NOVEL GCN2 MODULATOR HC-7366 INHIBITS MYELOID-DERIVED SUPPRESSOR CELLS AND REDUCES PULMONARY METASTASES

SITC 2022
P1327

Jeremy Drees[1], Anissa SH Chan[1], Yunfang Li[1], Takashi O. Kangas[1], Weiyu Zhang[1], Maria Fumagalli[1], Iman Dewji[1], Kathryn Bieging-Rolett[1], Sho Fujisawa[1], Sharon Huang[1], Ben Harrison[1], Ashley LaCayo[1], Xiaohong Qiu[1], Nicholas Collette[1], Gemily Wang[1], Feven Tameire[1], Paulina Wojnarowicz[1], Crissy Dudgeon[1], Eric Lightcap[1], David Surguladze[1], Nandita Bose[1]

[1]HiberCell, Inc.

ABSTRACT

Background/Methods:

General controlled nonderepressible 2 (GCN2) is a central kinase in the integrated stress response (ISR) that responds to amino acid deprivation [1]. Cancer cells can utilize the ISR for survival, but prolonged or hyperactivation of the ISR reduces proliferation and induces apoptosis. We are developing HC-7366, a First-in-Class, First-in-Human GCN2 modulator that activates GCN2, resulting in anti-tumor activity. HC-7366, currently in a phase 1 trial (NCT05121948), has demonstrated robust efficacy in multiple pre-clinical solid tumor and AML models.

Myeloid-derived suppressor cells (MDSC) inhibit anti-tumor T cell immunity and promote metastatic spread [2]. Immature myeloid cells are also marked by ISR activation. We hypothesized that HC-7366 treatment could further activate the ISR in MDSC, leading to cell death or reduced suppressive function and improving anti-tumor immunity. To test this, we used the 4T1 murine breast cancer model, which is characterized by expansion of MDSC that facilitate lung metastasis. 4T1 cells were orthotopically transplanted into BALB/c mice. Tumor volume was monitored, and tissues or blood were collected at various timepoints for flow cytometry, IHC, or JESS analysis. Mouse bone marrow derived MDSC were cultured with T-cells in the presence of HC-7366 *in vitro*, and anti-proliferative function was evaluated.

Results:

HC-7366 treatment showed consistent anti-metastatic efficacy, reducing lung metastases by an average of ~75% across multiple studies. Primary tumors and metastases in treated mice demonstrated GCN2 pathway activation by increases in downstream signaling proteins, including the amino acid biosynthesis proteins ASNS and PSAT1. Anti-tumor efficacy was correlated with a >70% reduction in splenomegaly and an increased stimulatory gene expression profile in the spleen. HC-7366 treatment also significantly decreased Ly6G+ PMN-MDSC frequency in the lungs and spleen and increased their expression of the activation markers CD86 and MHCII. HC-7366 treatment also significantly increased T-cell infiltration, activation, and proliferation in metastatic lungs as measured by increased expression of IL-2, Ki67, T-bet, and Granzyme B. Furthermore, *in vitro* functional assays of bone marrow-derived MDSC co-cultured with T-cells demonstrated reduced T-cell inhibition when treated with HC-7366.

Additionally, HC-7366 treatment significantly increased CD98 expression on MDSC in the lungs but not on CD8 T-cells, potentially indicating greater ISR activation by HC-7366 in MDSC than in T-cells. HC-7366 also reduced the S100A8/A9 calcium binding proteins in myeloid cells in both metastatic and normal lung tissue, which have been implicated in facilitating MDSC recruitment and proliferation [3-5]. Reductions in S100A8/A9 were also detectable in PBMCs and plasma.

Conclusions:

Collectively, these data demonstrate the anti-metastatic efficacy of HC-7366 and its inhibitory effects on MDSC, outlining its potential as a monotherapy and in combination with other immunotherapeutics to treat MDSC-enriched metastatic cancers.

BACKGROUND

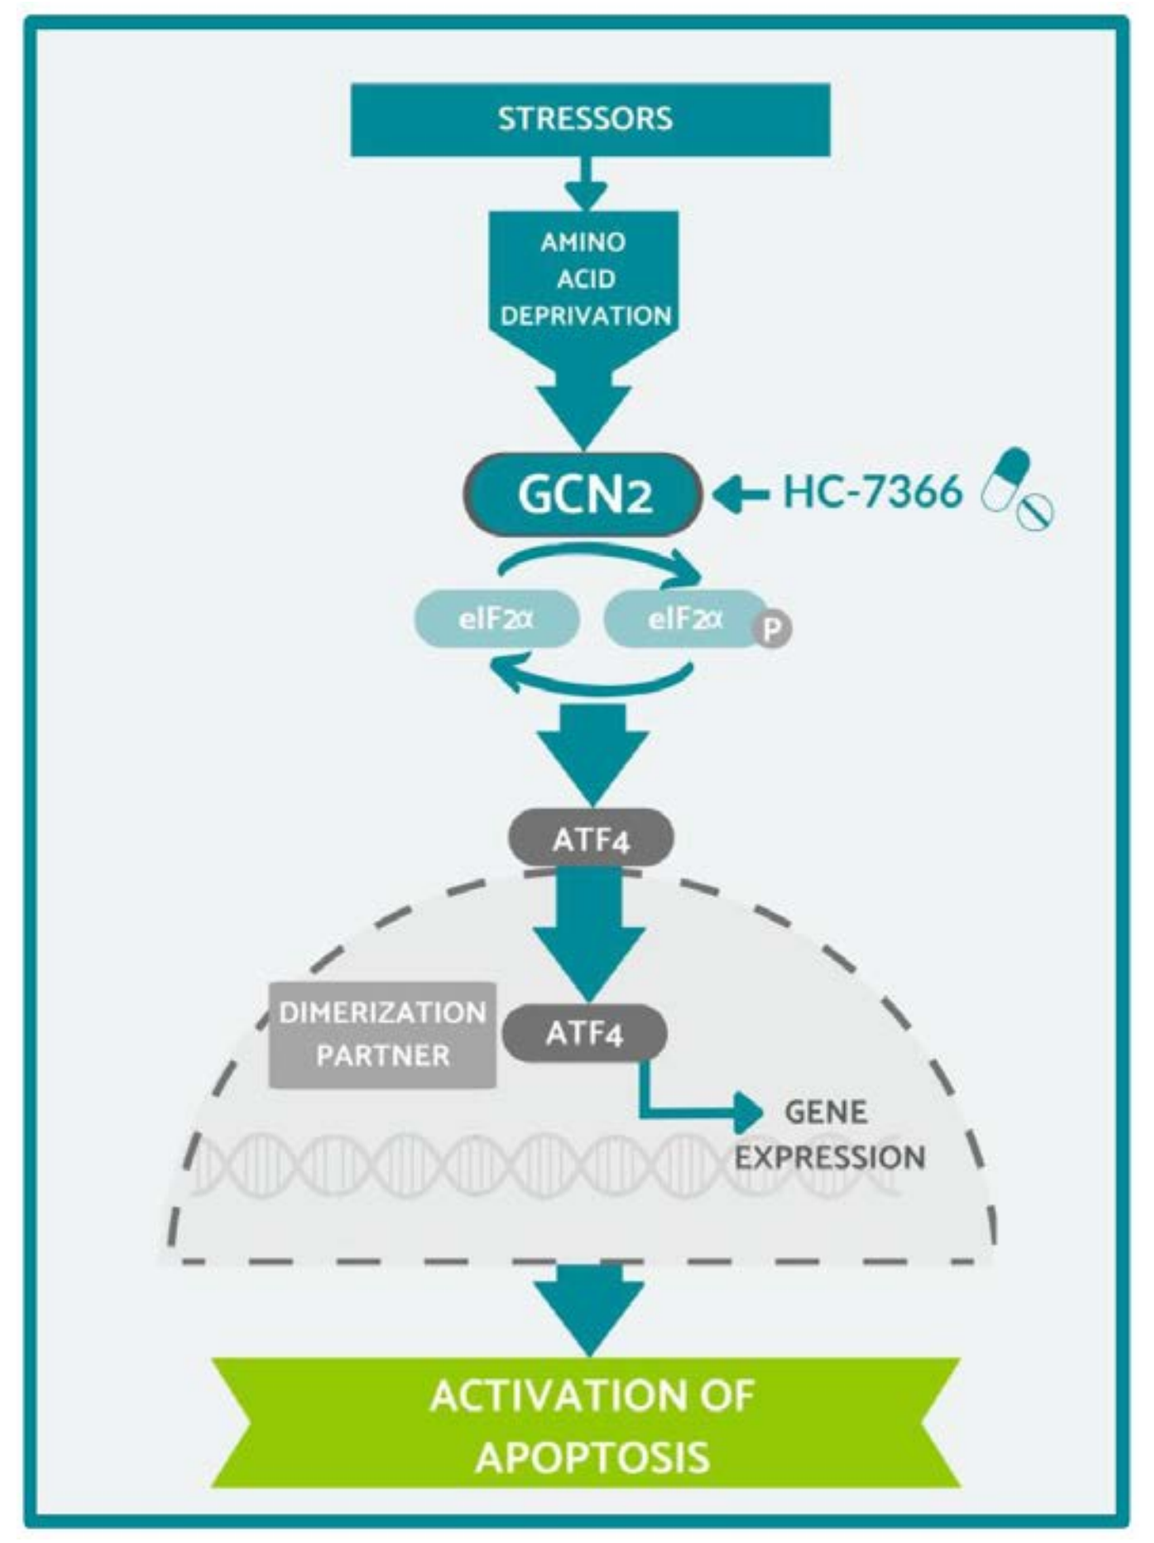
STRESSORS
AMINO ACID DEPRIVATION
GCN2
HC-7366
eIF2α
eIF2α
P
ATF4
DIMERIZATION PARTNER
ATF4
GENE EXPRESSION
ACTIVATION OF APOPTOSIS

Figure 1: GCN2 and HC-7366 in the Integrated Stress Response (ISR). GCN2 is one of the kinases in the ISR family and is a key metabolic stress sensor in cells. Amino acid deprivation causes phosphorylation of GCN2, which in turn phosphorylates eIF2α. Phosphorylation of eIF2α attenuates 5'Cap-dependent translation, while inducing translation of selected mRNAs that contain a short upstream open reading frame in their 5' UTR, including the transcription factor ATF4. Elevated ATF4 results in transcriptional upregulation of stress-responsive genes that lead to cell survival or apoptosis depending on context [1].

While cancer cells are known to utilize the ISR for survival, prolonged or hyper activation of the ISR has been shown to induce apoptosis [1]. HC-7366 is a First-in-Class, First-in-Human GCN2 modulator that activates GCN2, resulting in anti-tumor activity through lethal activation of the ISR pathway.

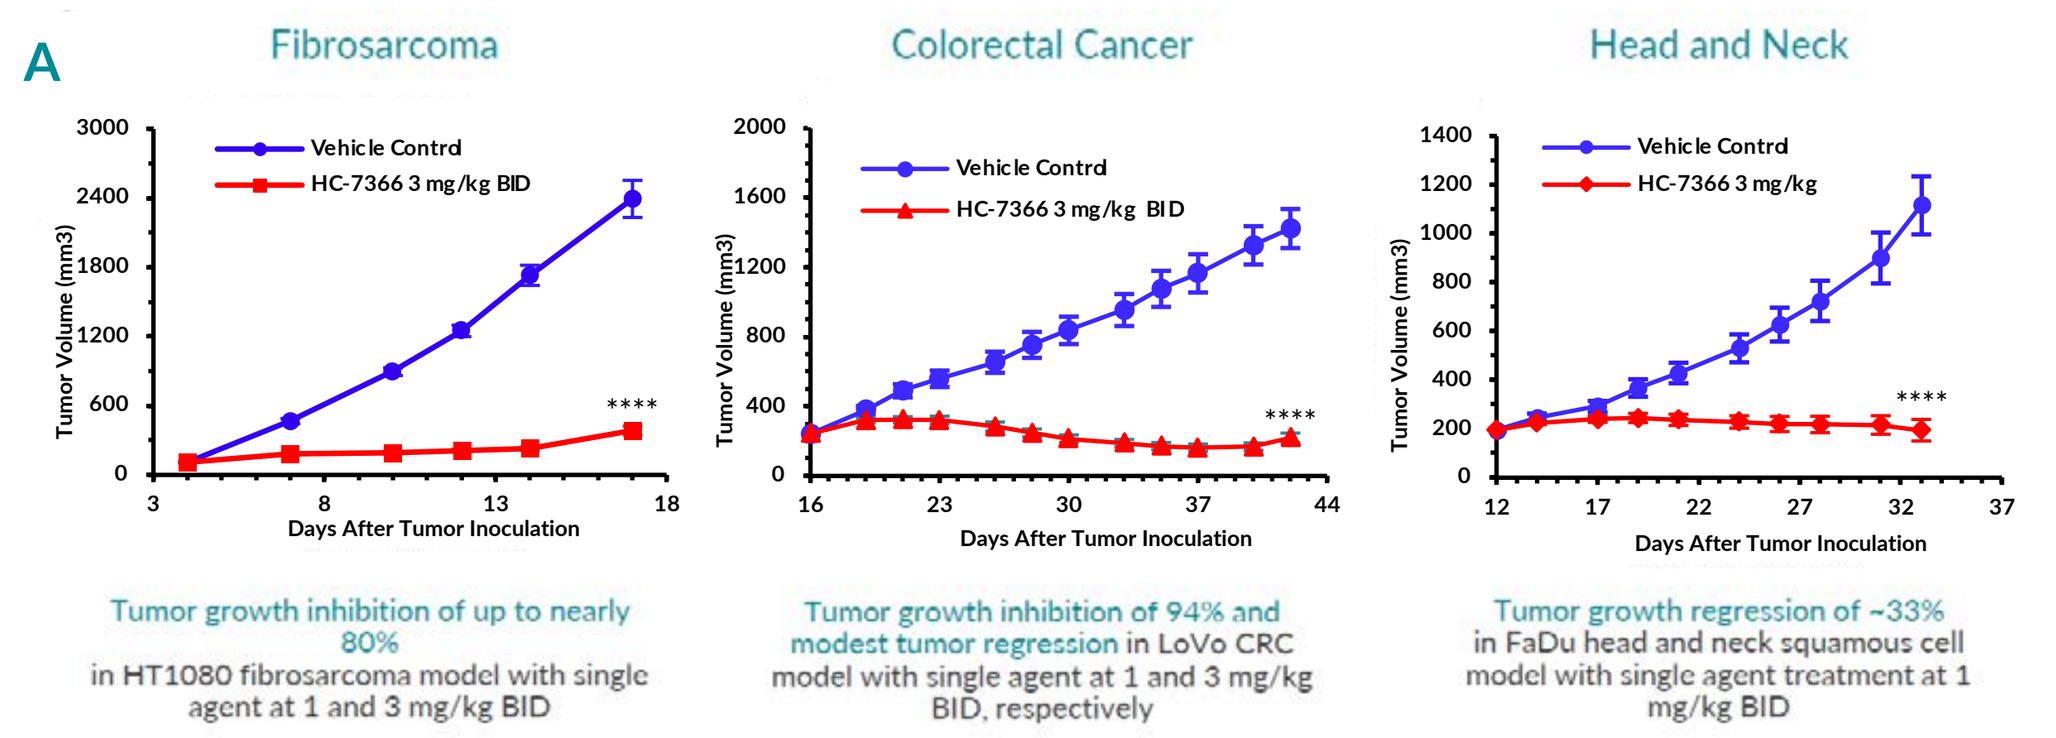
A
Fibrosarcoma
Colorectal Cancer
Head and Neck
Vehicle Control
HC-7366 3 mg/kg BID
Tumor Volume (mm3)
Days After Tumor Inoculation
Tumor growth inhibition of up to nearly 80% in HT1080 fibrosarcoma model with single agent at 1 and 3 mg/kg BID
Tumor growth inhibition of 94% and modest tumor regression in LoVo CRC model with single agent at 1 and 3 mg/kg BID, respectively
Tumor growth regression of ~33% in FaDu head and neck squamous cell model with single agent treatment at 1 mg/kg BID

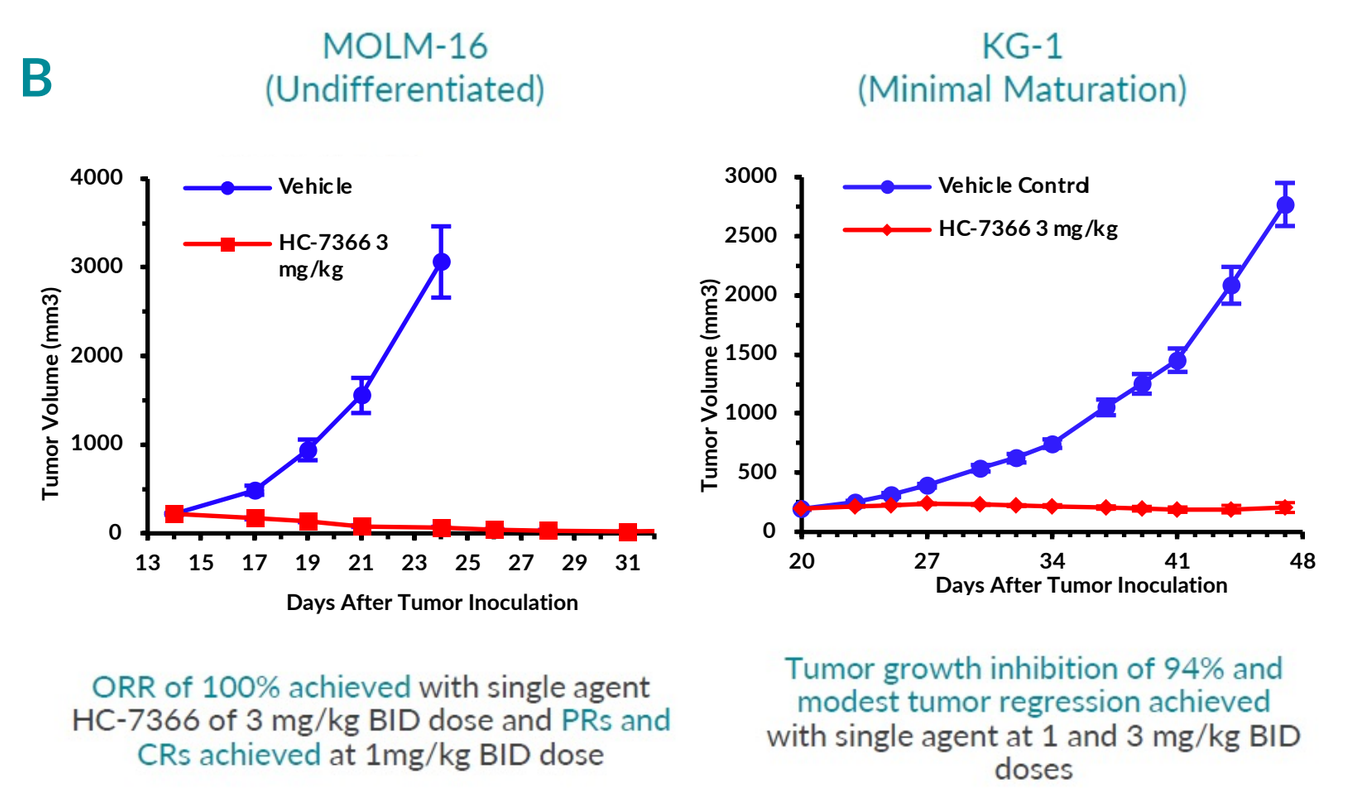
B
MOLM-16 (Undifferentiated)
KG-1 (Minimal Maturation)
Vehicle
HC-7366 3 mg/kg
Days After Tumor Inoculation
ORR of 100% achieved with single agent HC-7366 of 3 mg/kg BID dose and PRs and CRs achieved at 1mg/kg BID dose
Tumor growth inhibition of 94% and modest tumor regression achieved with single agent at 1 and 3 mg/kg BID doses

Figure 2: HC-7366 Shows Potent Single Agent Activity in Solid and Liquid Tumor Models. Robust anti-tumor efficacy is observed with HC-7366 treatment in multiple solid tumor (**A**) and AML (**B**) pre-clinical models of different types and maturation status. Efficacy coincides with activation of the ISR pathway.

RESULTS

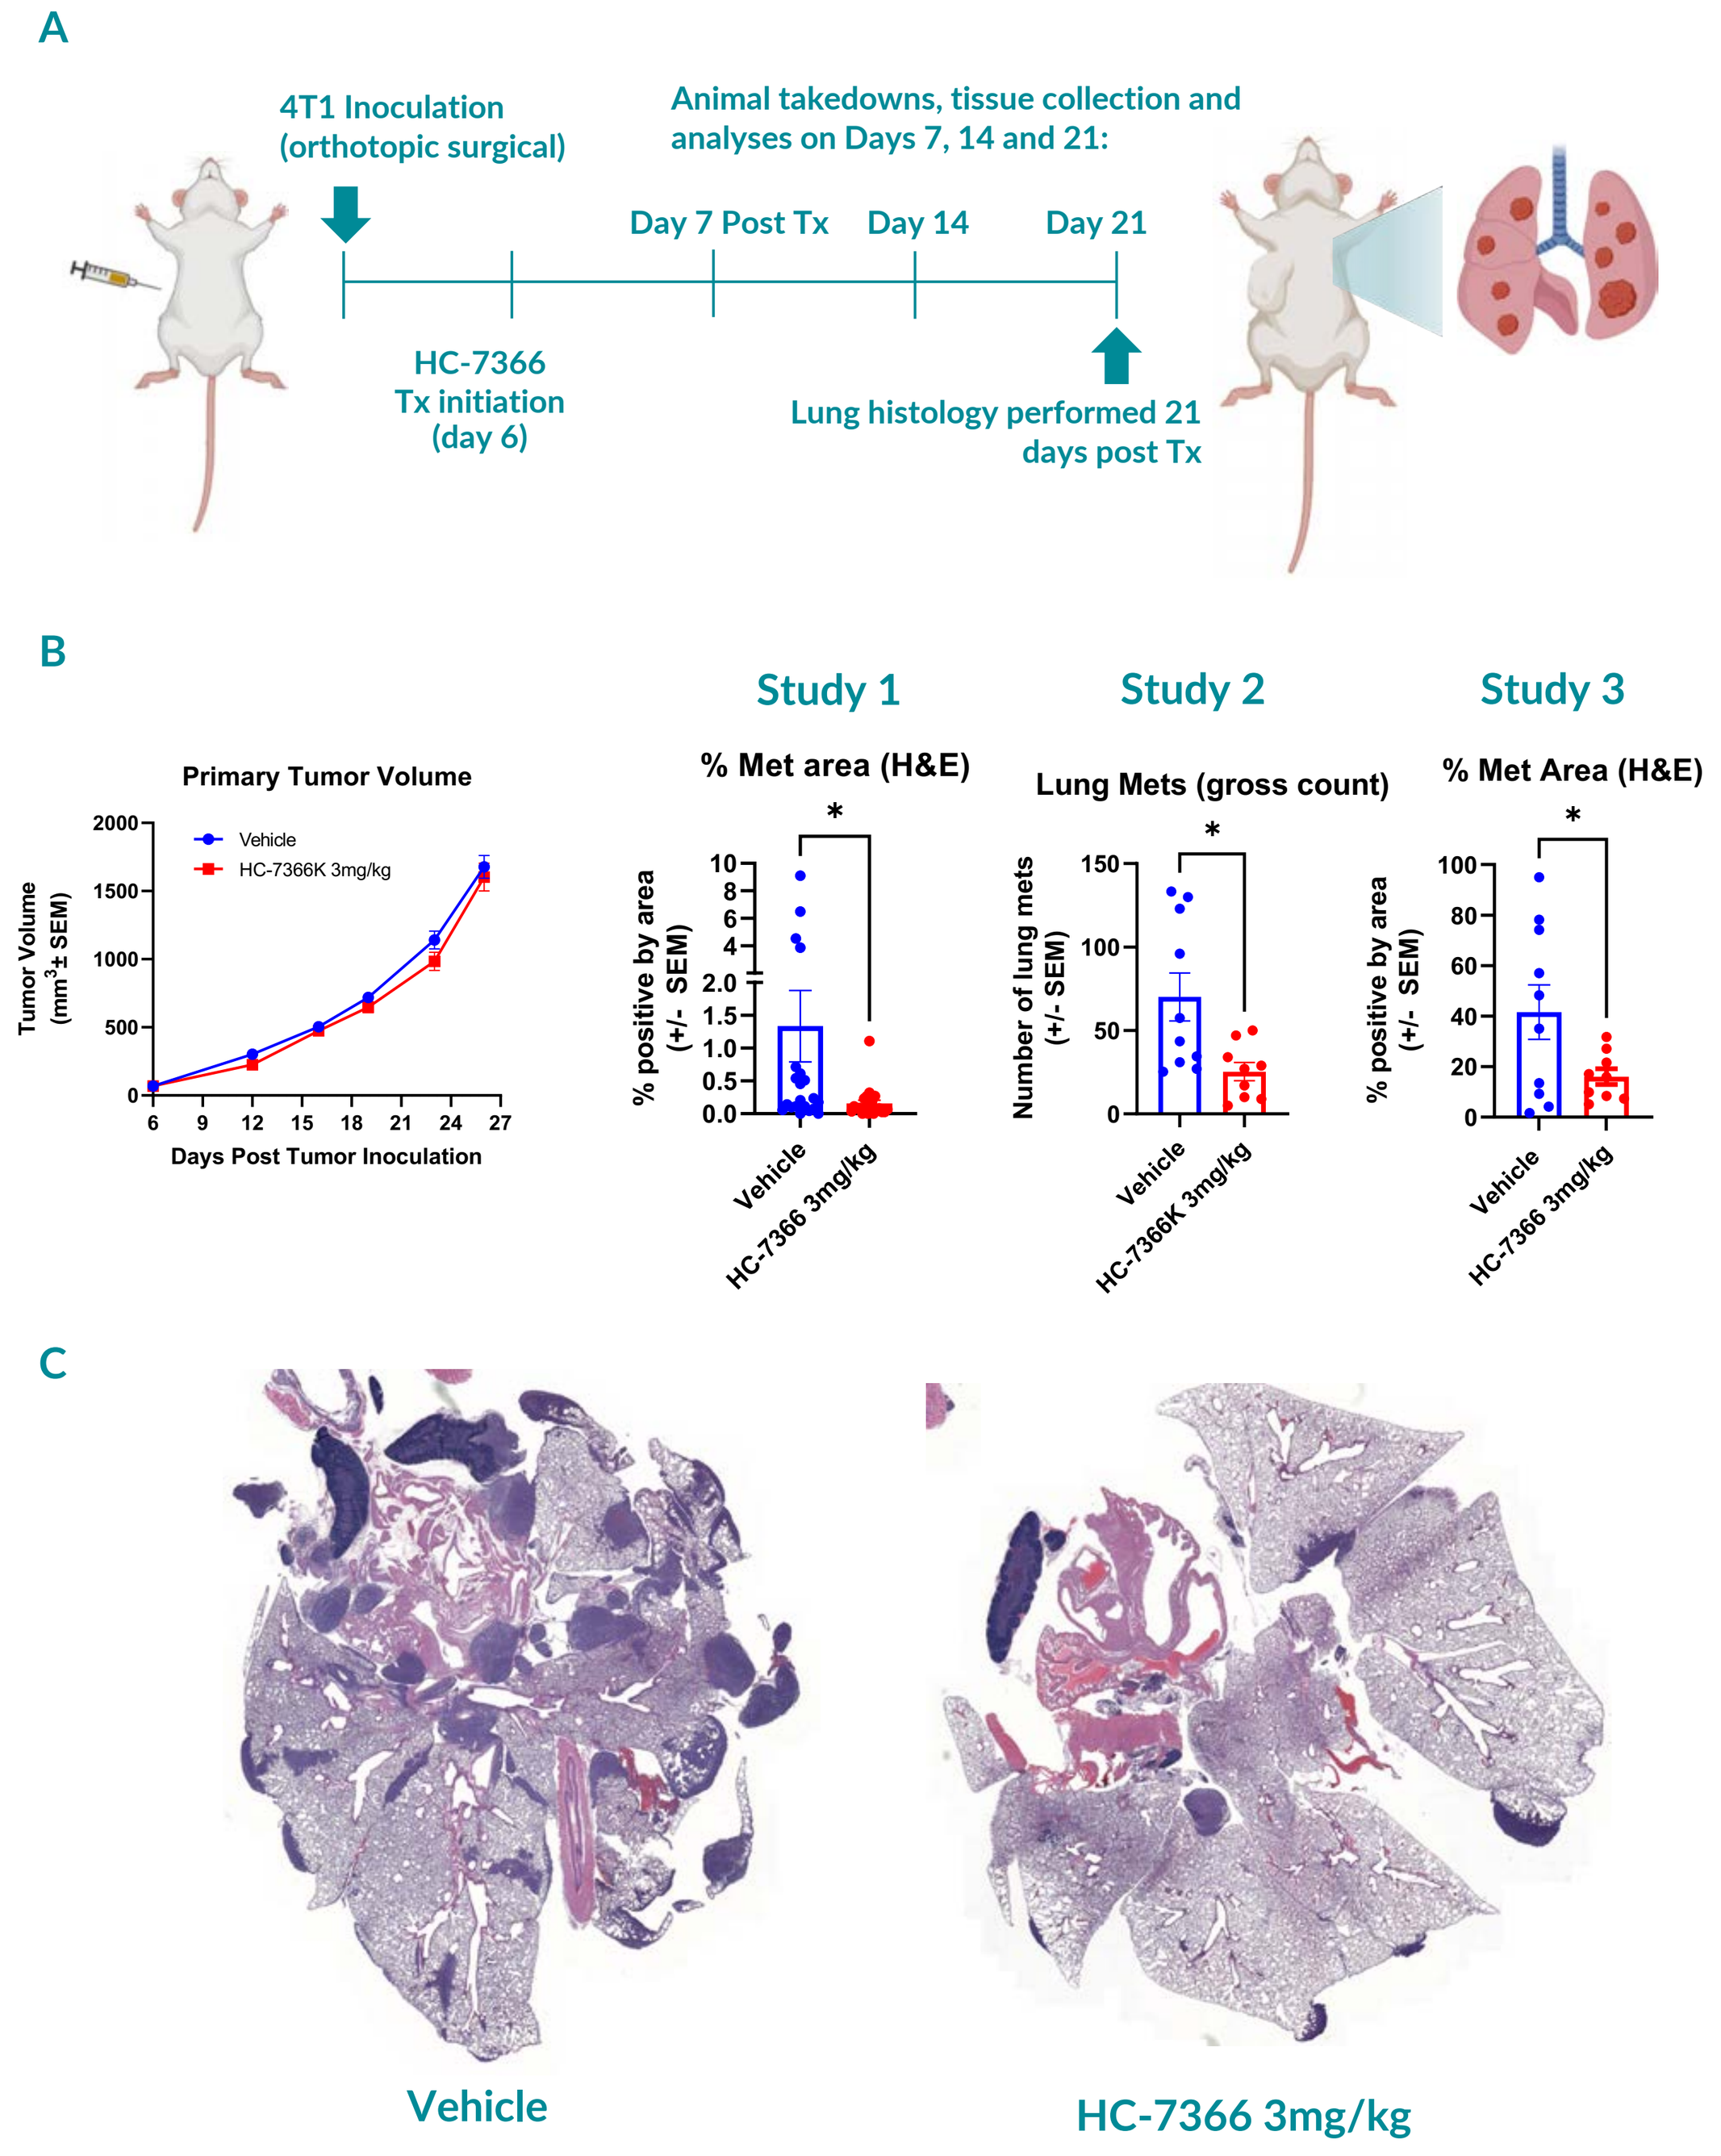
A
4T1 Inoculation (orthotopic surgical)
Animal takedowns, tissue collection and analyses on Days 7, 14 and 21:
Day 7 Post Tx
Day 14
Day 21
HC-7366 Tx initiation (day 6)
Lung histology performed 21 days post Tx
B
Primary Tumor Volume
Study 1
% Met area (H&E)
Study 2
Lung Mets (gross count)
Study 3
% Met Area (H&E)
C
Vehicle
HC-7366 3mg/kg

Figure 3: HC-7366 Reduces Pulmonary Metastases in 4T1 Breast Cancer. A) 4T1 study schema. 5x10^5 4T1 breast cancer cells were surgically implanted into the mammary fat pad of female BALB/c mice. Animals were randomized for treatment when tumors reached an average of 50-100mm^3. Treatment was administered PO, BID. Animals were sacrificed for analyses at 7-, 14-, and 21-days post treatment initiation. **B)** While HC-7366 showed limited effect on primary 4T1 tumors, it showed a consistent anti-metastatic effect in lungs of tumor bearing mice administered 3mg/kg HC-7366. Lungs were collected 21 days post treatment initiation (three independent studies). **C)** Representative H&E-stained images of lungs.

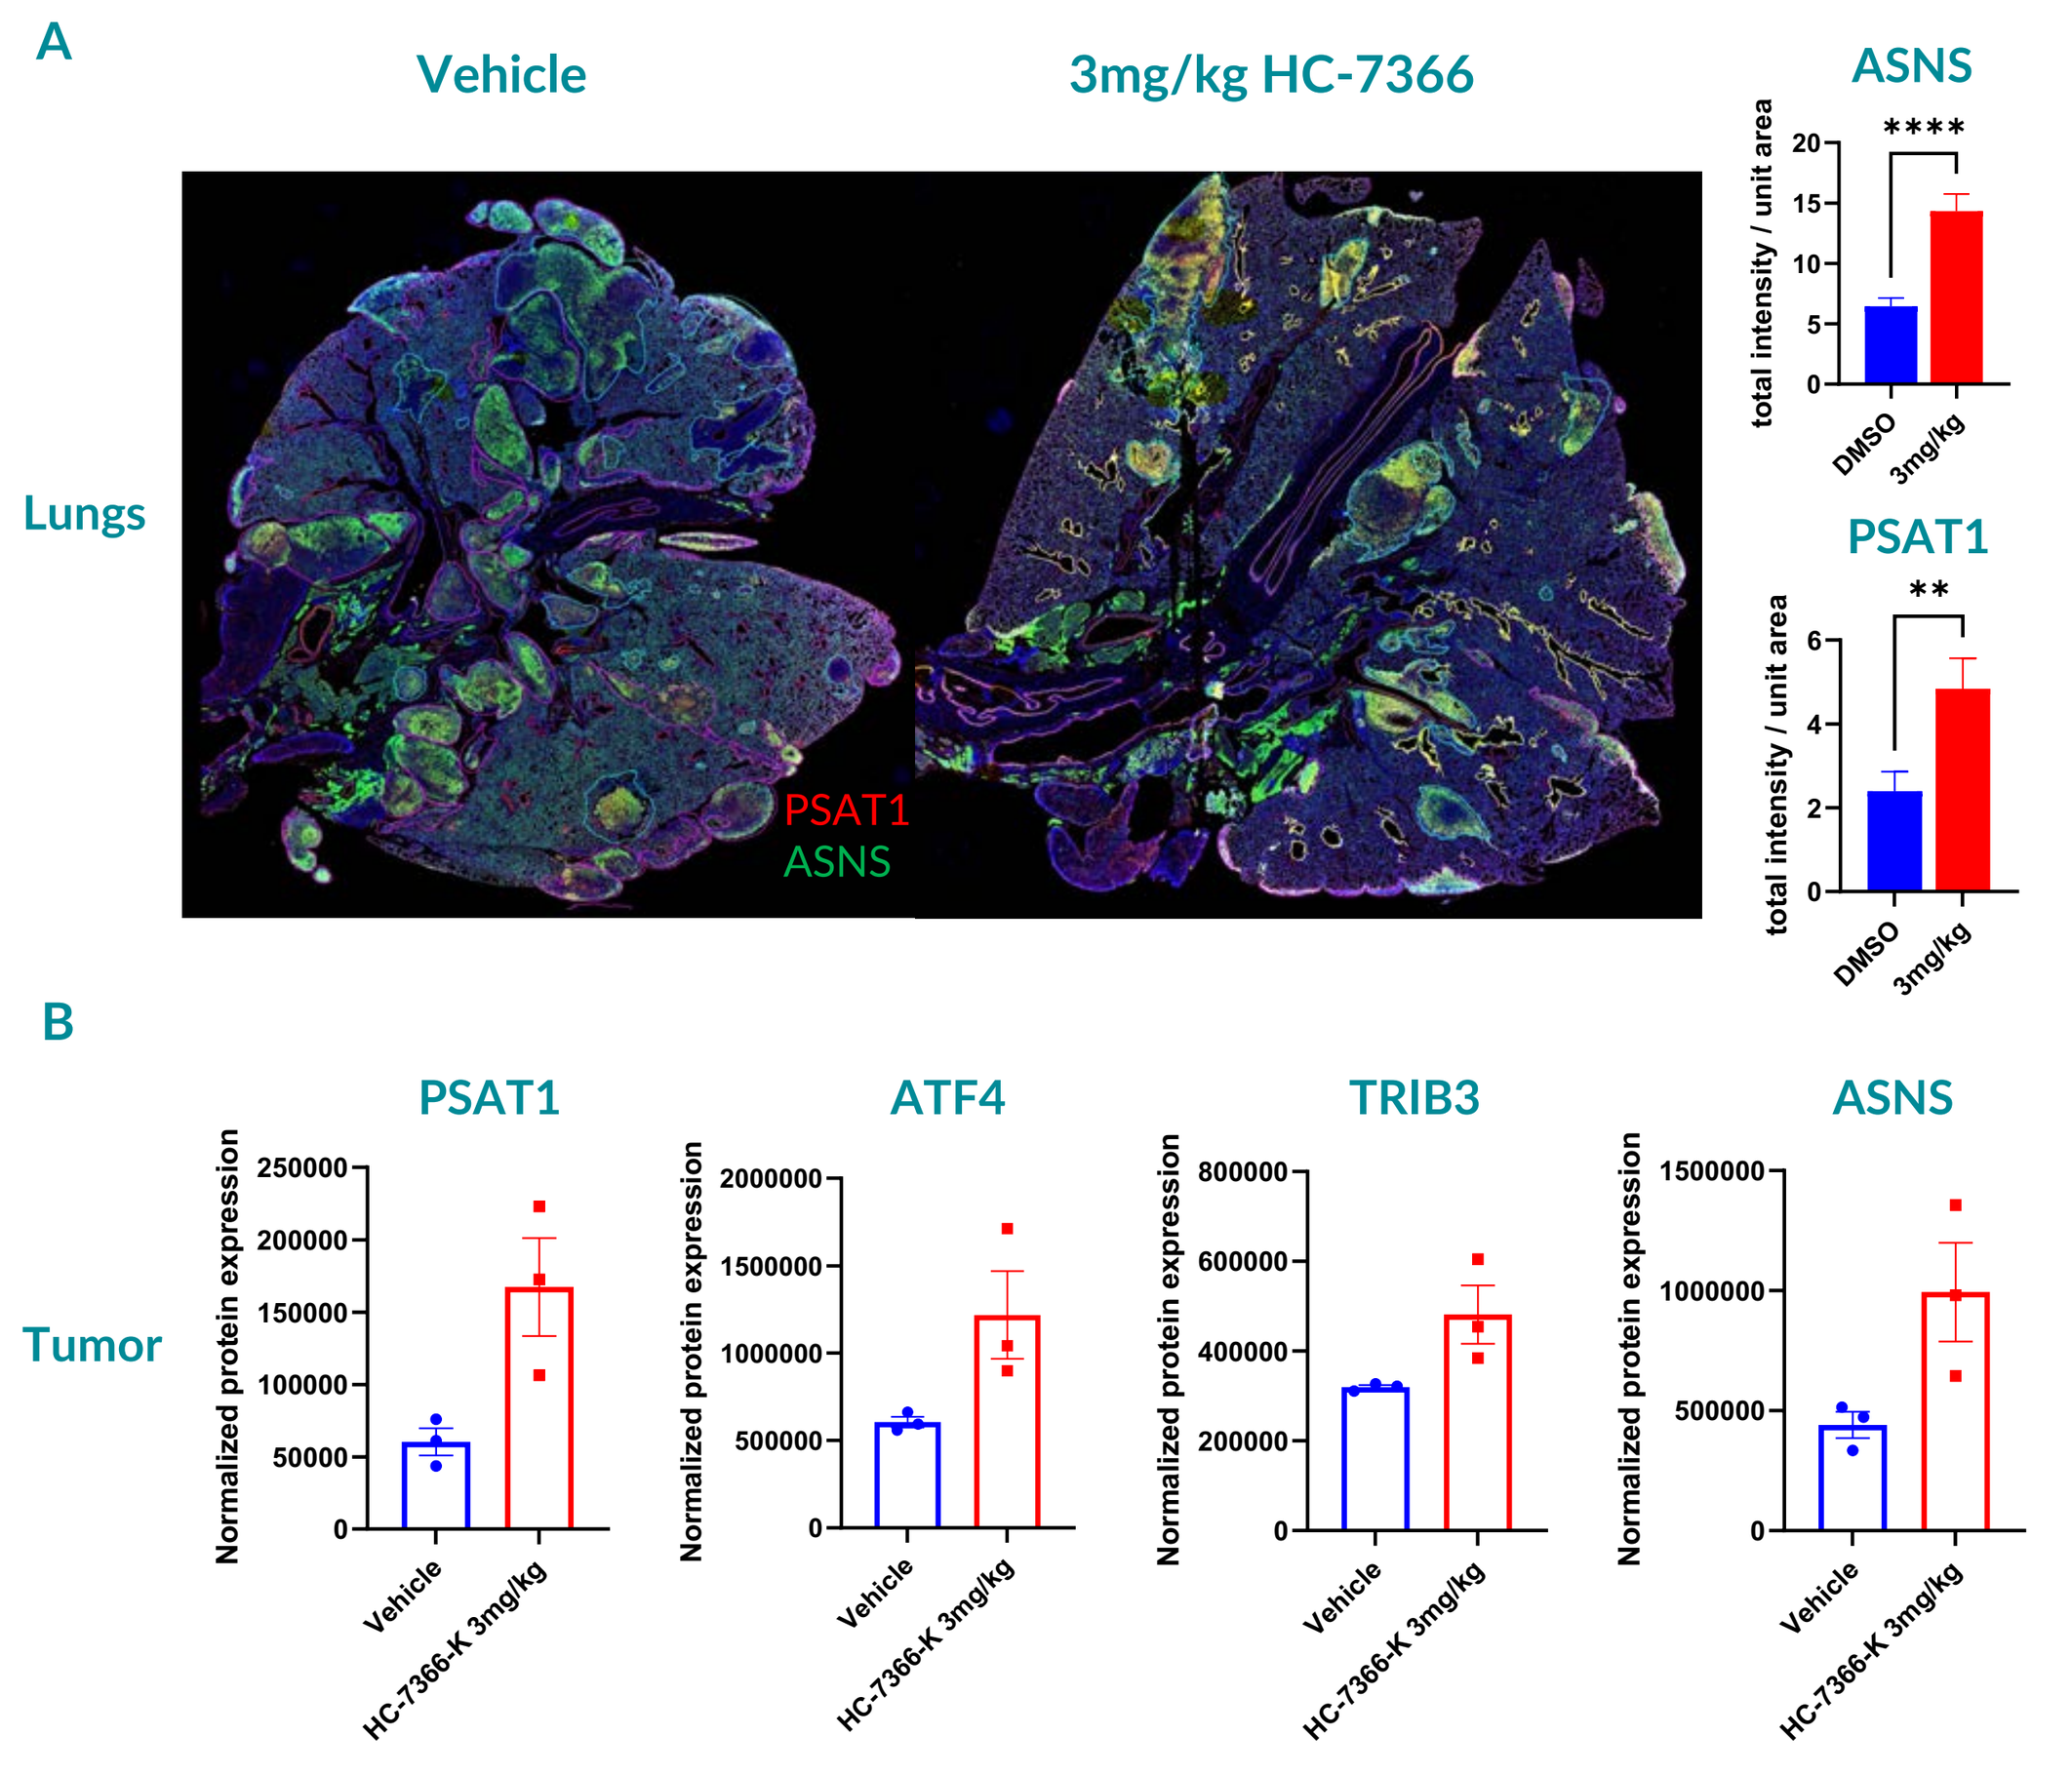
A
Vehicle
3mg/kg HC-7366
Lungs
PSAT1
ASNS
B
Tumor
PSAT1
ATF4
TRIB3
ASNS

Figure 4: HC-7366 Induces ISR Pathway Activation in Lung Metastases and Primary Tumor. A) Lungs from 4T1 tumor-bearing mice were collected, fixed, and paraffin embedded 21 days post HC-7366 treatment initiation. IHC was performed to measure expression of the ISR-induced amino acid biosynthesis proteins asparagine synthetase (ASNS) and phosphoserine aminotransferase 1 (PSAT1). Significant increases in both targets were observed with 3mg/kg HC-7366 treatment. **B)** Primary 4T1 tumors collected 7 days post treatment initiation were homogenized and measured via JESS for ISR-induced proteins. Increases in PSAT1, ATF4, TRIB3, and ASNS were observed with kg HC-7366 treatment.

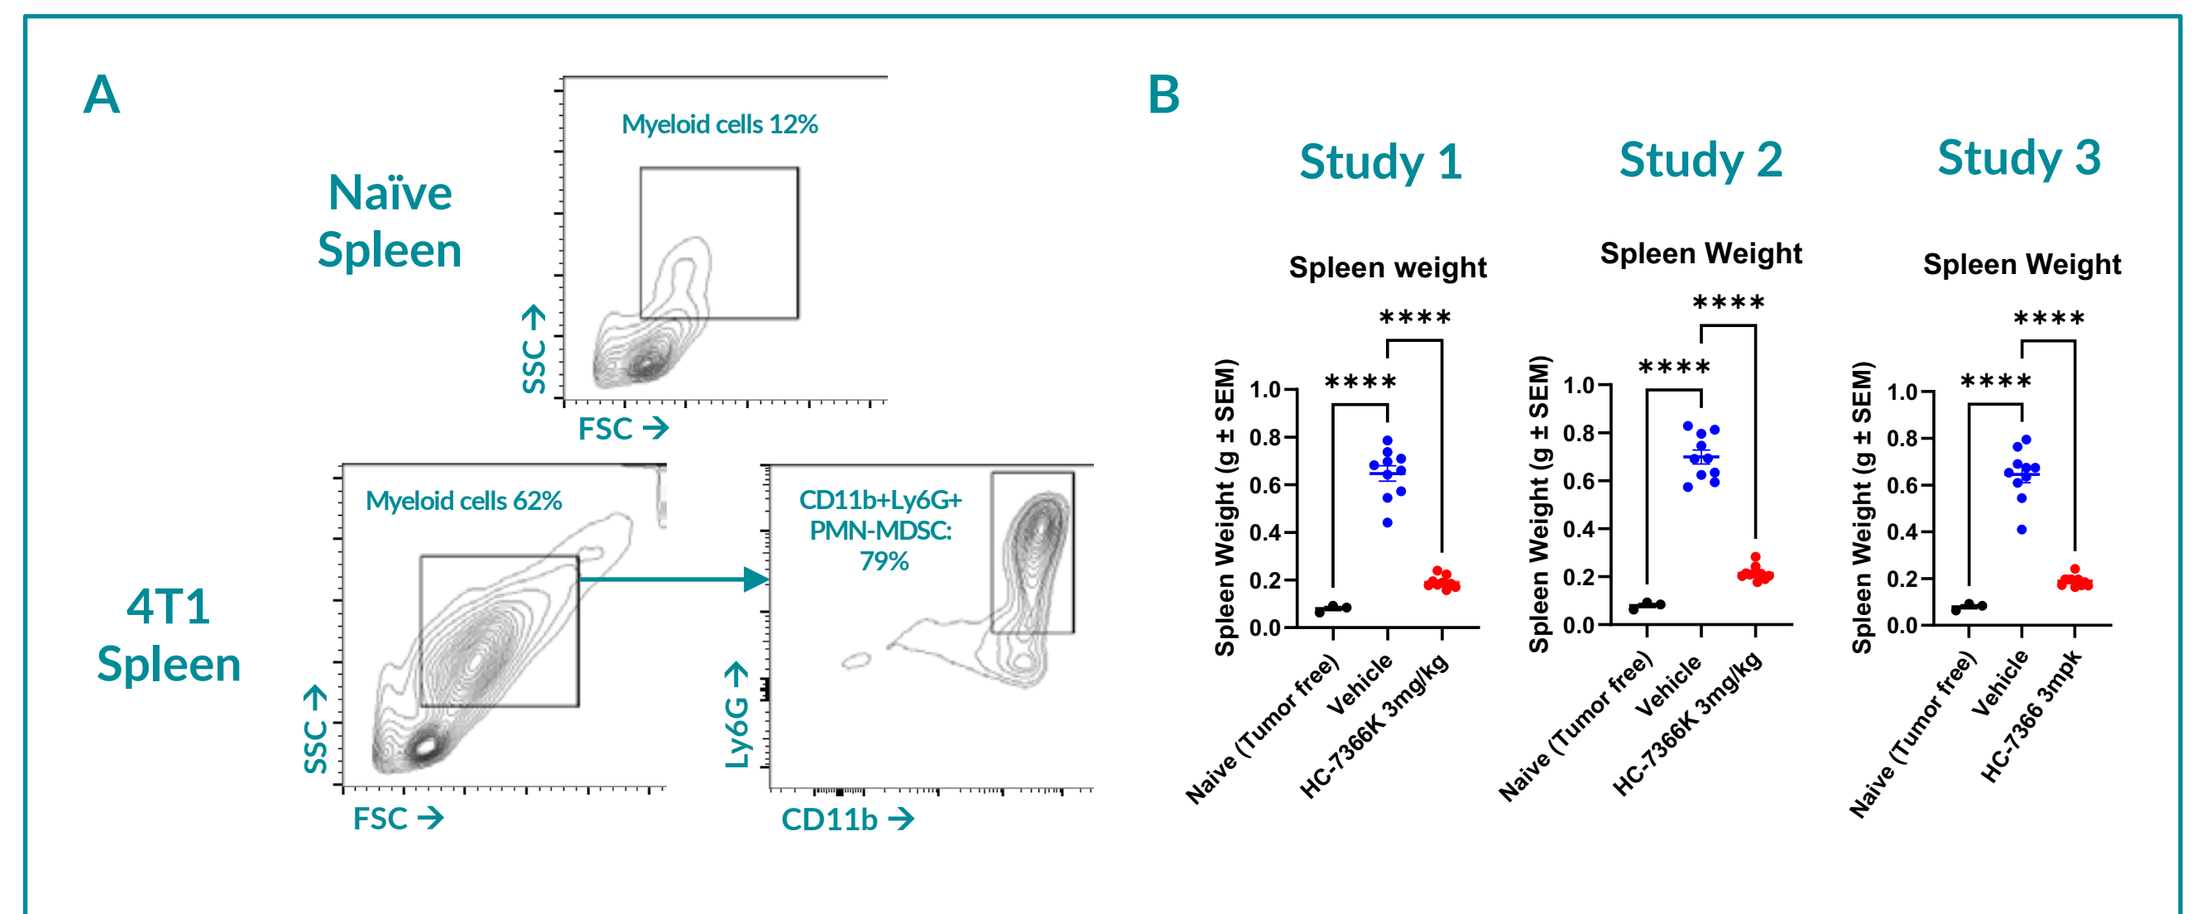
A
Naïve Spleen
Myeloid cells 12%
4T1 Spleen
Myeloid cells 62%
CD11b+Ly6G+ PMN-MDSC: 79%
B
Study 1
Study 2
Study 3
Spleen weight

Figure 5: HC-7366 Treatment Reduces 4T1-Induced Splenomegaly. A) Spleens in 4T1 tumor-bearing mice are enriched for CD11b+Ly6G+ PMN-MDSC. **B)** MDSC expansion in 4T1 mice dramatically increases spleen weight compared to naïve mice. Treatment with HC-7366 significantly decreases splenomegaly, especially at the activating 3mg/kg dose level. Graphs represent 3 independent studies.

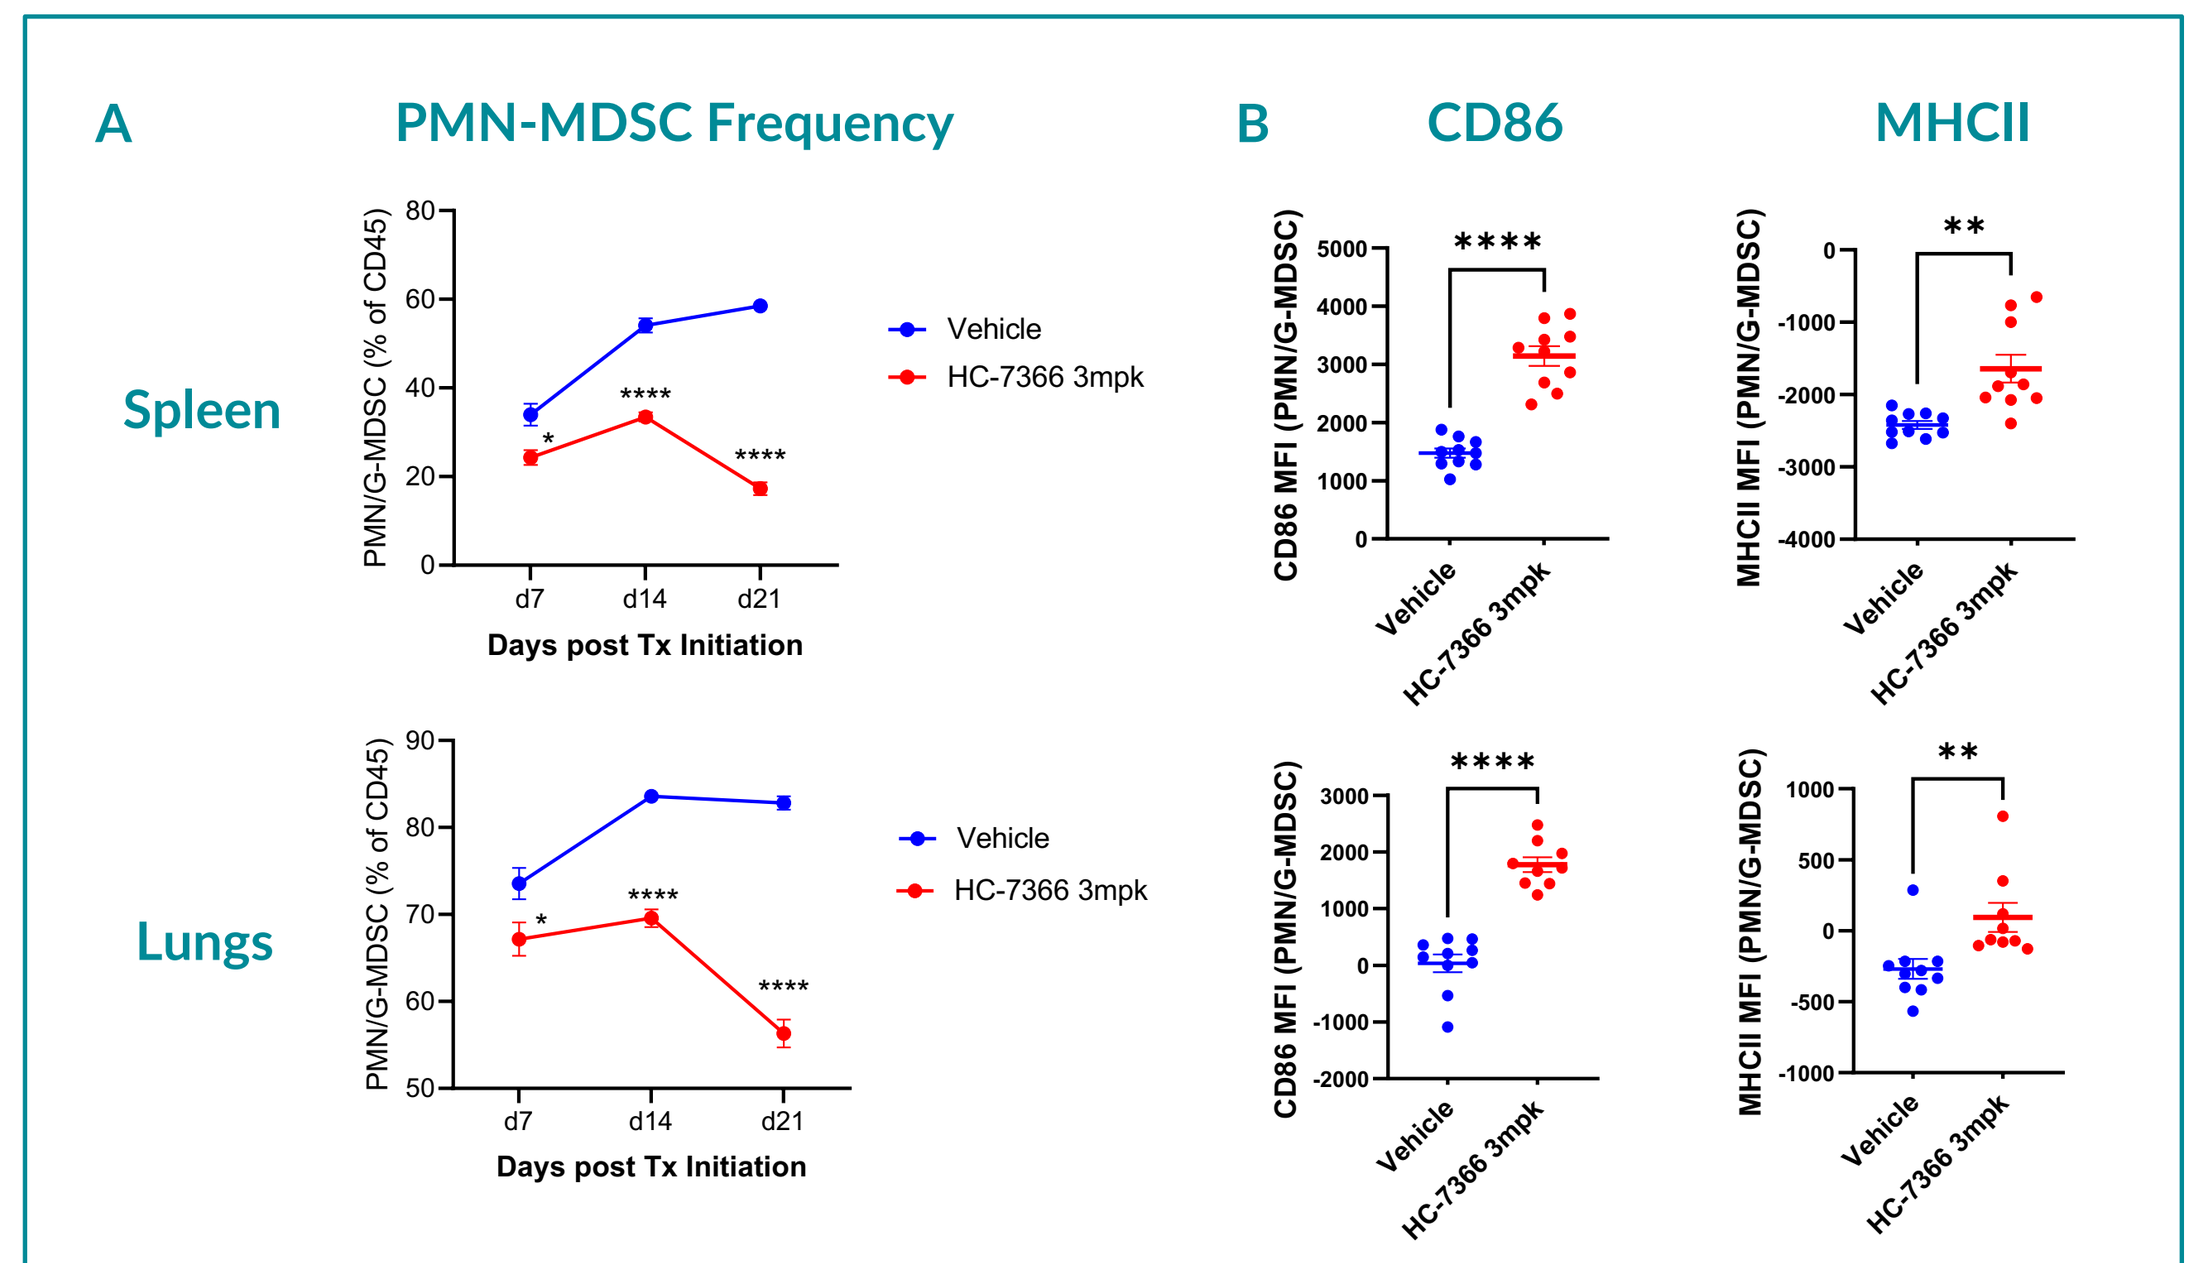
A
PMN-MDSC Frequency
Spleen
Lungs
Days post Tx Initiation
Vehicle
HC-7366 3mpk
B
CD86
MHCII

Figure 6: HC-7366 Reduces MDSC Frequency and Increases Expression of Activation Markers in the Spleen and Lungs. Single cell suspensions from spleens and lungs of HC-7366 treated 4T1 mice were evaluated via multi-color flow cytometry. **A)** MDSC frequency in both organs was significantly reduced with 3mg/kg HC-7366 treatment. **B)** The activation and co-stimulation markers MHCII and CD86 were significantly increased on MDSC in the spleen and lungs with 3mg/kg HC-7366 treatment (Day 14).

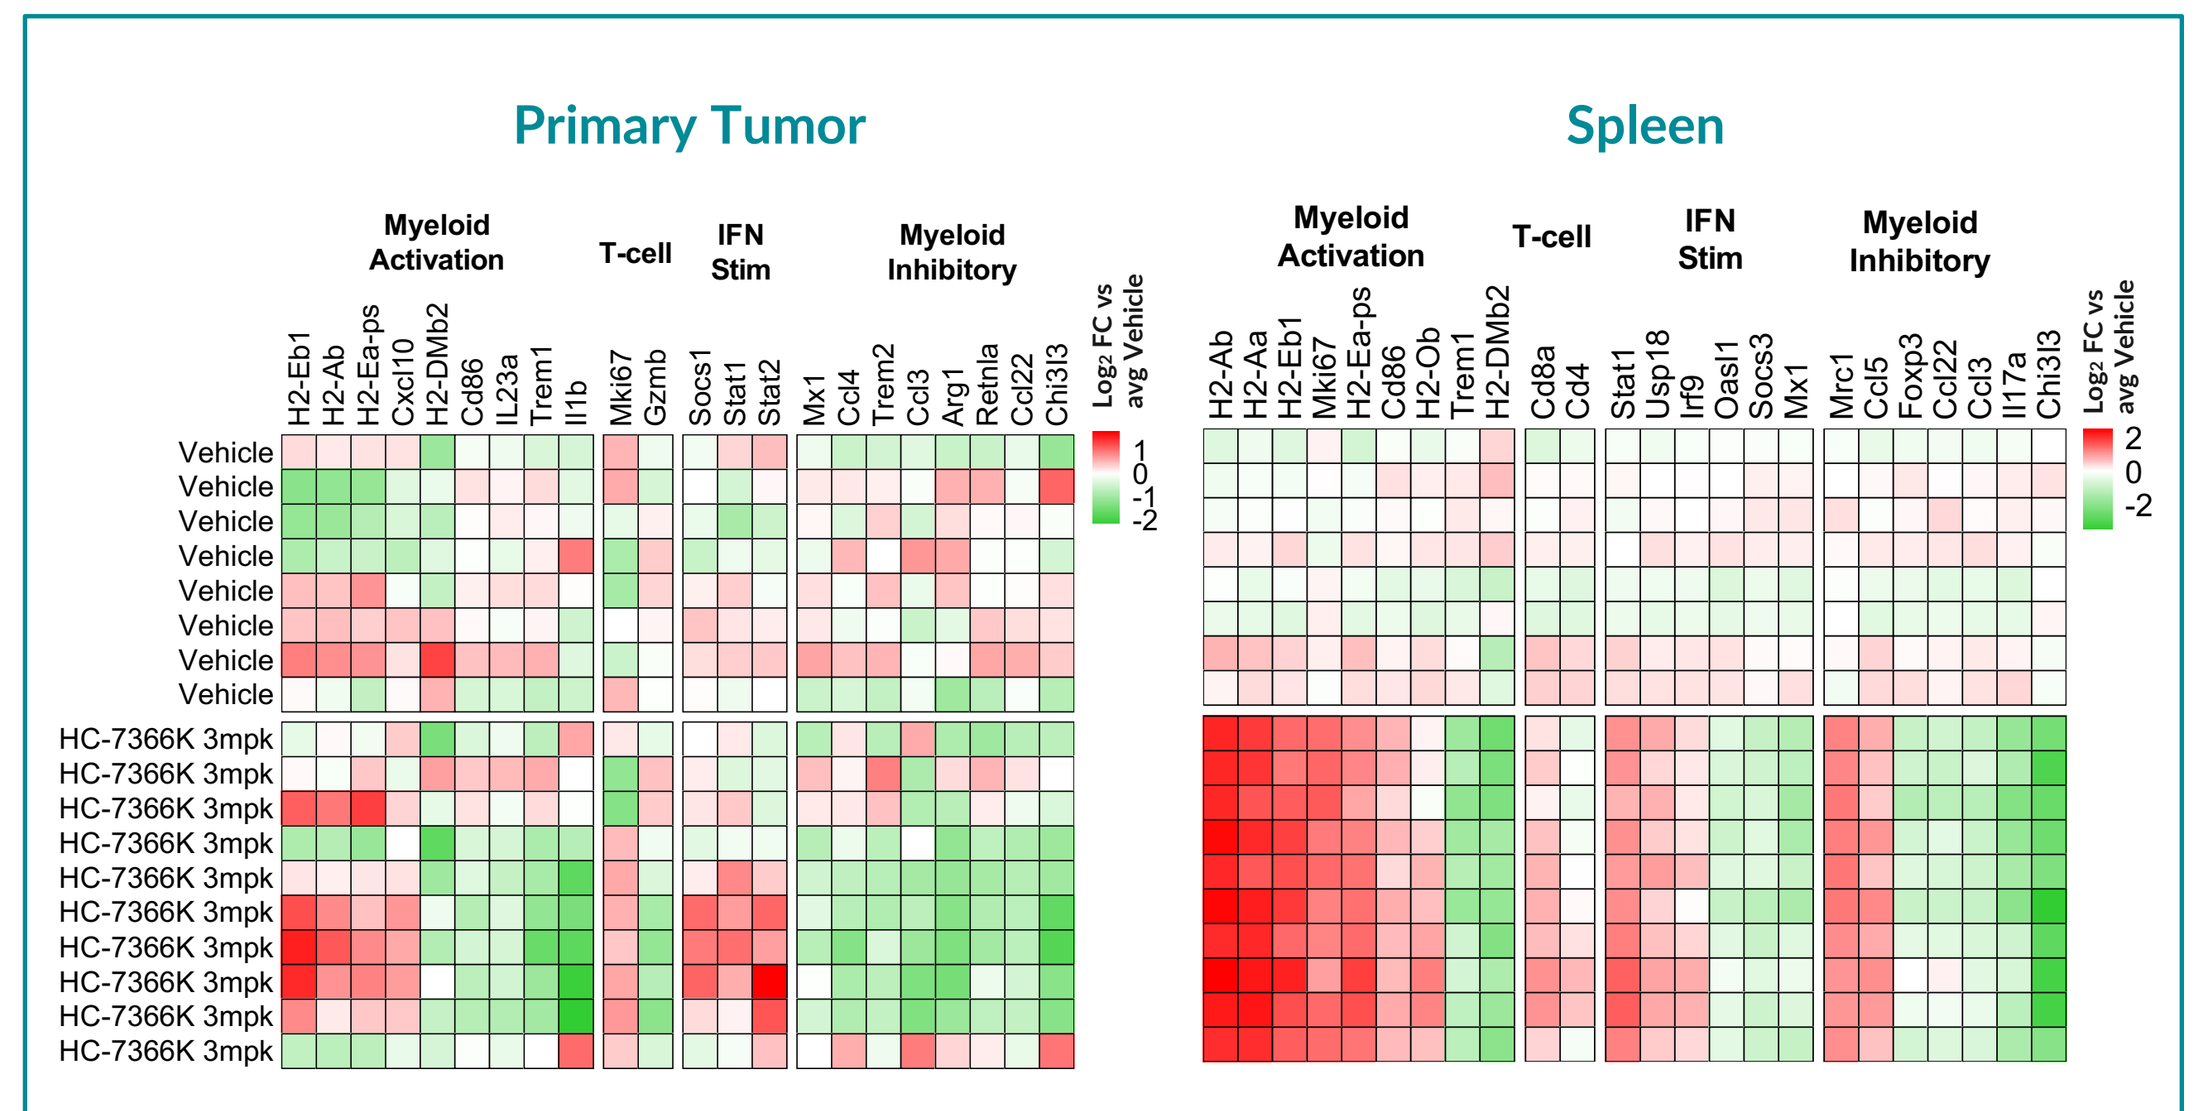
Primary Tumor
Spleen
Myeloid Activation
T-cell
IFN Stim
Myeloid Inhibitory

Figure 7: HC-7366-Induced Transcriptional Changes Indicate Immune Activation of Myeloid cells. RNA isolated from tumors and spleens of HC-7366 treated 4T1 mice was analyzed for gene expression via Quantigene (Day 21 of Tx). While tumor tissue did not show consistent effects, the MDSC-enriched spleens showed consistent increases in immune activation transcripts and reductions in inhibitory transcripts with 3mg/kg HC-7366. Rows = individual mice. 77 genes surveyed, differentially expressed genes are shown.

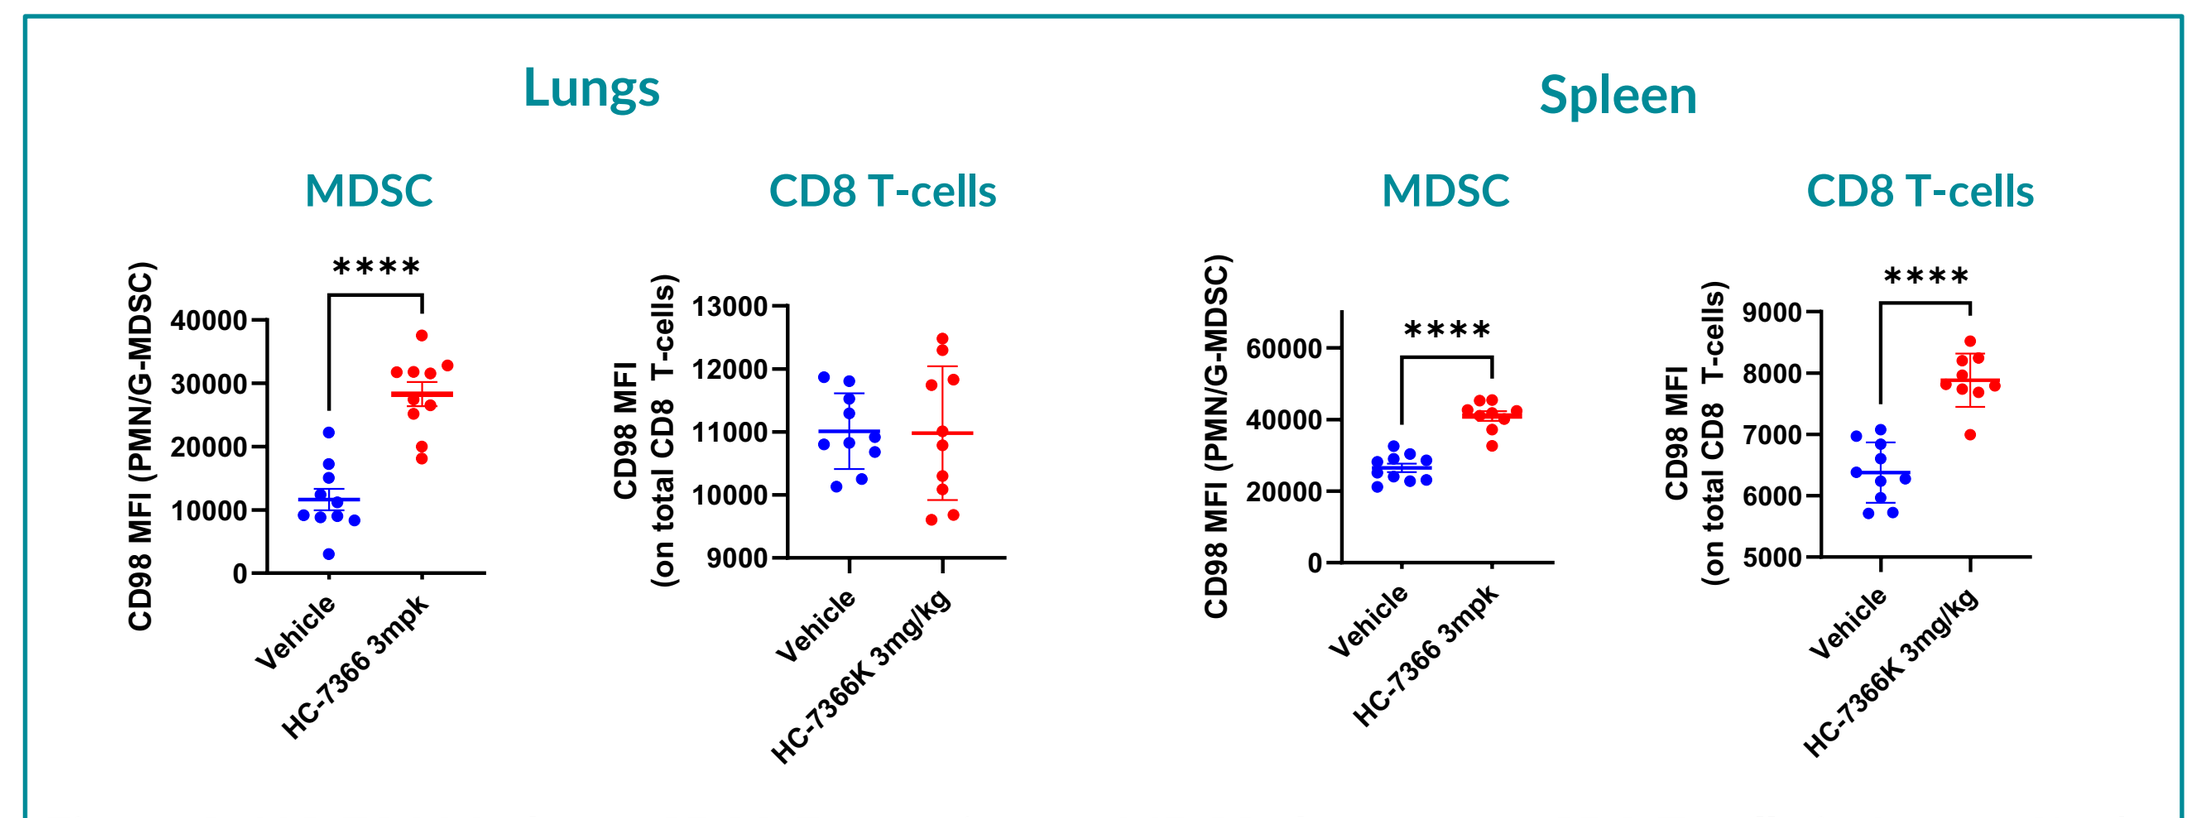
Lungs
Spleen
MDSC
CD8 T-cells
MDSC
CD8 T-cells

Figure 8: HC-7366 Induces CD98 Expression on MDCSs but not on CD8 T-cells in Lungs. Single cell suspensions from lungs and spleens of 4T1 mice 21 days post-Tx initiation were evaluated for CD98 expression via flow cytometry. CD98 is downstream of ATF4 and forms a complex with LAT1 on the cell surface to mediate import of essential amino acids. 3mg/kg HC-7366 treatment significantly increased expression of CD98 on MDSC, indicating ISR pathway activation, but not on CD8 T-cells in the lungs, while both cell subsets showed increased CD98 expression in the spleen.

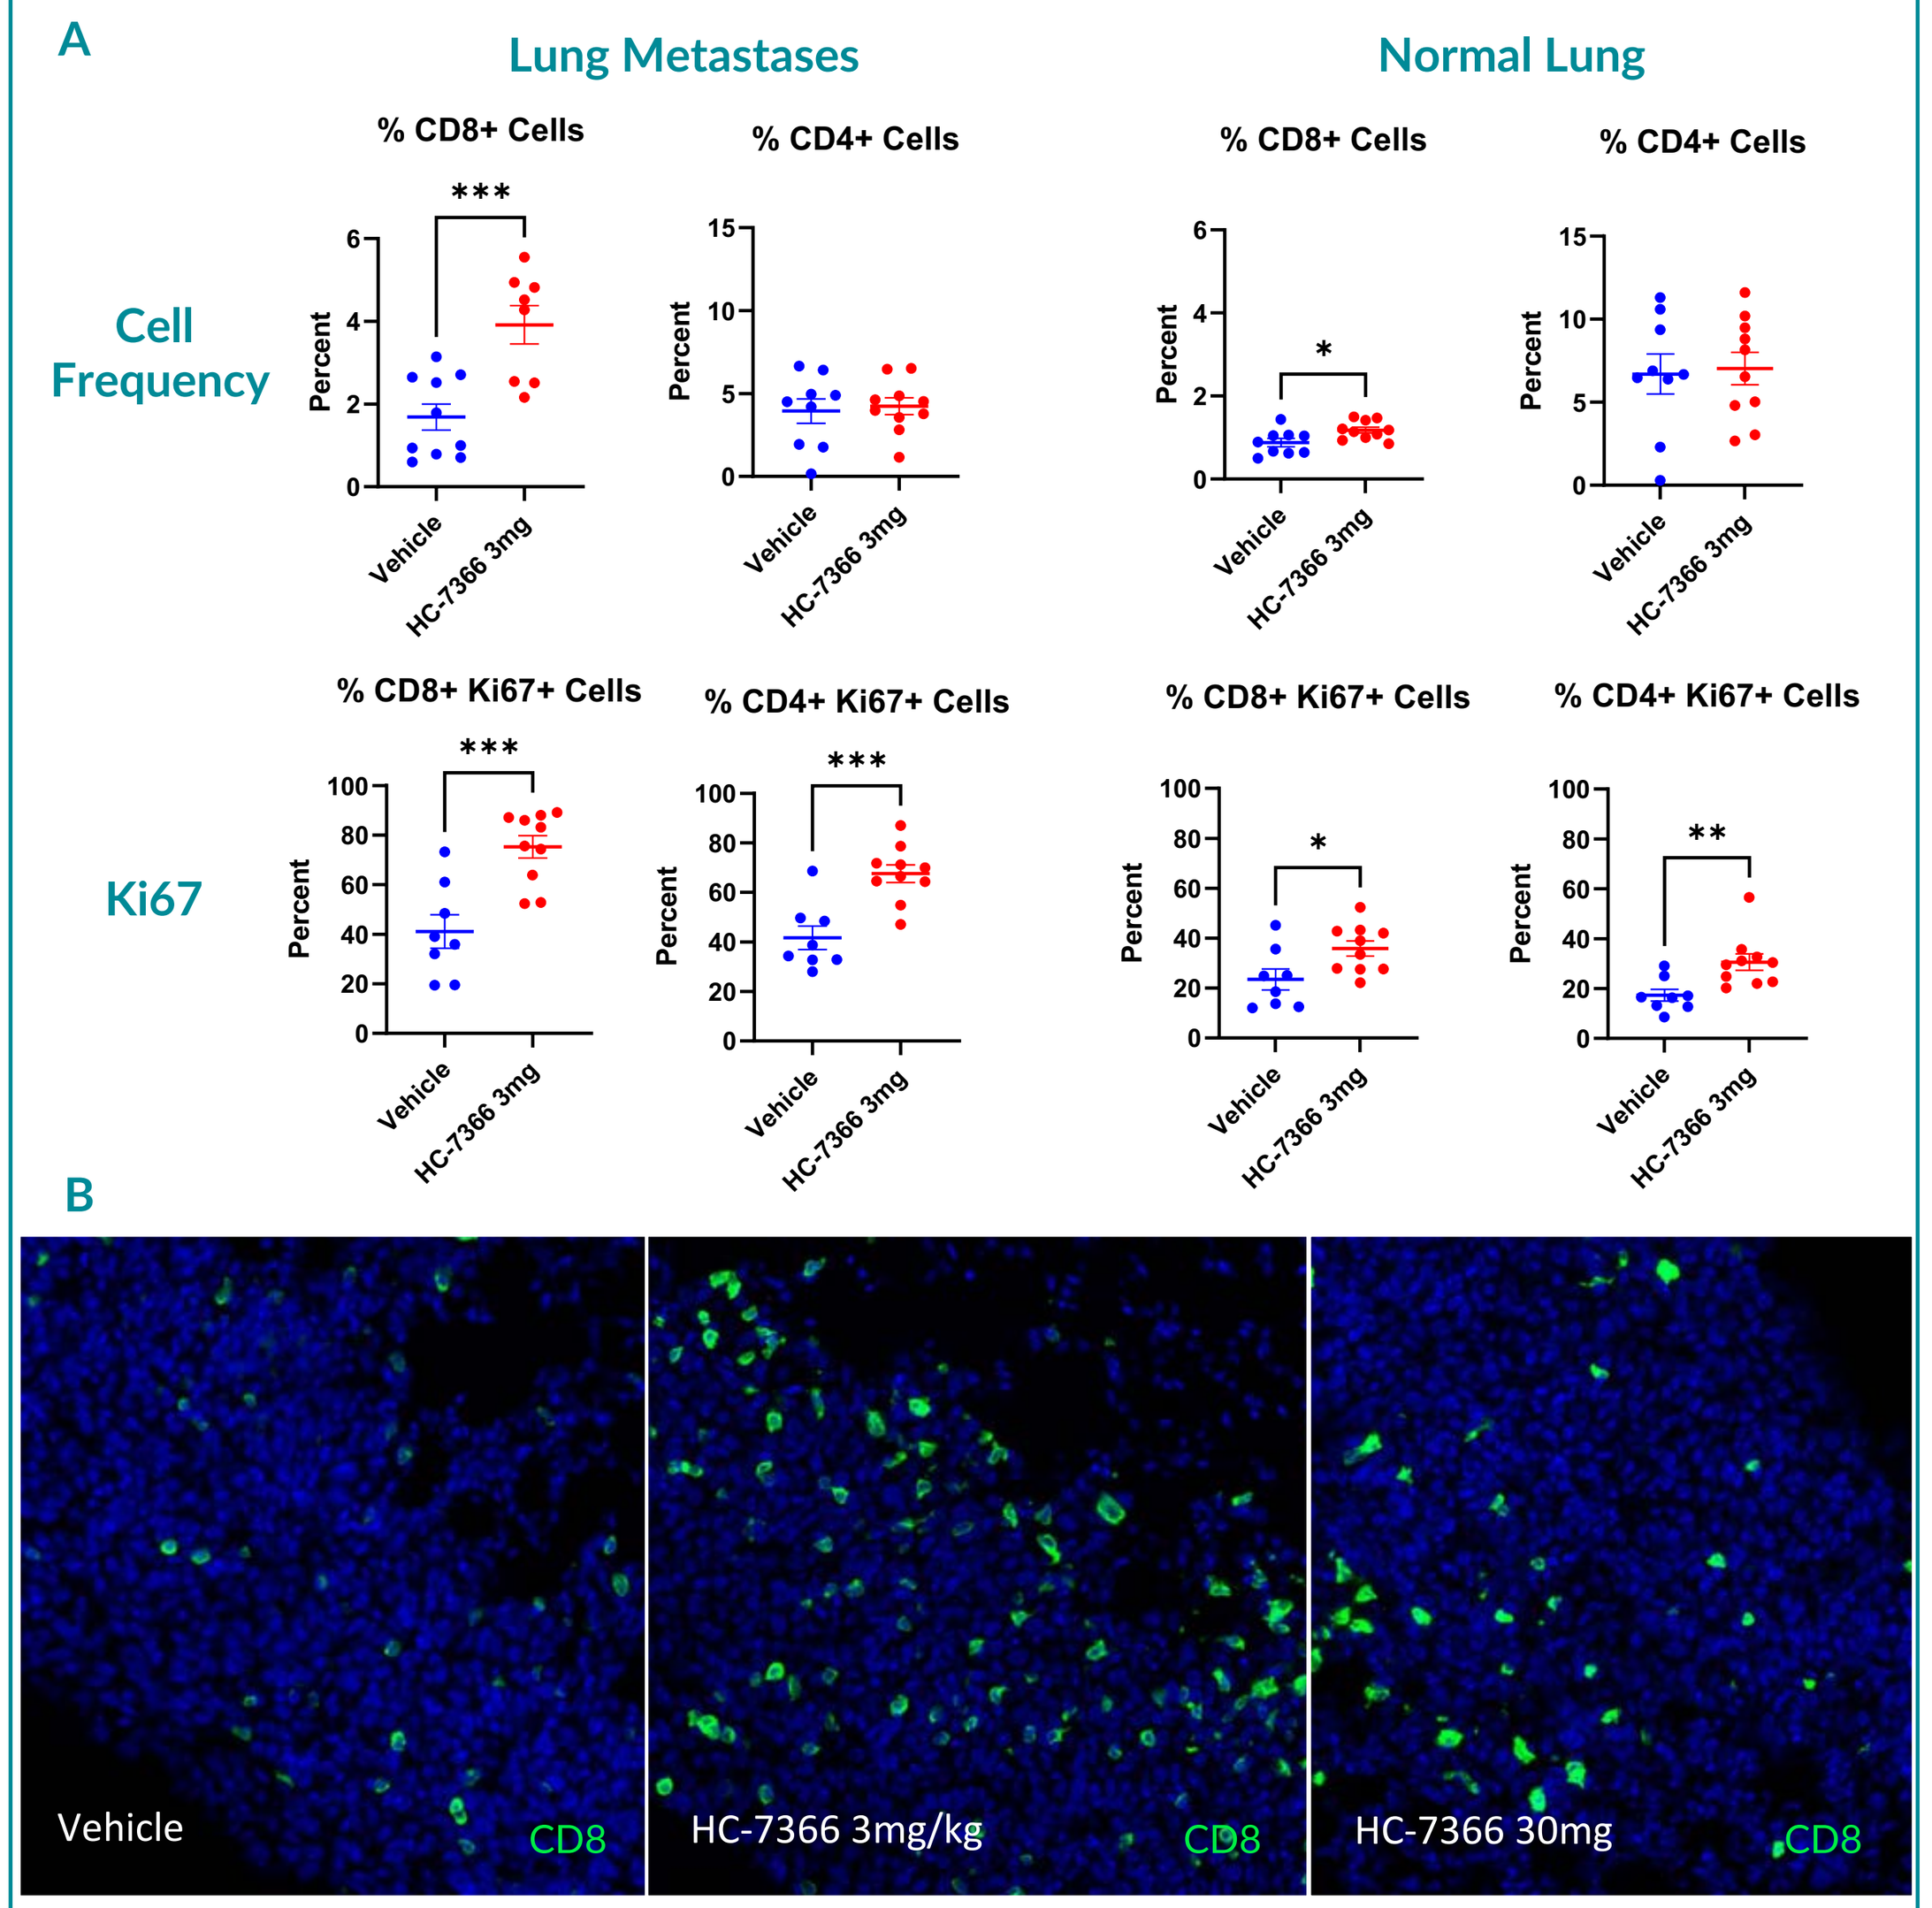
A
Lung Metastases
Normal Lung
Cell Frequency
Ki67
B
Vehicle
CD8
HC-7366 3mg/kg
HC-7366 30mg

Figure 9: HC-7366 Increases CD8 T-cells and T-cell proliferation in Pulmonary Metastases. IHC was performed on FFPE lungs collected from 4T1 mice treated with HC-7366 (Day 21 of Tx). Samples were stained for CD4, CD8, and Ki67. **A)** CD8 T-cells were significantly increased in metastatic lesions and to a lesser degree in normal tissue. Both CD4 and CD8 T-cells showed significantly increased Ki67 expression, which was greater in metastatic lesions than in normal tissue. **B)** Representative CD8 T-cell staining is shown.

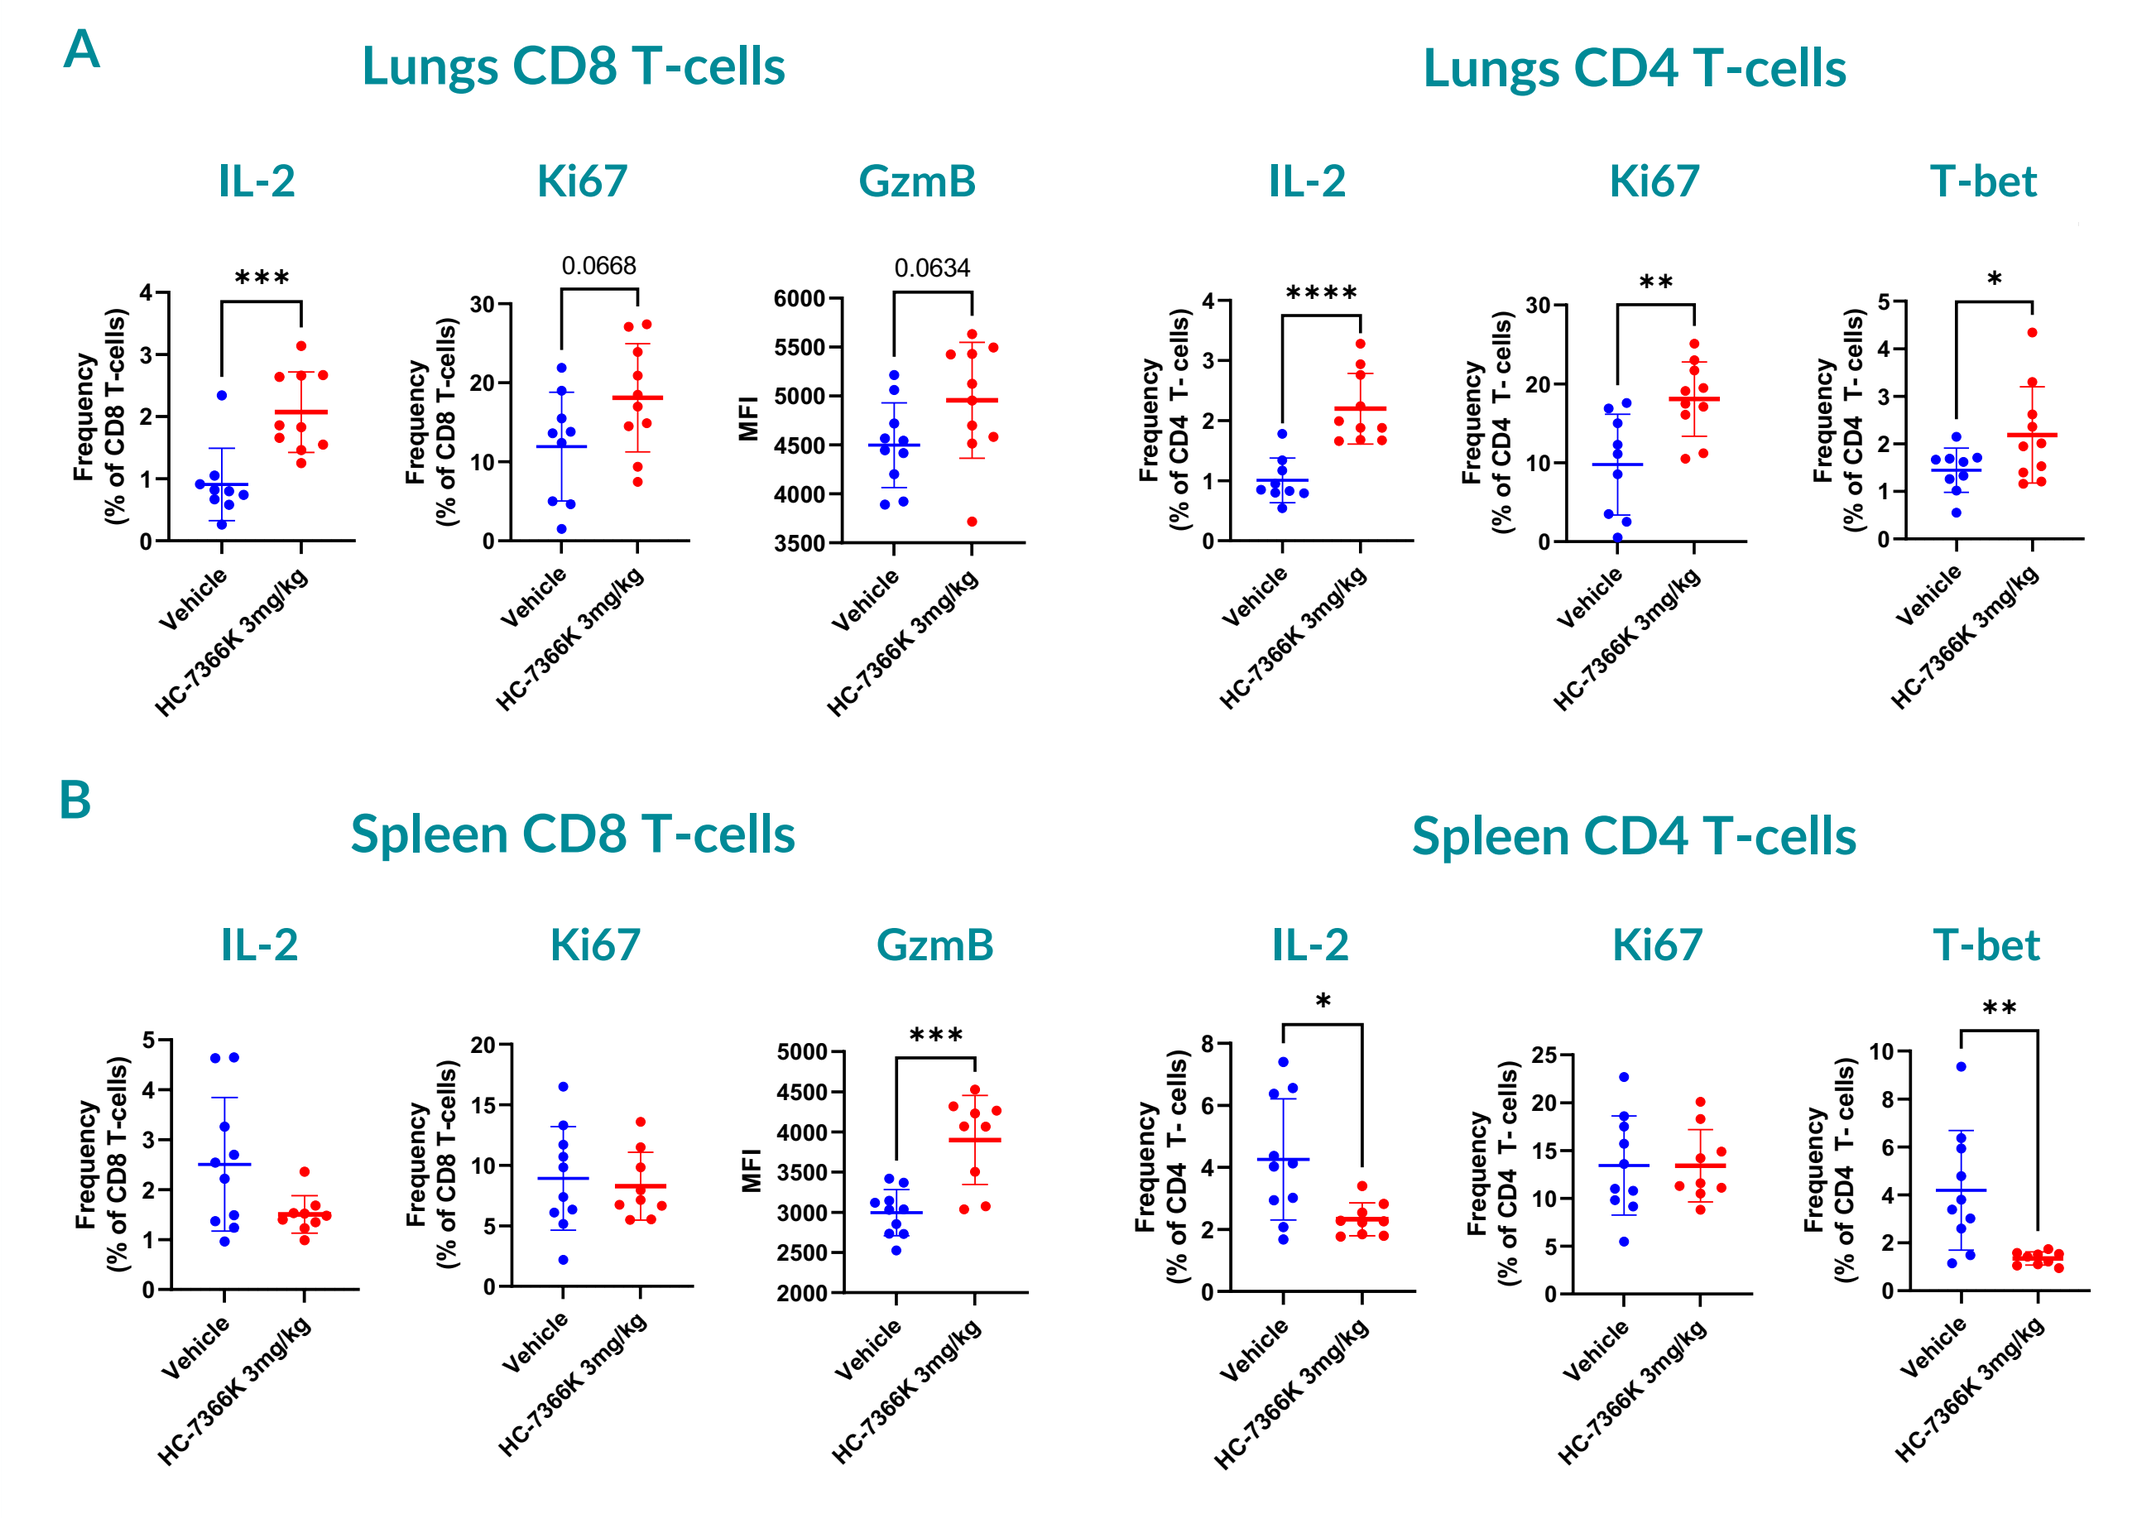
A
Lungs CD8 T-cells
Lungs CD4 T-cells
IL-2
Ki67
GzmB
T-bet
B
Spleen CD8 T-cells
Spleen CD4 T-cells

Figure 10: HC-7366 Increases T-cell Activation in Lungs. Single cell suspensions from lungs and spleens of HC-7366 treated 4T1 mice were evaluated via intracellular flow cytometry (Day 21 of Tx). CD4 and CD8 T-cells showed significantly increased activation markers in lungs **(A)**, but not in spleens **(B).**

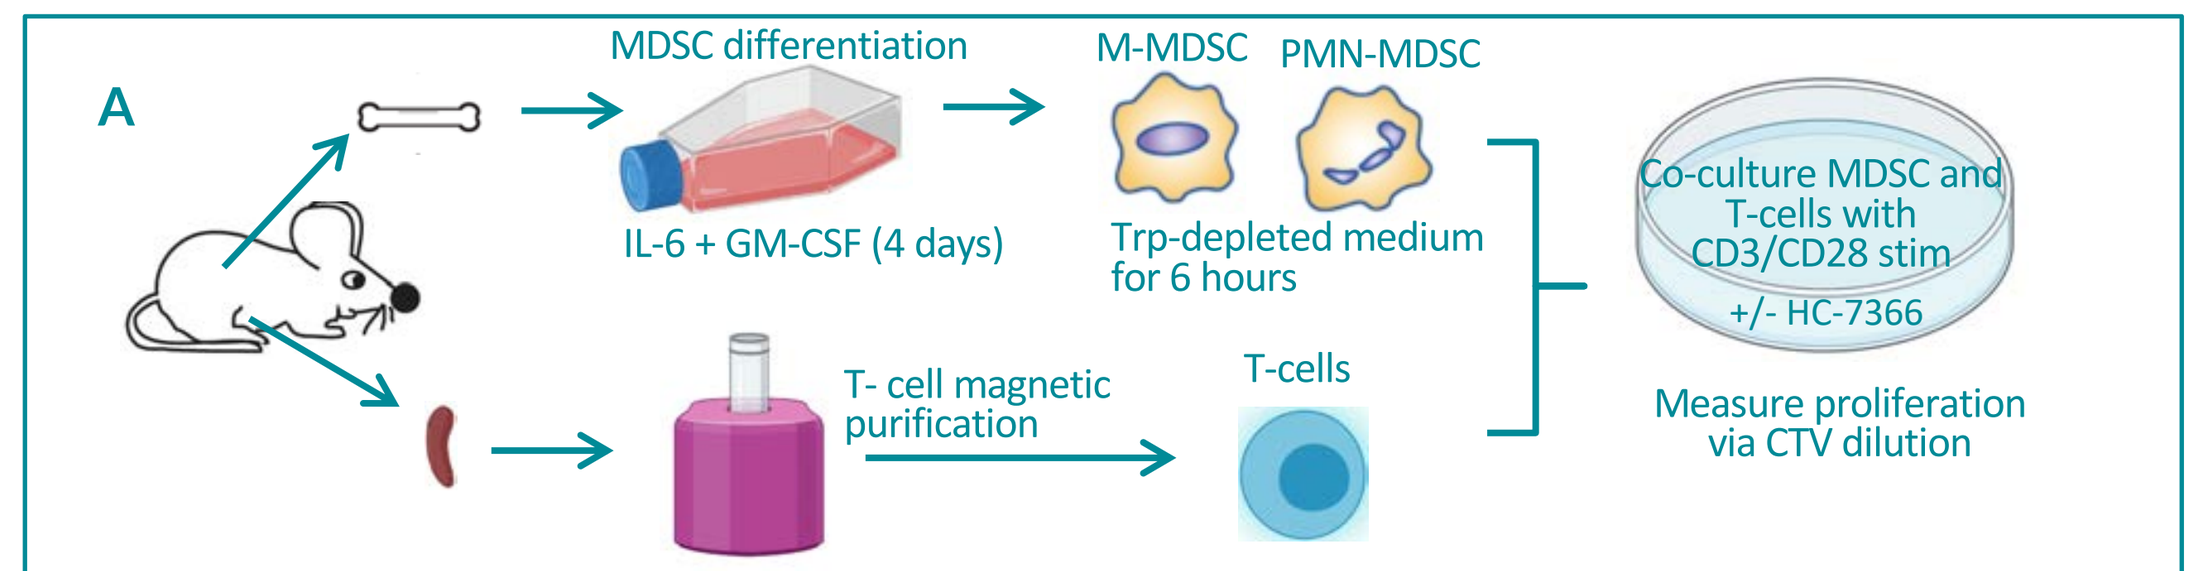
A
MDSC differentiation
IL-6 + GM-CSF (4 days)
M-MDSC
PMN-MDSC
Trp-depleted medium for 6 hours
Co-culture MDSC and T-cells with CD3/CD28 stim +/- HC-7366
T- cell magnetic purification
T-cells
Measure proliferation via CTV dilution

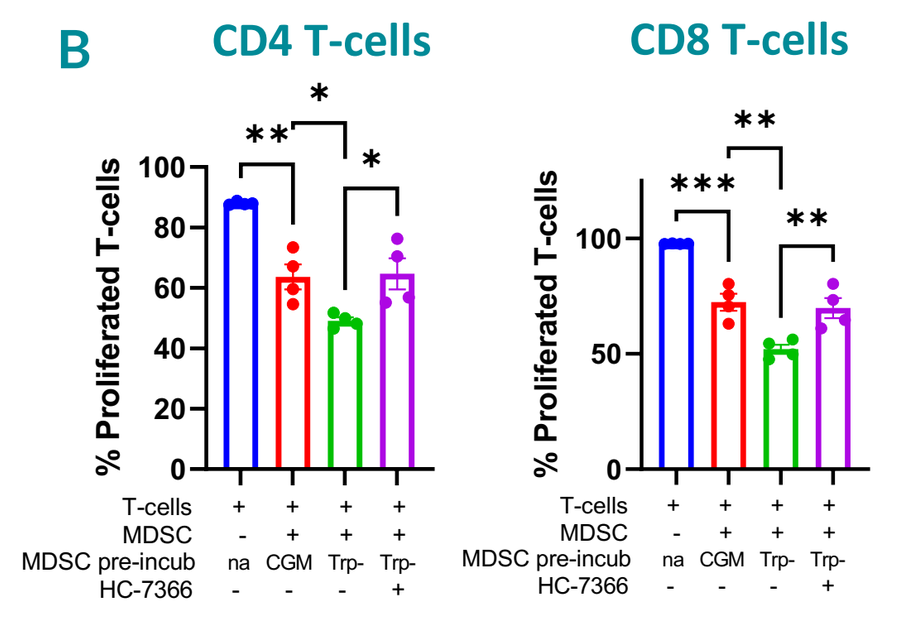
B
CD4 T-cells
CD8 T-cells
% Proliferated T-cells

Figure 11: ***Ex Vivo*** **Treatment of Bone Marrow-Derived MDSC Reduces Suppressive Activity. (A)** MDSC were differentiated from mouse bone marrow and pre-incubated with tryptophan-depleted (Trp-) or complete growth medium (CGM) before co-culture with stimulated T-cells +/- 0.1µM HC-7366. **(B)** Proliferation of CD4 or CD8 T-cells was measured by CTV dilution using flow cytometry. HC-7366 treatment significantly rescued T-cell proliferation in the presence of BM-derived MDSC.

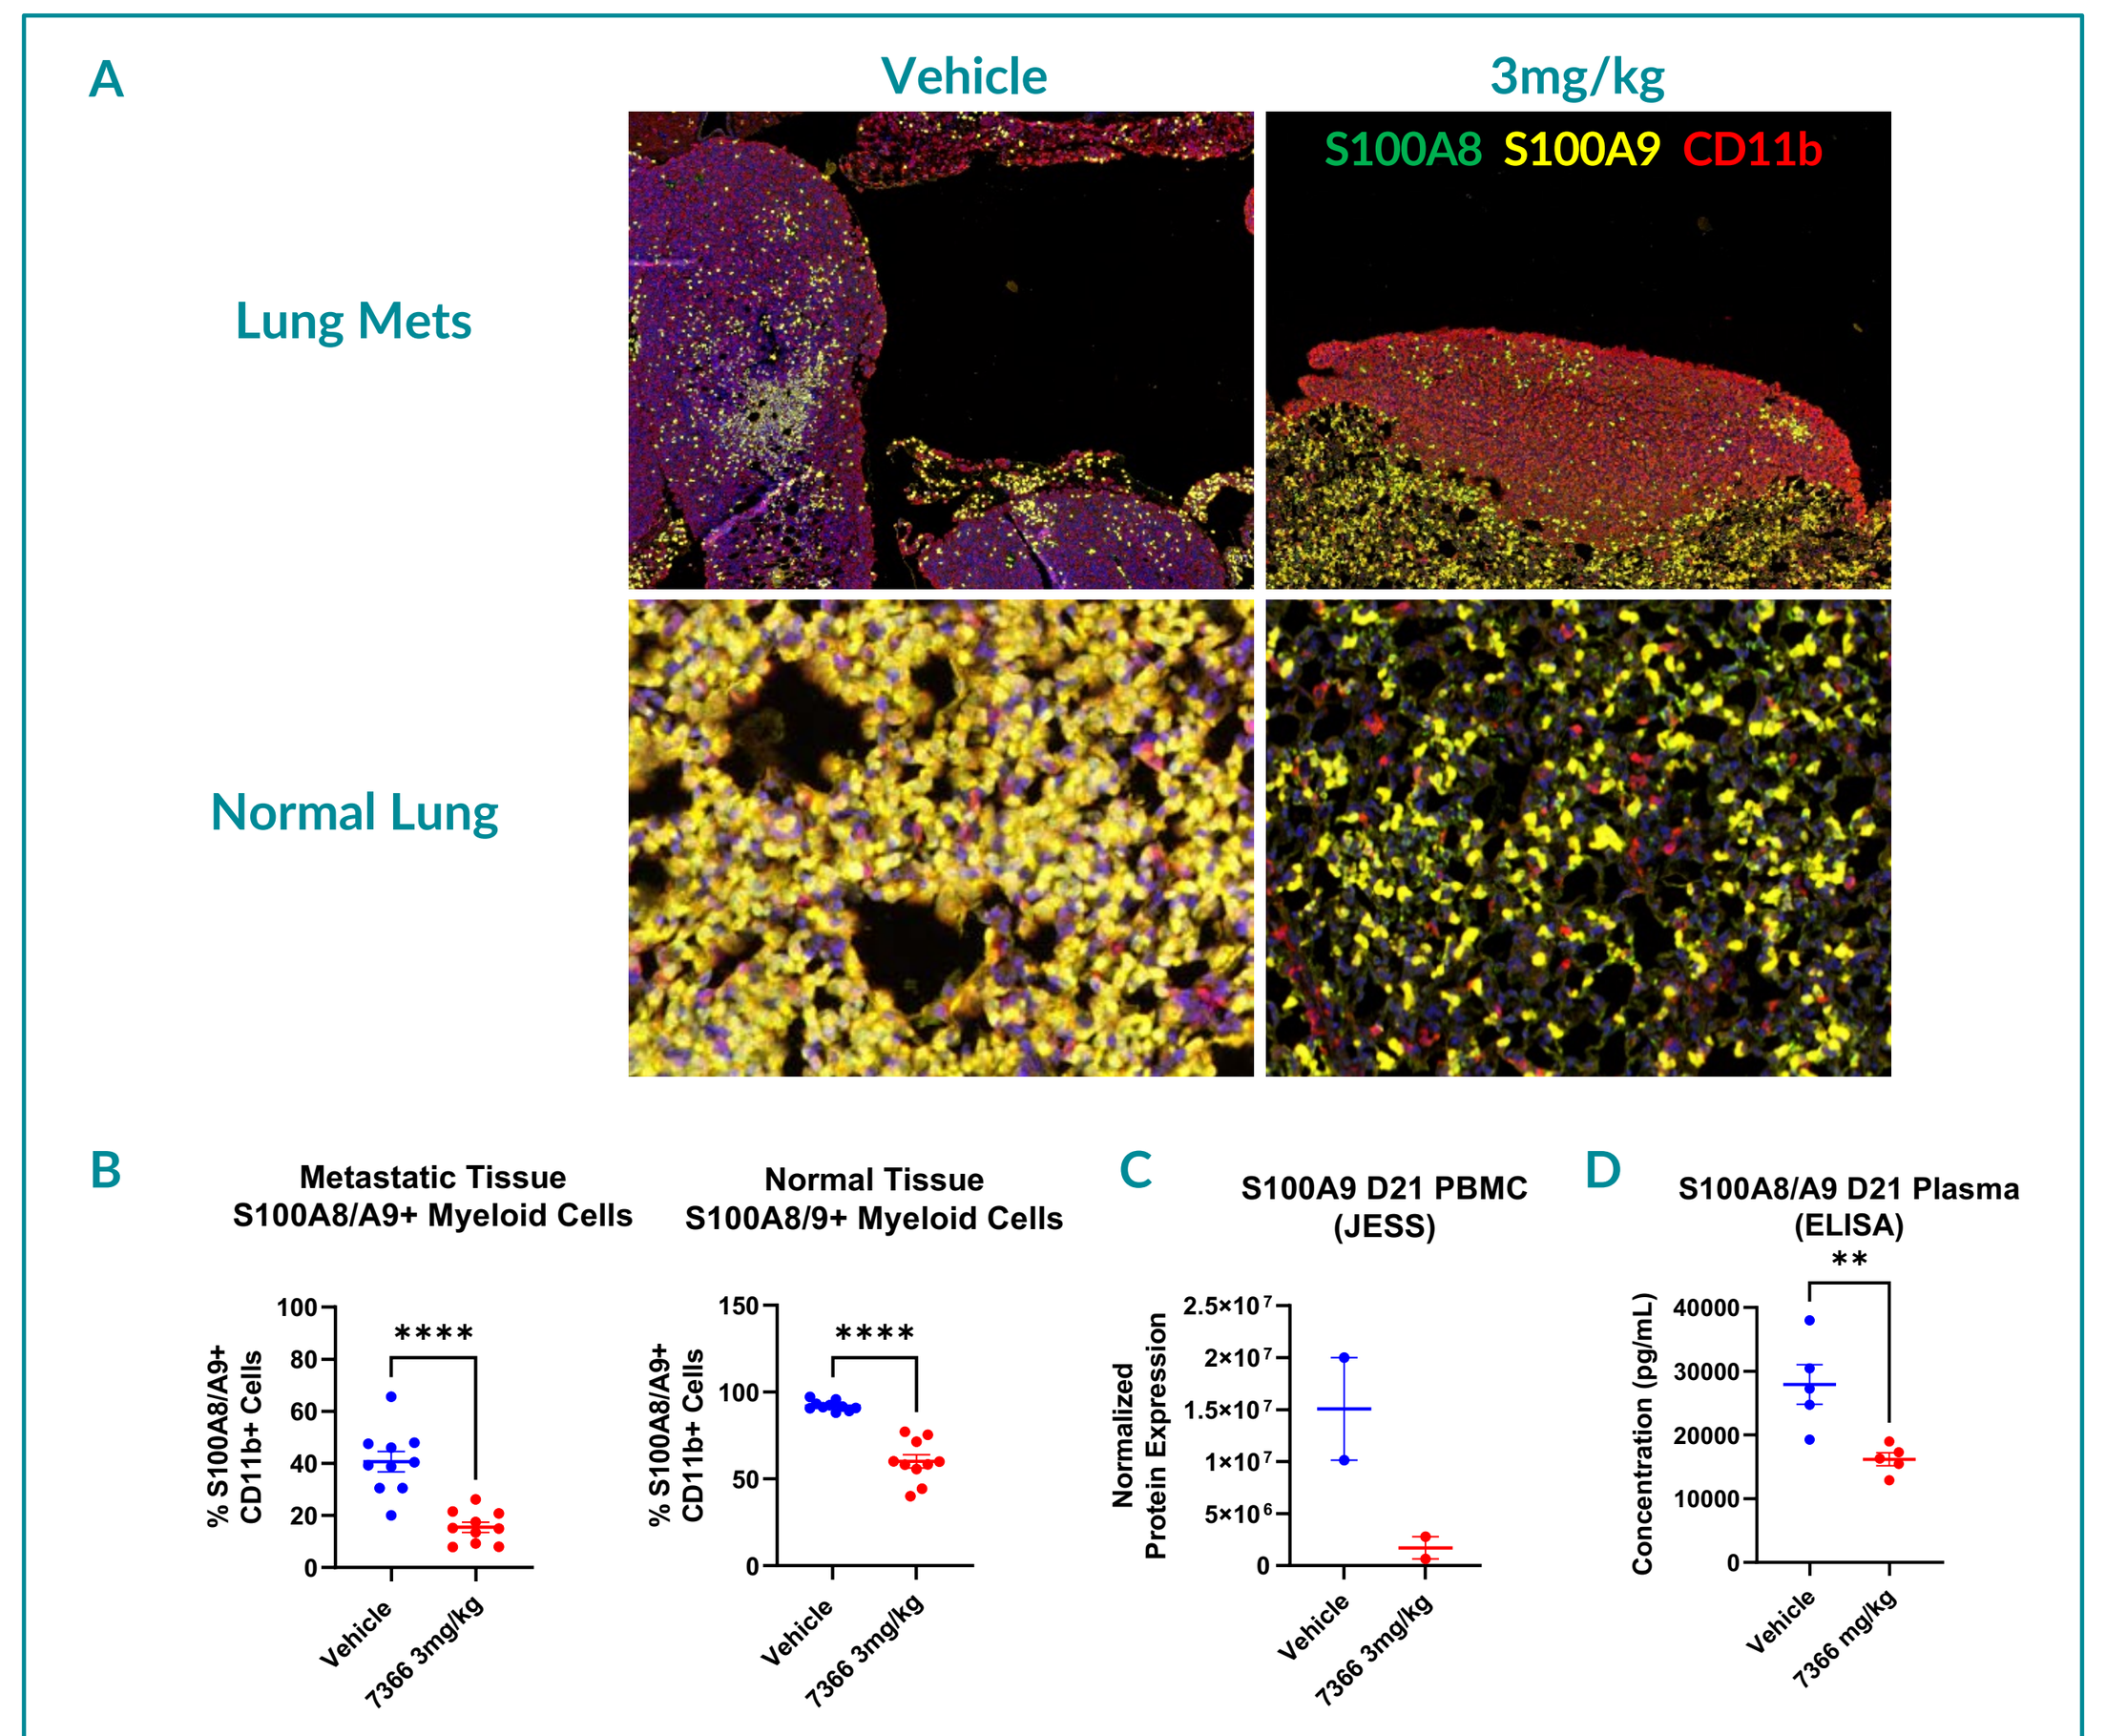
A
Vehicle
3mg/kg
S100A8 S100A9 CD11b
Lung Mets
Normal Lung
B
Metastatic Tissue S100A8/A9+ Myeloid Cells
Normal Tissue S100A8/9+ Myeloid Cells
C
S100A9 D21 PBMC (JESS)
D
S100A8/A9 D21 Plasma (ELISA)

Figure 12: HC-7366 Reduces S100A8/A9 Proteins in Lung Myeloid Cells, PBMCs, and Plasma of 4T1 Mice. S100A8/A9 proteins are constitutively expressed in myeloid cells, and their overexpression in has been shown to increase trafficking, expansion, and activation of MDSC to establish an immunosuppressive microenvironment and promote metastatic spread [3-5]. S100A8/A9 proteins were measured by IHC in metastatic and normal lung tissue **(A-B)**, in PBMCs by JESS **(C)**, and in plasma by ELISA **(D)**. Significant reductions in S100 protein levels were observed in each case (statistics were not performed on JESS analysis, as multiple PBMC samples were pooled for two each data point).

SUMMARY/CONCLUSIONS

- The GCN2 modulator HC-7366 showed anti-metastatic efficacy in the lungs of 4T1 tumor-bearing mice that correlated with ISR activation in primary tumor and metastases.
- HC-7366 substantially reduced MDSC frequency and reduced their immunosuppressive phenotype *in vivo* and *in vitro*.
- HC-7366's effect on MDSC was correlated with increased T-cell infiltration, activation and proliferation in metastatic lesions.
- The ISR downstream AA transport protein CD98 was increased in MDSC but not on CD8 T-cells in lung tissue, potentially indicating greater ISR induction in MDSC than in T-cells.
- HC-7366 reduced S100A8/A9 protein levels in lungs, PBMCs, and plasma, indicating a potential mechanistic or PD marker for HC-7366 activity.
- Studies are ongoing to further investigate the link between HC-7366's effects on MDSC and its anti-metastatic activity.
- Taken together, these data demonstrate the potential of HC-7366 to treat MDSC-enriched metastatic cancers.

ACKNOWLEDGEMENTS

- We would like to thank Amy Swearingen and Nicole Meinhardt (HiberCell) for assistance in preparation of this poster.
- We would also like to thank the HiberCell Chemistry Manufacturing and Controls team for assistance with all compound-related work.

REFERENCES

1. Pakos-Zebrucka K, *et al.* (2016) EMBO Reports 17: 1374–1395. doi: 10.15252/embr.201642195
2. Trovato R, *et al.* (2020) Front. Oncol. 10:165. doi: 10.3389/fonc.2020.00165
3. Sinha P, *et al.* (2008) J Immunol. 181(7):4666-4675. doi: 10.4049/jimmunol.181.7.4666
4. Huang M, *et al.* (2019). Front. Immunol. 10:2243. doi: 10.3389/fimmu.2019.02243
5. Wang, L, *et al.* (2013) J Immunol 190 (2) 794-804. doi:10.4049/jimmunol.1202088

SCAN ME

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SITC and the author of this poster.

HiberCell