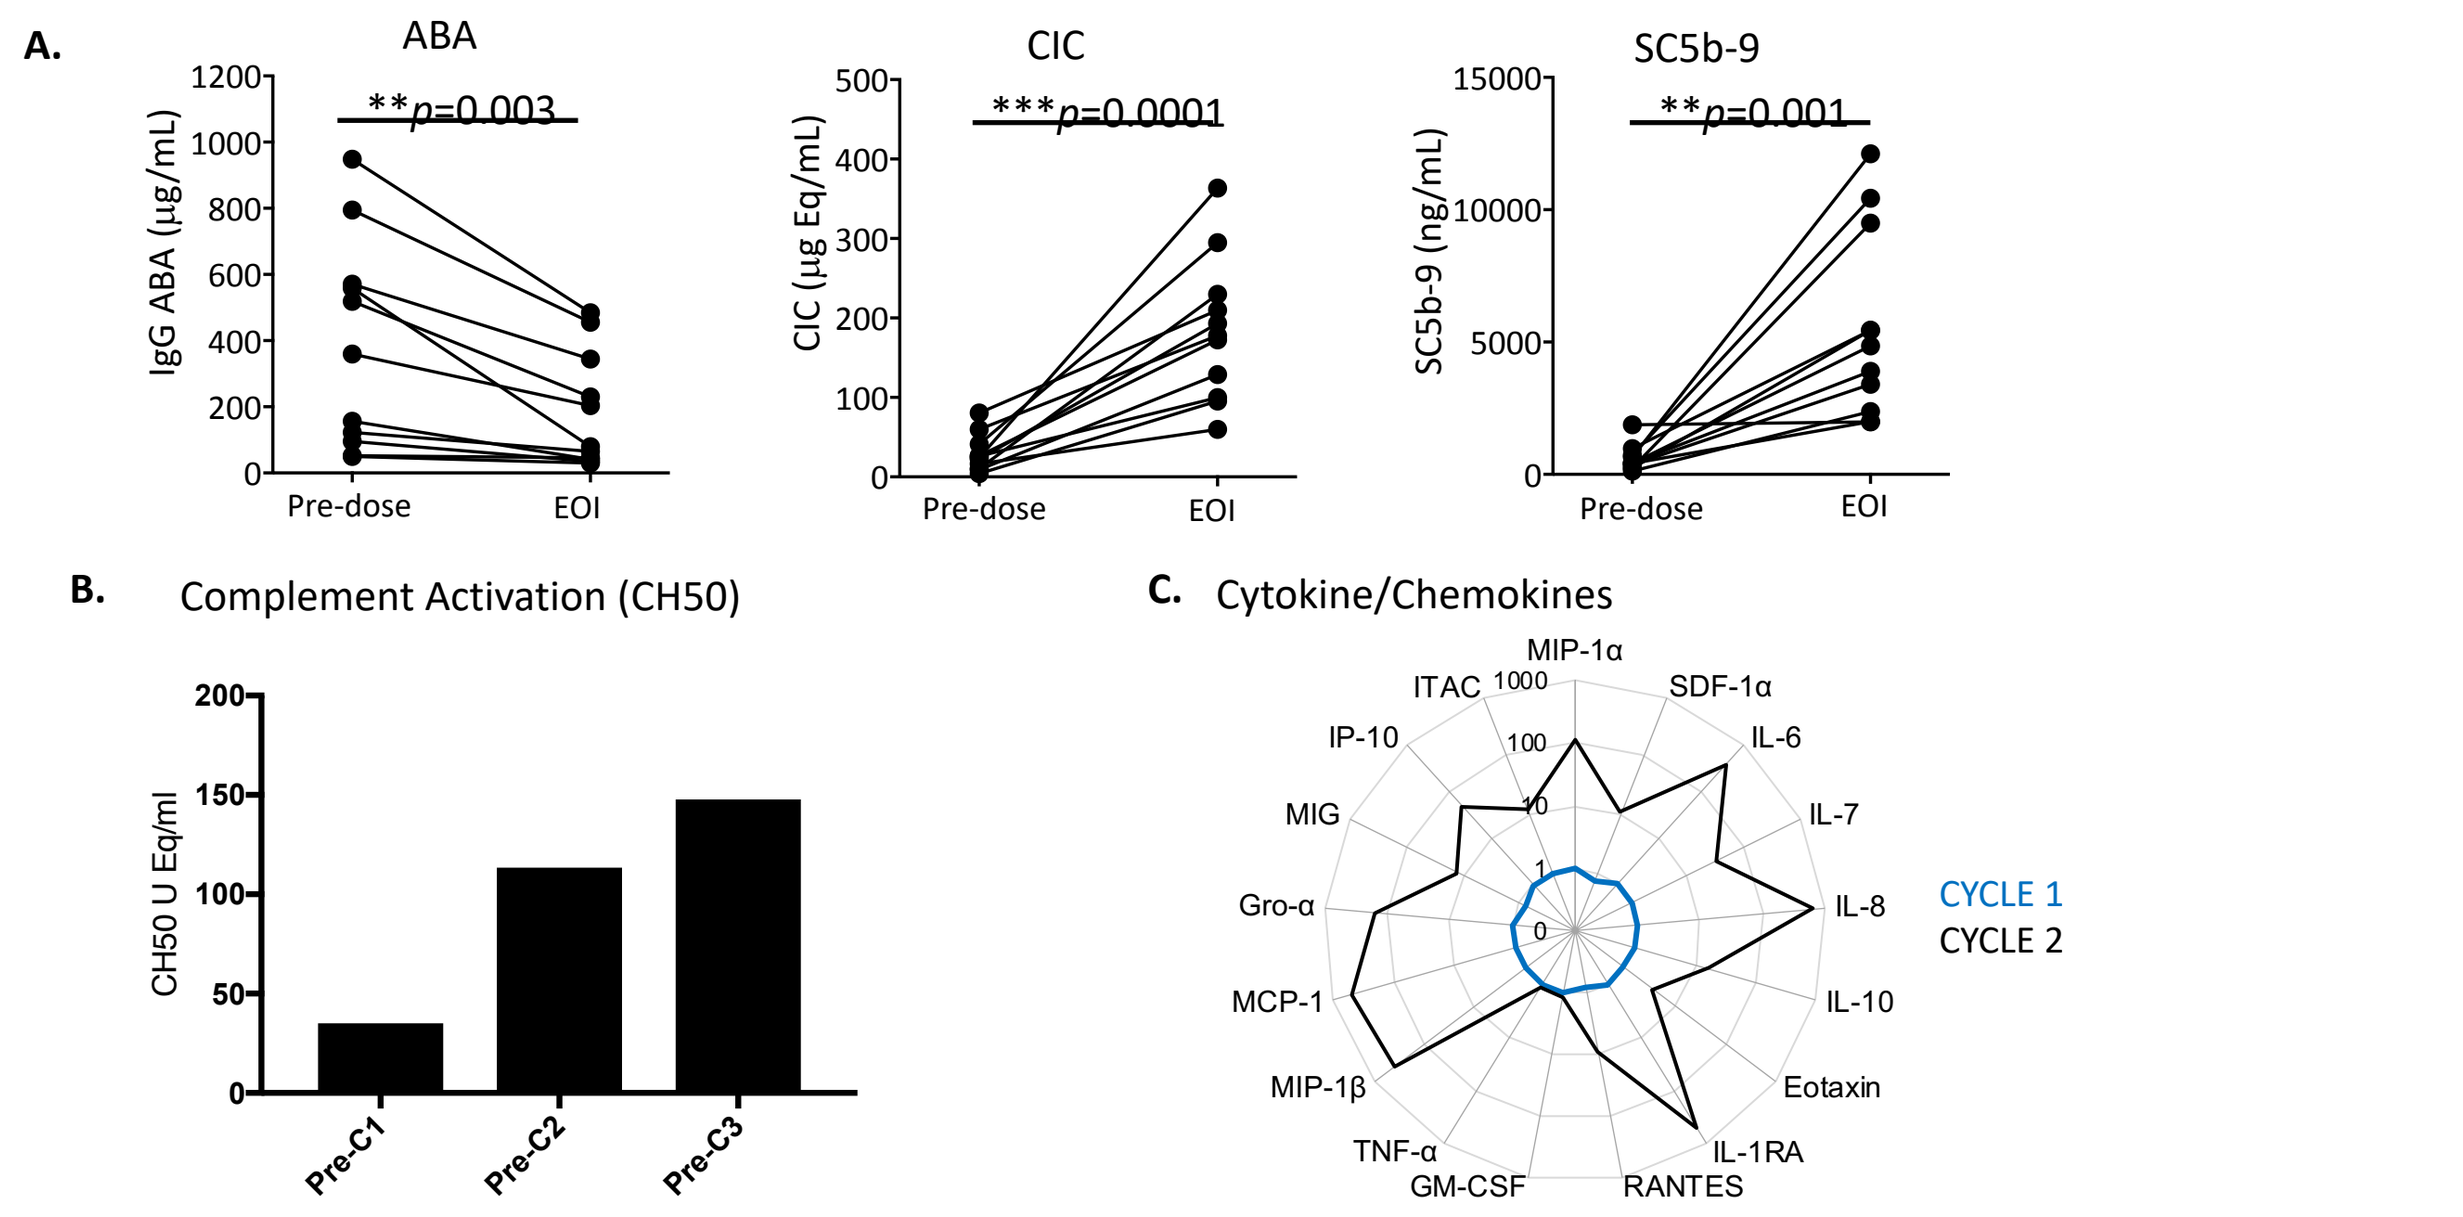
A.
ABA
**p=0.003
IgG ABA (µg/mL)
CIC
***p=0.0001
CIC (µg Eq/mL)
SC5b-9
**p=0.001
SC5b-9 (ng/mL)
Pre-dose
EOI
B. Complement Activation (CH50)
CH50 U Eq/ml
Pre-C1
Pre-C2
Pre-C3
C. Cytokine/Chemokines
MIP-1α
SDF-1α
IL-6
IL-7
IL-8
IL-10
Eotaxin
IL-1RA
RANTES
GM-CSF
TNF-α
MIP-1β
MCP-1
Gro-α
MIG
IP-10
ITAC
CYCLE 1
CYCLE 2

Figure 2. Imprime and pembrolizumab dosing resulted in several innate and adaptive immune activating pharmacodynamic changes that generally peaked between cycle 1 and cycle 2. We explored peripheral blood changes at pre-dose and end of infusion (EOI) of Cycles 1, 2, 3, 4, 5, 6, and beyond. The peak IPD responses are observed between cycles 1 and 2. **(A)** Maximum fold-increase in serum levels of free ABA, CIC, and complement-activation product SC5b-9 measured at cycle 2 EOI by ELISA, in the first 12 subjects **(B)** Total complement, as measured by CH50 is increased over after weekly dosing with Imprime in a representative subject. **(C)** Fold increase over C1 in cytokine levels measured at cycles 1-2 EOI by Luminex in a representative subject (103102)

Imprime Binds to Monocytes and Dendritic Cells, Evidence for Target Engagement (Dectin-1): TNBC Subject

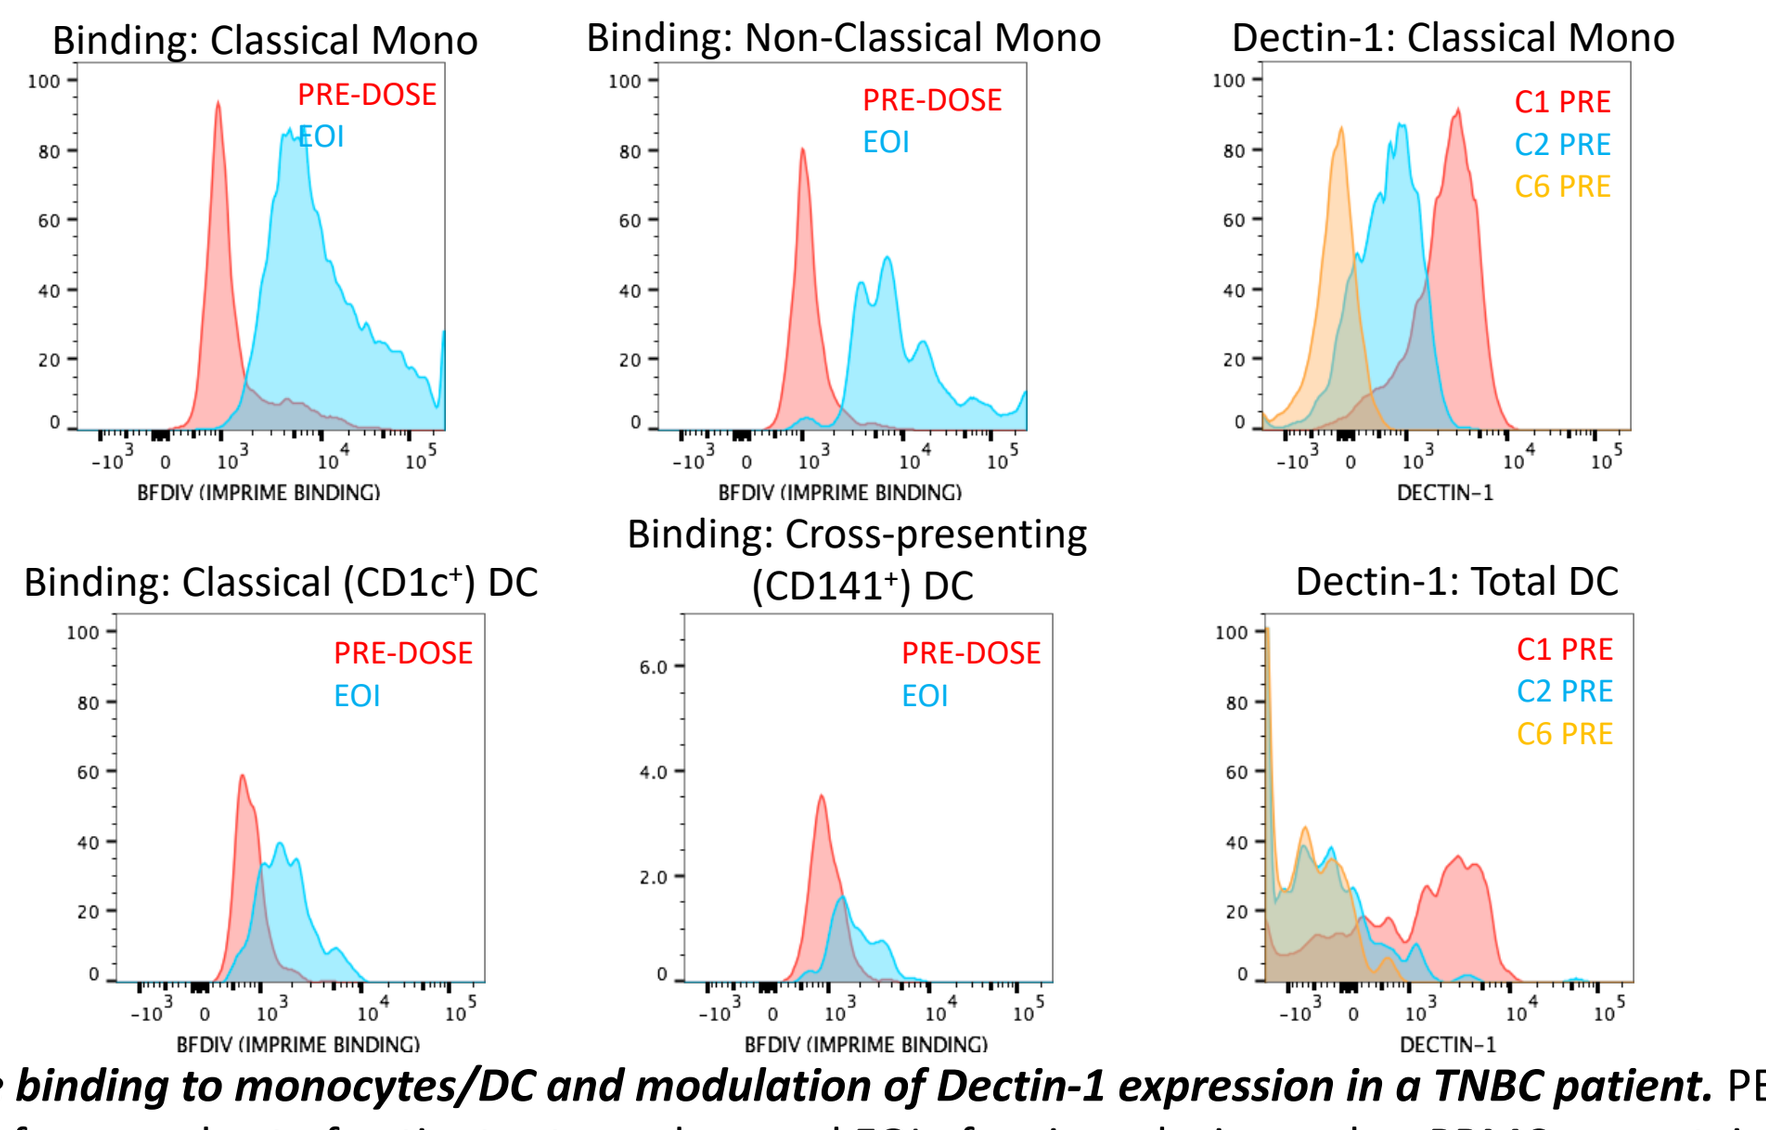
Binding: Classical Mono
Binding: Non-Classical Mono
Dectin-1: Classical Mono
Binding: Classical (CD1c+) DC
Binding: Cross-presenting (CD141+) DC
Dectin-1: Total DC
PRE-DOSE
EOI
C1 PRE
C2 PRE
C6 PRE
BFDIV (IMPRIME BINDING)
DECTIN-1

Figure 3. Imprime binding to monocytes/DC and modulation of Dectin-1 expression in a TNBC patient. PBMC were isolated from whole blood from a subset of patients at pre-dose and EOI of various dosing cycles. PBMC were stained with anti-beta glucan antibody (BfDIV) and markers for monocytes/DC subsets. Imprime binding and modulation of Dectin-1 expression on different monocytes and DC subsets were analyzed by flow cytometry.

Imprime Activates Monocyte and DC Subsets in a TNBC Subject

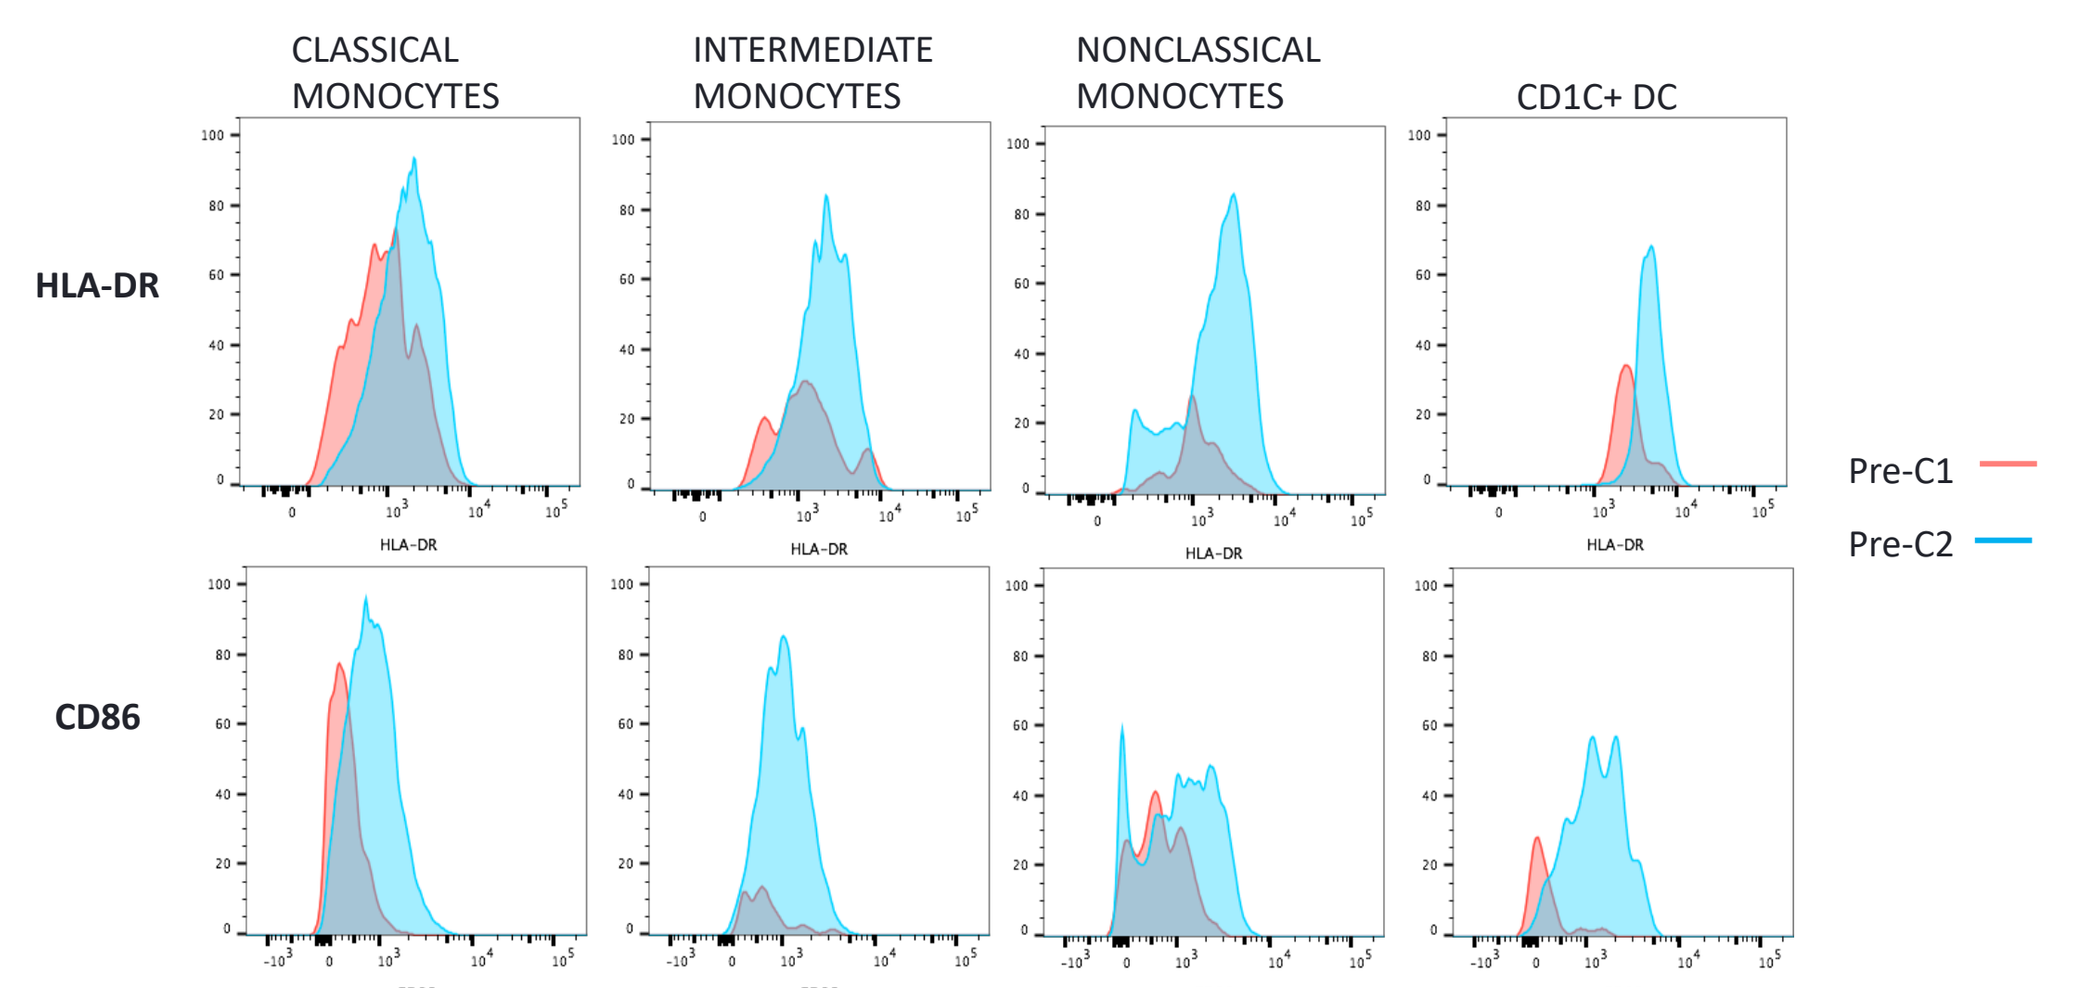
CLASSICAL MONOCYTES
INTERMEDIATE MONOCYTES
NONCLASSICAL MONOCYTES
CD1C+ DC
HLA-DR
CD86
Pre-C1
Pre-C2

Figure 4. Monocyte and DC activation in TNBC patients. PBMC were isolated from whole blood of patients at pre-dose and EOI of various dosing cycles. PBMC were stained with identification markers for monocytes or DC subsets and analyzed by flow cytometry. Each of the populations were assessed for expression of activation markers HLA-DR and CD86. Data shown here is for one of the RECIST responders.

Serum IPD Responses Associated with Clinical Benefit

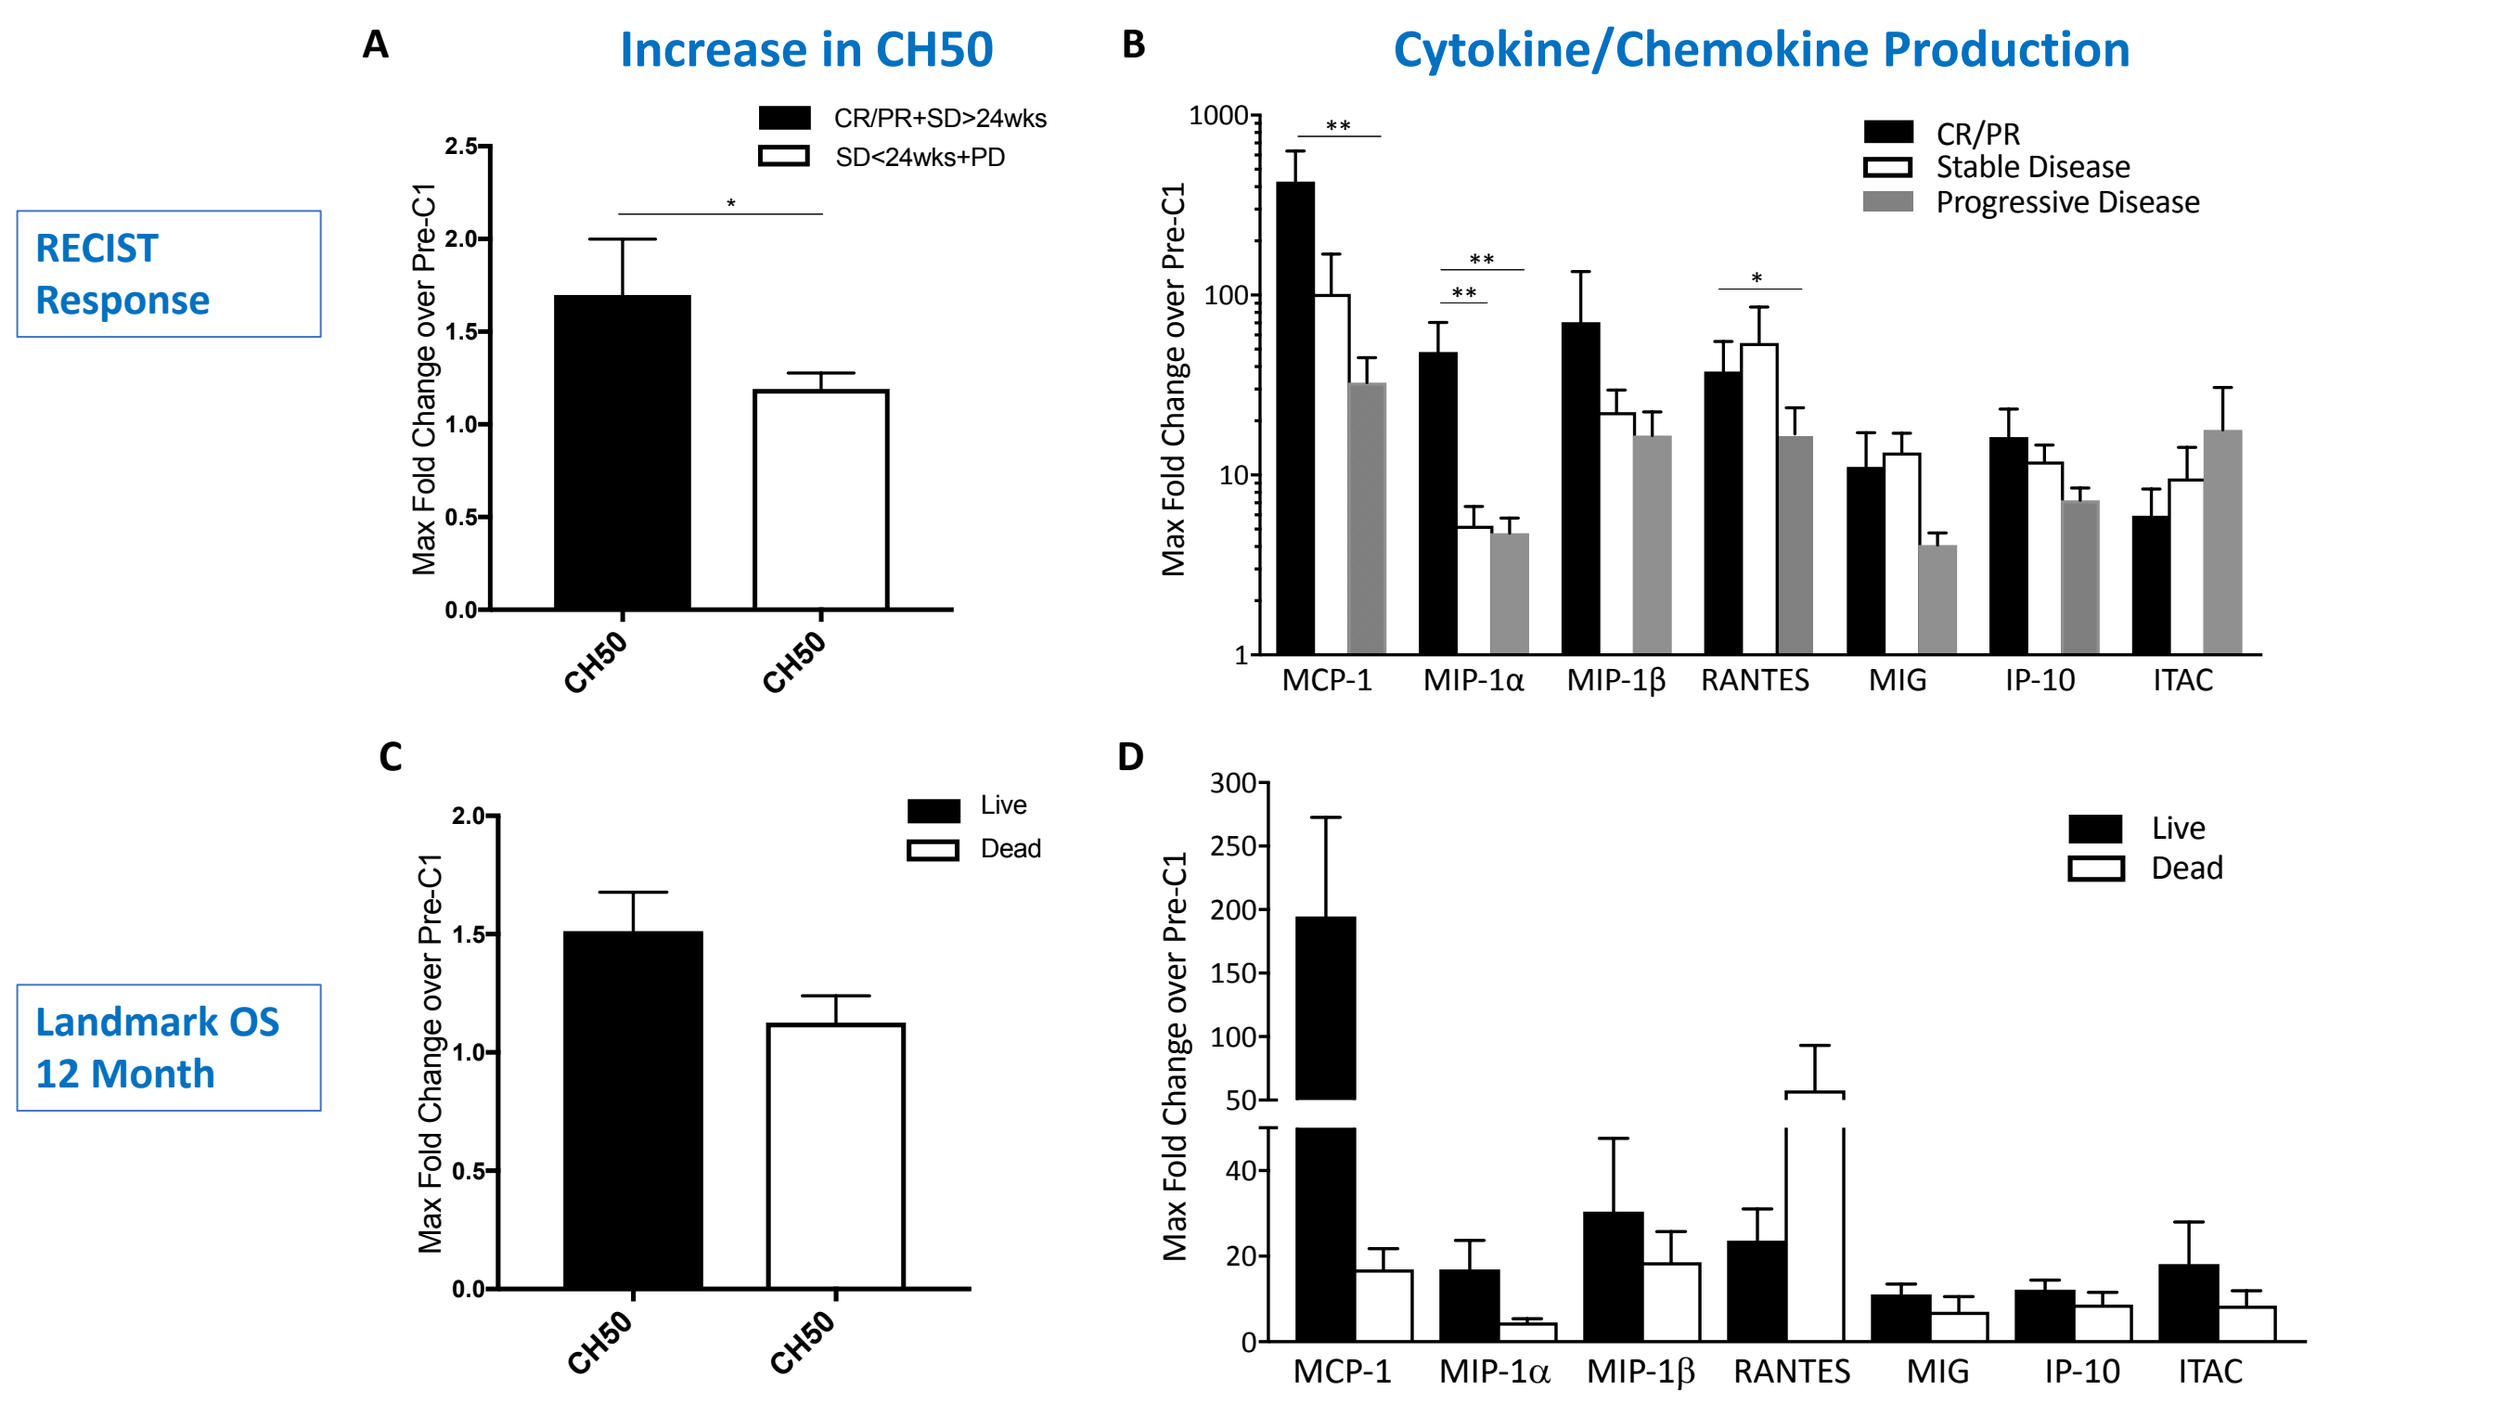
A
Increase in CH50
B
Cytokine/Chemokine Production
RECIST Response
CR/PR+SD>24wks
SD<24wks+PD
CR/PR
Stable Disease
Progressive Disease
Max Fold Change over Pre-C1
CH50
MCP-1
MIP-1α
MIP-1β
RANTES
MIG
IP-10
ITAC
C
D
Landmark OS 12 Month
Live
Dead

Figure 5. Increase in complement function measured by CH50 and chemokine/cytokine production associated with both RECIST response and enhanced overall survival. **(A and C)** CH50, a marker of classical complement pathway activation, is measured in serum collected at pre-dose and end of infusion (EOI) of various dosing cycles using commercial enzyme immunoassay (EIA) kits. Shown here are bars representing the average maximum fold-change (occurring approximately between C2 to C6) over pre-C1 are shown. **(B and D)** Chemokine/cytokine levels in serum collected at EOI of various dosing cycles are measured using commercial multiplex Luminex assays. Shown here are bars representing the average max fold-change over baseline . The maximum change was typically observed at C2. *P<0.05, **P<0.005.
RECIST: CR + PR + SD>24 wks (N=11); SD<24 wks + PD (N=31)
2 subjects discontinued before pre-C2 Landmark OS 12 month: Live=22 and Dead=16

Cellular IPD Responses and Association with Clinical Benefit

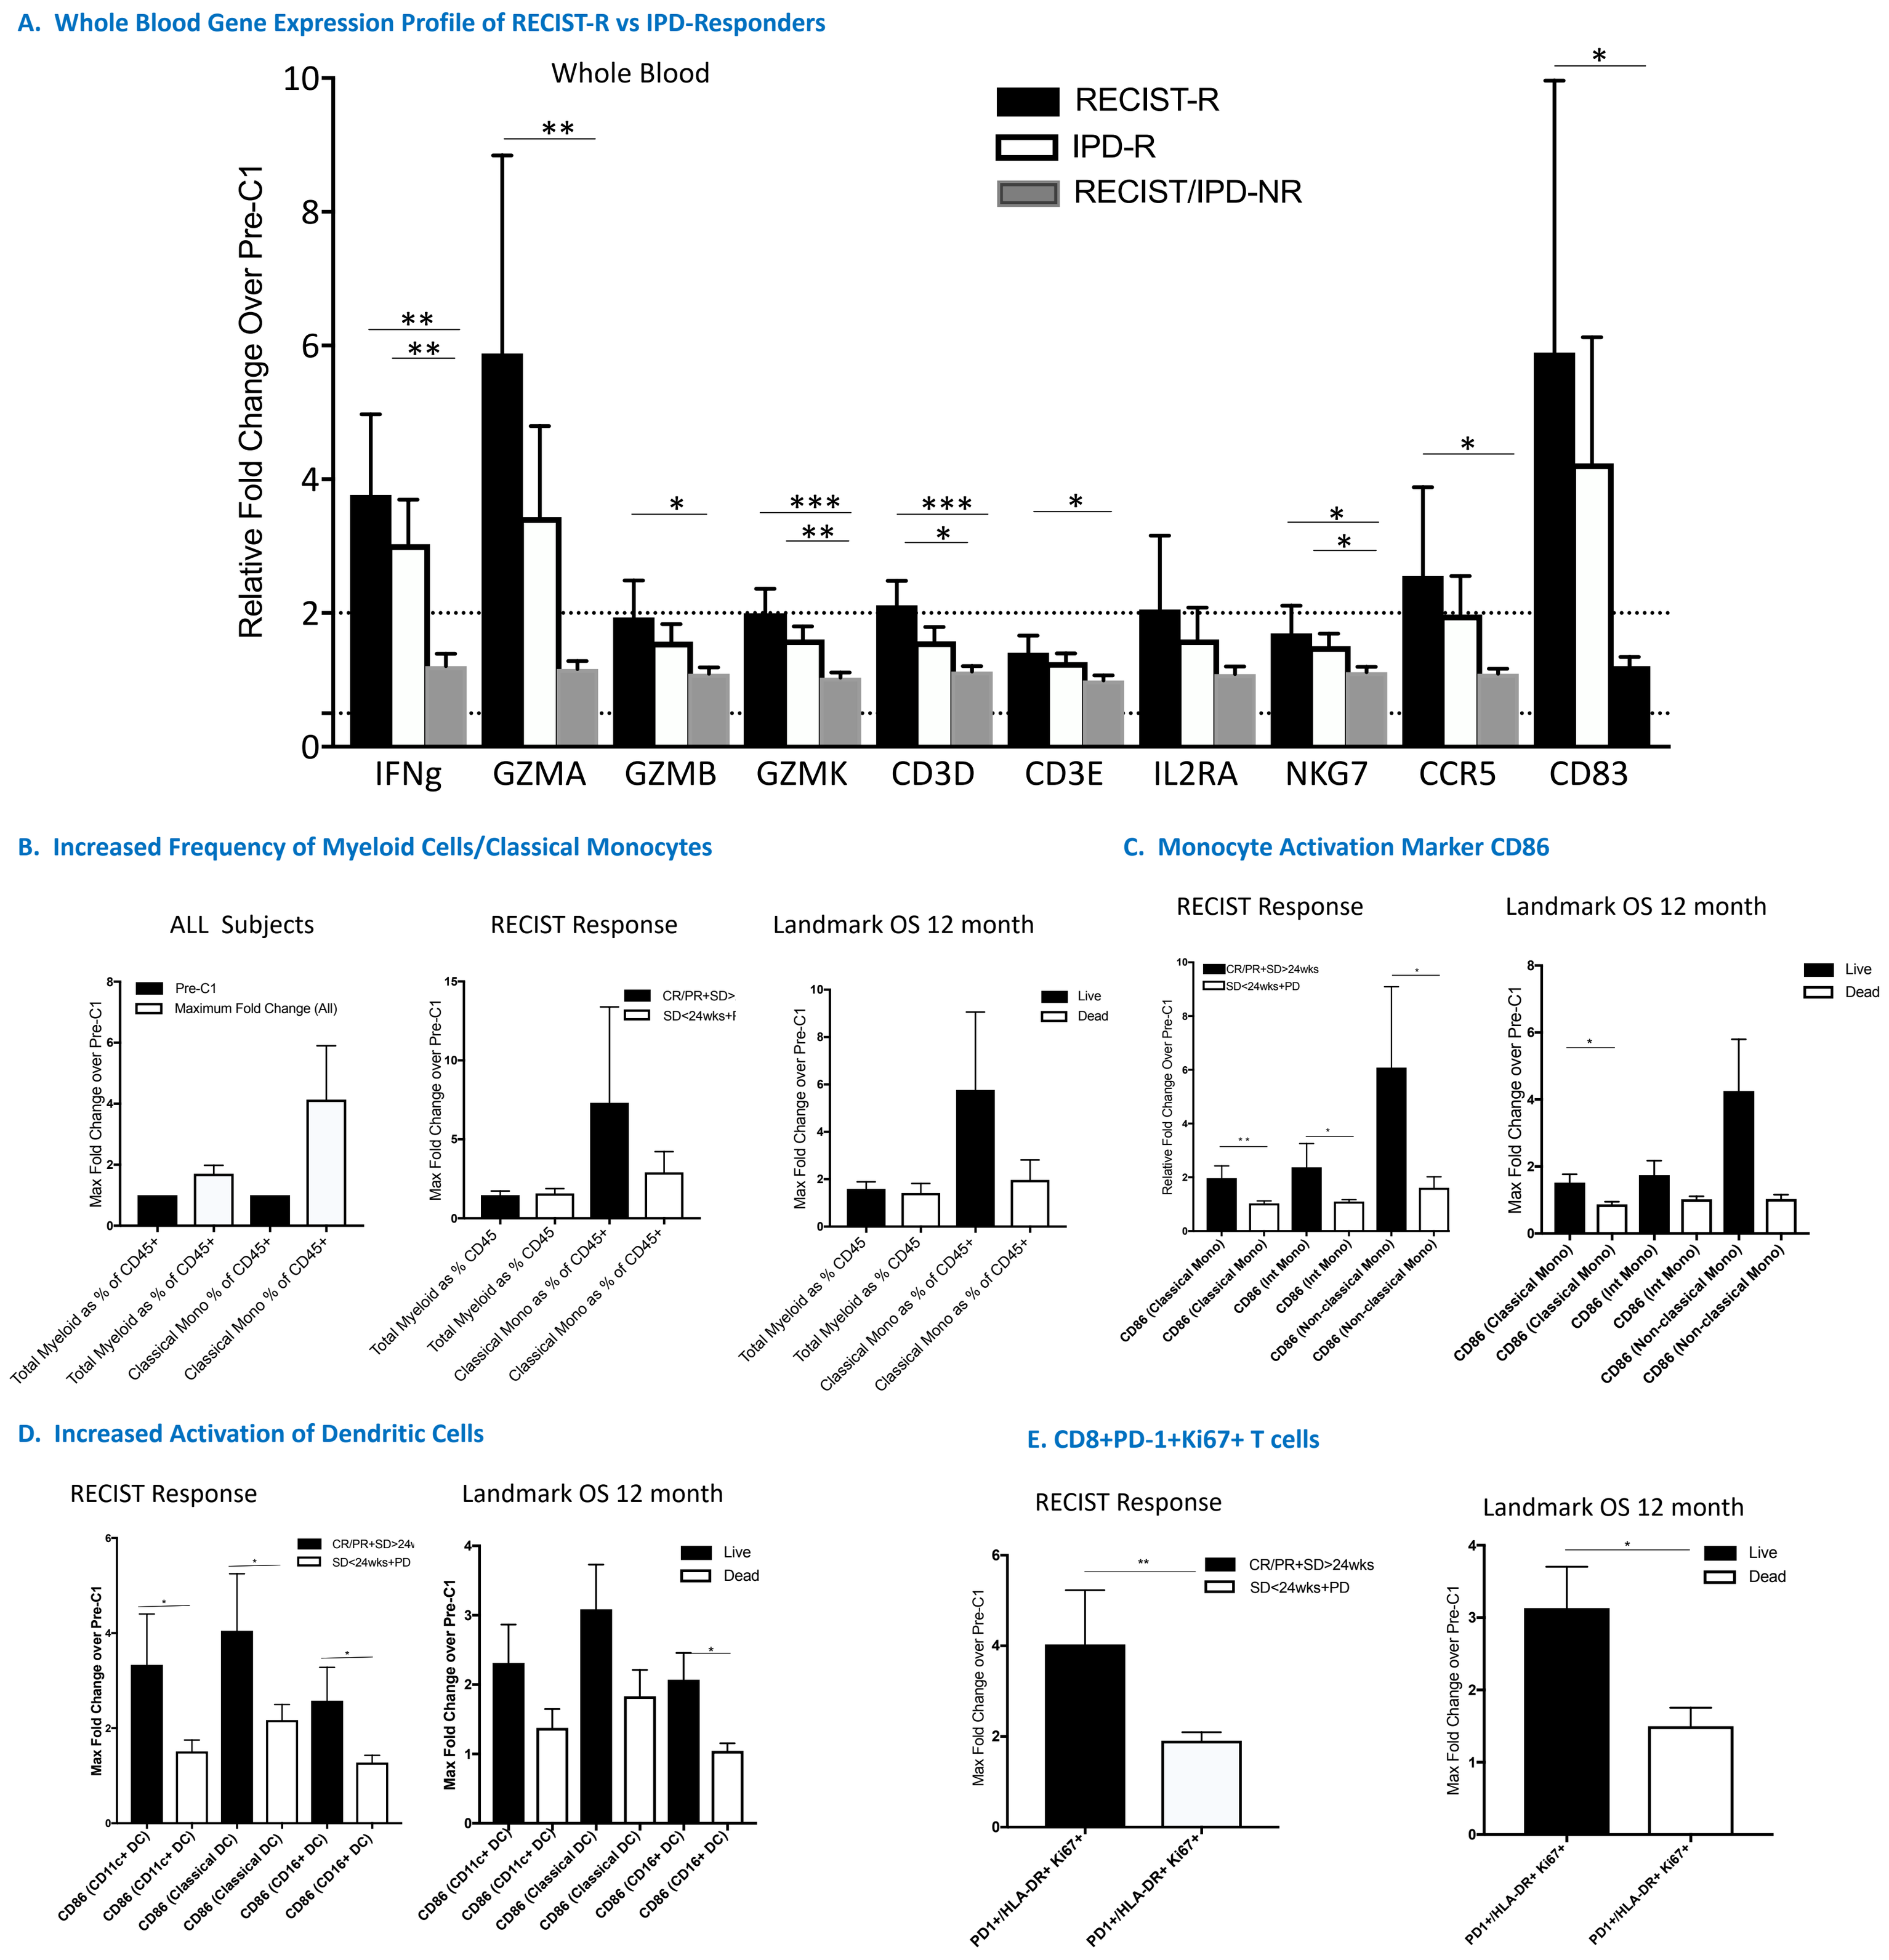
A. Whole Blood Gene Expression Profile of RECIST-R vs IPD-Responders
Whole Blood
Relative Fold Change Over Pre-C1
RECIST-R
IPD-R
RECIST/IPD-NR
IFNg
GZMA
GZMB
GZMK
CD3D
CD3E
IL2RA
NKG7
CCR5
CD83
B. Increased Frequency of Myeloid Cells/Classical Monocytes
ALL Subjects
RECIST Response
Landmark OS 12 month
C. Monocyte Activation Marker CD86
RECIST Response
Landmark OS 12 month
D. Increased Activation of Dendritic Cells
RECIST Response
Landmark OS 12 month
E. CD8+PD-1+Ki67+ T cells
RECIST Response
Landmark OS 12 month
Live
Dead

Figure 6. Changes in IPD responses are associated RECIST response and enhanced OS. **(A)** Expression of several innate and adaptive immune response gens were evaluated by QuantiGene technology. RNA was isolated from human whole blood by PAXgene Blood RNA kit. Gene expression values were normalized to the housekeeping genes and shown here is the max fold-change of the genes with >1.5-fold increase above pre-C1. RECIST-R (N=7), IPD-R (N=16), IPD-NR (N=24). **(B)** PBMC were isolated from whole blood of patients at pre-dose and EOI of various dosing cycles. Total myeloid cells (HLA-DR+) and classical monocytes (CD14+ CD16-) were measured as percentage of CD45+ immune cells in the peripheral blood by flow cytometry. The average max fold-change occurring at pre-dose C2/C6 over baseline (pre-C1) is plotted. **(C)** CD86 expression on the different monocyte subsets within PBMC, classical monocytes (CD14+ CD16-), intermediate monocytes (CD14+CD16+), and non-classical monocytes (CD14-CD16+) were measured by flow cytometry. The average max fold change occurring at pre-dose C2/C6 over baseline (pre-C1) is plotted. **(D)** CD86 expression on total DC (CD11c+), classical DC (CD1c+), inflammatory or monocyte-derived DC (CD16+) were measured by flow cytometry. The average max fold-change occurring at pre-dose C2/C6 over baseline (pre-C1) is plotted. **(E)** T cells and frequency of PD1+/HLA-DR+ Ki67+ CD8 T as a percentage of total CD8 T cells were analyzed by flow cytometry. The average max fold-change occurring at pre-dose C2/C6 over baseline (pre-C1) is plotted. P<0.05, **P<0.005, ***P<0.0005.

For **(B-E)**,
RECIST: CR + PR + SD>24 wks (N=11); SD<24 wks + PD (N=31)
2 subjects discontinued before pre-C2
Landmark OS 12 month: Live=22 and Dead=16

IPD Responders vs RECIST Responders

- Robust innate and adaptive immune responses were observed in RECIST responders
 - Some of these immunological responses were also observed in subjects who did not have RECIST responses but survived longer
- In order to delineate the immunological responses that best associated with increased overall survival, the subjects were stratified as **'IPD responders (IPD-R)' and 'IPD non-responders (IPD-NR)'** based on several of the innate and adaptive immune activation parameters:
 - >1.3 fold-increase in CH50 value post-treatment
 - >1.5 fold-increase in CD86 expression on classical monocytes post-treatment
 - >2-fold increase in the the PD1+/HLA-DR+ Ki67+ CD8 T cells post-treatment

A. CH50 and OS

B. CD86 Expression and OS

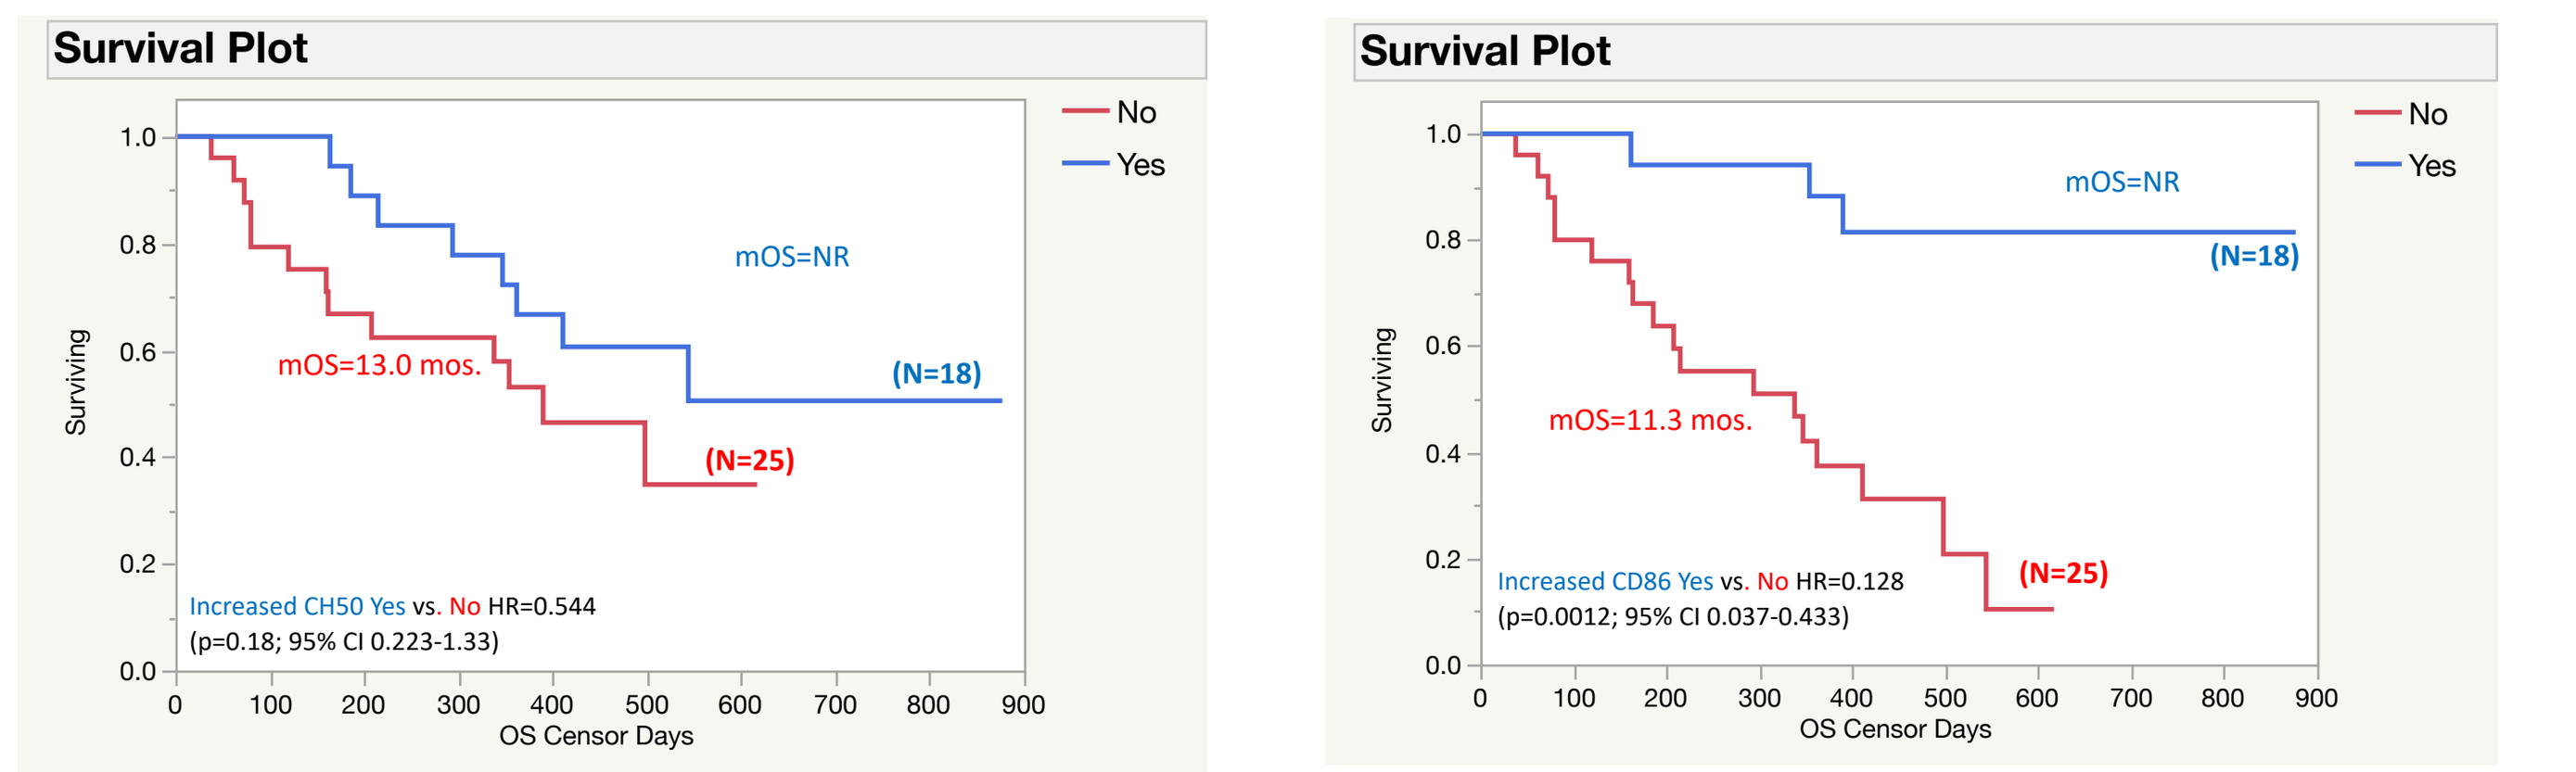
Survival Plot
No
Yes
mOS=NR
(N=18)
mOS=13.0 mos.
(N=25)
Increased CH50 Yes vs. No HR=0.544
(p=0.18; 95% CI 0.223-1.33)
OS Censor Days
Surviving
Survival Plot
mOS=NR
(N=18)
mOS=11.3 mos.
(N=25)
Increased CD86 Yes vs. No HR=0.128
(p=0.0012; 95% CI 0.037-0.433)

C. Activated CD8 T cells (PD1+/HLA-DR+ Ki67+) and Progression Free Survival (PFS)

D. Activated CD8 T cells (PD1+/HLA-DR+ Ki67+) and OS

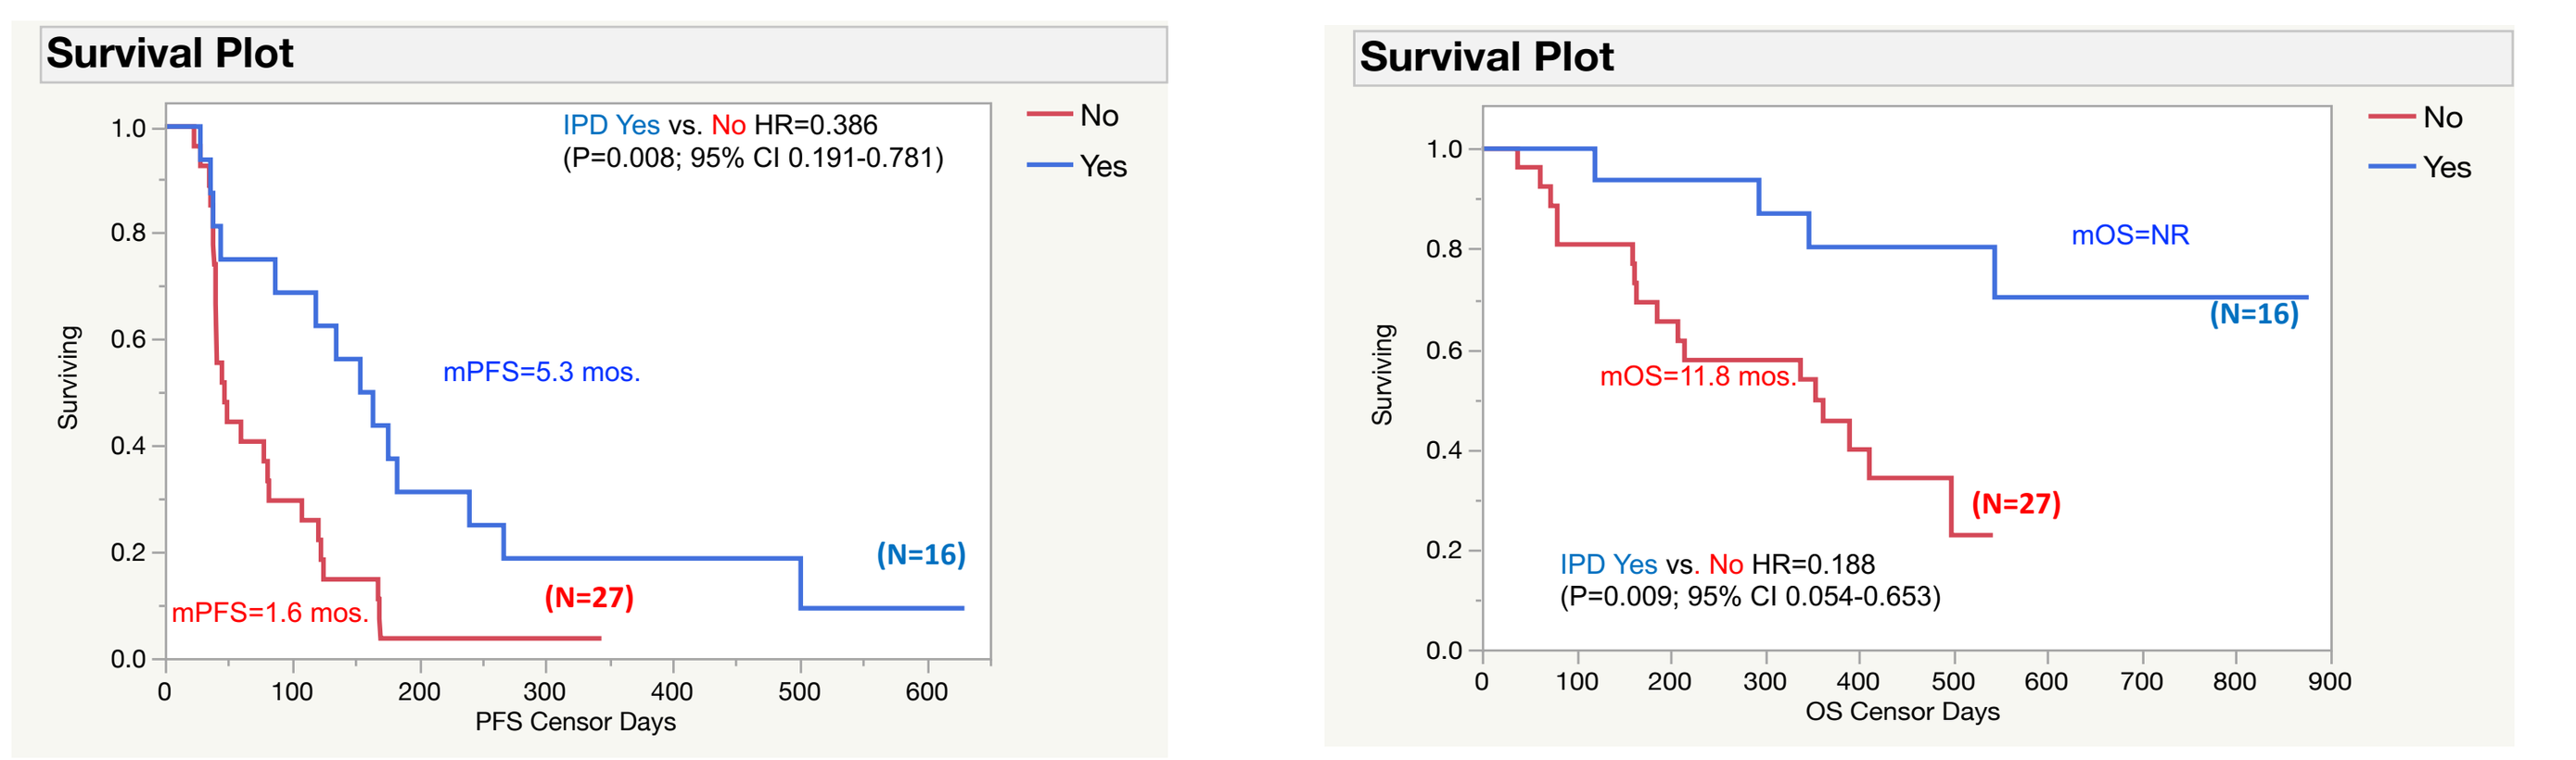
Survival Plot
IPD Yes vs. No HR=0.386
(P=0.008; 95% CI 0.191-0.781)
mPFS=5.3 mos.
(N=16)
mPFS=1.6 mos.
(N=27)
PFS Censor Days
Survival Plot
mOS=NR
(N=16)
mOS=11.8 mos.
(N=27)
IPD Yes vs. No HR=0.188
(P=0.009; 95% CI 0.054-0.653)
OS Censor Days

Figure 7. Imprime-mediated IPD effects mark patients with enhanced OS and PFS. **(A)** CH50, measure of activation of the classical complement pathway is measured in serum collected at pre-dose and end of infusion (EOI) of various dosing cycles using commercial EIA kits. Analyses of longitudinal blood samples from patients showed that a subset of patients had elevated CH50 as early as C2 (three weeks of treatment). Data shown represent a 1.3X or greater increase in CH50 vs baseline pre-treatment. **(B)** CD86 expression on classical monocytes (CD14+ CD16-) was measured by flow cytometry. Analyses of longitudinal blood samples from patients showed that a subset of patients had increased CD86 expression as early as C2 (three weeks of treatment). Data shown represent a 1.25X or greater increase in CD86 expression vs baseline pre-treatment. **(C-D)** PBMC were stained with markers for T cells and frequency of PD1+/HLA-DR+ Ki67+ CD8 T as a percentage of total CD8 T cells were analyzed by flow cytometry. Analyses of longitudinal blood samples from patients showed that a subset of patients had increased percentage of activated T cells as early as C2 (three weeks of treatment). Data shown in **(C)** represent a 1.5X or greater increase in PD1+/HLA-DR+ Ki67+ CD8 T vs baseline pre-treatment. Data shown in **(D)** represent a 2X or greater increase in PD1+/HLA-DR+ Ki67+ CD8 T vs baseline pre-treatment.

Summary

The IPD Responses in TNBC Demonstrate Both Innate and Adaptive Immune Activation

- Treatment with Imprime (in combination with pembrolizumab) elicits peripheral innate immune-activating immunopharmacodynamic changes including complement activity, select chemokine production, increased myeloid cells, and phenotypic activation of monocytes and DC. These activities have been previously evident in pre-clinical efficacy tumor models as well as healthy human volunteers treated only with Imprime.
- An increase in activated CD8 T cells was also observed in many patients (n =16). Since TNBC is not as immunogenic as other cancers like melanoma and there is less pre-existing T Cell immunity, robust innate immune activation may be key to inducing anti-tumor T cell responses.
- Concomitant with the peripheral immunological responses, the tumor biopsies showed a significant infiltration of activated myeloid and T cells.
- The strong association between the clinical responses and the innate/adaptive immune responses are suggestive of interplay between the therapeutic mechanisms of Imprime and pembrolizumab.